Abstract No:
3527

Monitoring botensilimab- and balstilimab-induced T cell dynamics in refractory mismatch repair proficient metastatic colorectal cancer

Authors:
Gertjan Rasschaert (1, 2), Ana Mendizabal-Sasieta (3), Allyson Peddle (2), Marta Grzelak (3), Robin Govaerts (1), Joseph Elan Grossman (4), Marta Soto (3), Qian Wu (2), Benny Johnson (4), Filip Van Herpe (1, 2), Steven O'Day (4), Dhan Chand (4), Sabine Tejpar (1, 2), Holger Heyn (3)

Affiliations:
1. Gastrointestinal Oncology department, Leuven University Hospitals, Leuven, Belgium
2. Laboratory for Digestive Oncology, Katholieke Universiteit Leuven, Leuven, Belgium
3. Omniscope Inc., Palo Alto, CA, United States
4. Agenus Inc, Lexington, MA, United States of America

Background

- Mismatch repair proficient (pMMR) metastatic colorectal cancer (mCRC) responds poorly to immune checkpoint inhibition (ICI).
- Assessment per RECIST does not always capture clinically valuable response.
- Understanding immune cell activities is critical for improving ICI treatment efficacy.
- T cells elicit anti-tumor specificity through their T cell receptors (TCR) and dynamic changes in the TCR repertoire are associated with clinical outcomes.
- Circulating T cells in the blood provide an accessible liquid biomarker to longitudinally quantify and track T cell activity at systems level.
- Here, we present temporal T cell tracking as a correlate of ICI efficacy in refractory pMMR mCRC patients treated with botensilimab (BOT; Fc-enhanced anti-CTLA-4 antibody) with or without balstilimab (BAL; anti-PD-1 antibody).

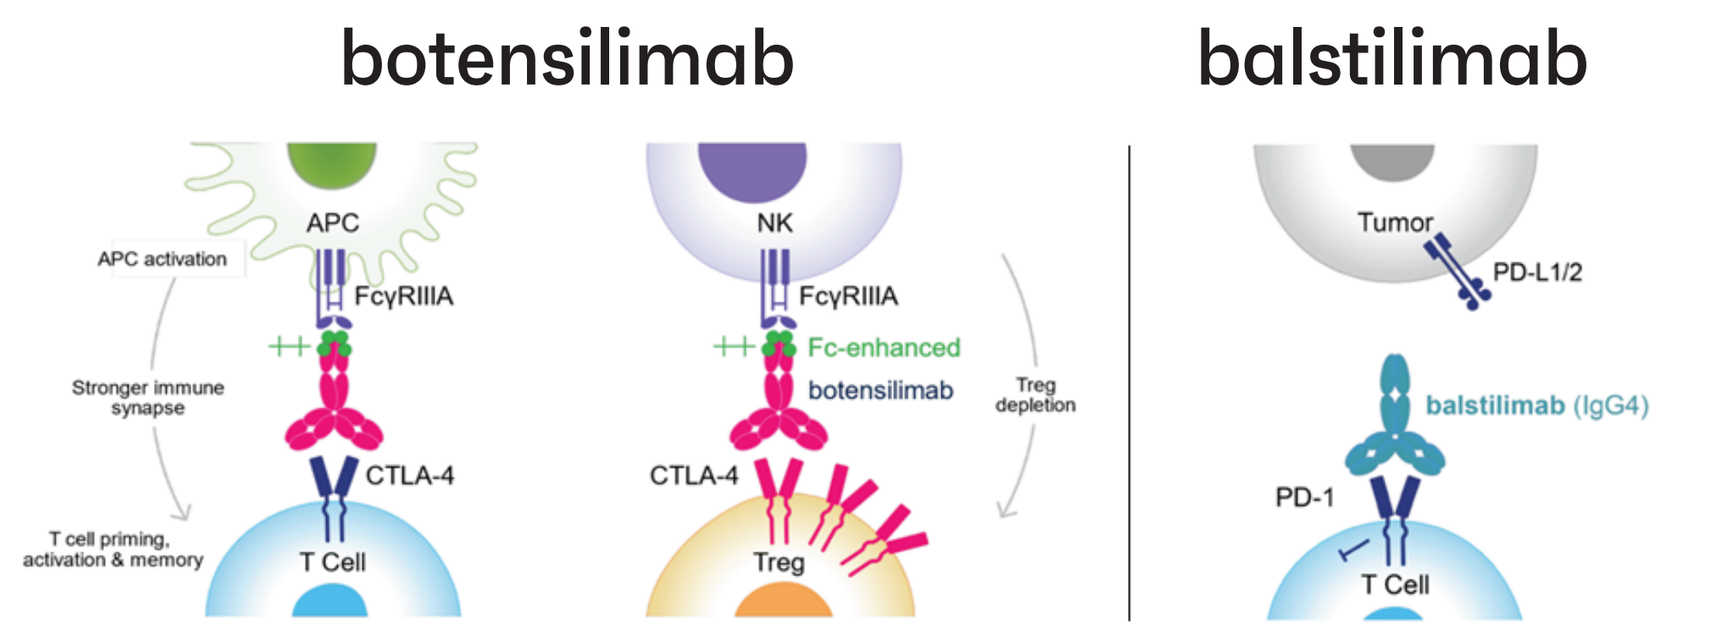
botensilimab
balstilimab
APC
APC activation
FcγRIIIA
Stronger immune synapse
CTLA-4
T cell priming, activation & memory
T Cell
NK
Fc-enhanced
botensilimab
Treg depletion
Treg
Tumor
PD-L1/2
balstilimab (IgG4)
PD-1

Figure 1. Botensilimab is an Fc-enhanced anti-CTLA-4 antibody designed to bind with high affinity to activating FcγRs and promote potent, complete blockade of CTLA-4. Balstilimab is an IgG4 anti-PD-1 (Programmed Death-1) antibody designed to bind with high affinity to PD-1 expressed on T cells and completely block the interaction between PD-1 and its ligands PD-L1/2 expressed by tumor cells and other immune cells.

Methods

10 patients from the open-label, phase 2 study (**NCT05608044**) with BOT in refractory pMMR CRC (without metastatic liver disease) were included. All patients had a primary tumour resection. In this trial, patients were randomized into BOT (75mg or 150mg Q6W, 4x) monotherapy or in combination with BAL (240mg Q2W, for 2 years), versus standard of care (regorafenib or trifluridine / tipiracil). Longitudinal assessment of TCR dynamics was performed on circulating T cells collected at baseline (week 0) and during treatment (weeks 2, 4, 6, and 12) using deep TCR sequencing (osTCR). T cell activity was quantified at each timepoint using the T cell Activity Score (TAS), which is calculated by an AI model (osTAS) that detects and quantifies therapy-induced T cell clones based on their expansion kinetics and functional clustering. Additionally, T cell diversity dynamics were analyzed.

ID	Age	Sex	Site primary	Differentiation grade	Mutation profile	Metastatic sites	Timing metastases	Response group
ITP-3	60	F	Rectum	Moderate	KRAS G12V	Lung	Synchronous	Progressive disease and alive
ITP-4	53	M	Right	Moderate	KRAS G12V, SMAD4, PIK3CA, APC, TP53	Peritoneum	Metachronous	Progressive disease and dead
ITP-5	73	F	Left	Moderate	KRAS G12D, PIK3CA, double APC, TP53	Lung, mediastinum	Synchronous	Progressive disease and dead
ITP-7	74	M	Rectum	Moderate	RAS wild type	Local, bone	Metachronous	Partial response
ITP-8	71	F	Rectum	Moderate	KRAS G12A	Lung	Synchronous	Progressive disease and dead
ITP-9	60	M	Left	Well	RAS mt	Lung	Metachronous	Partial response
ITP-11	49	F	Left	Well	KRAS G12D	Lung, mediastiinum	Metachronous	Progressive disease and alive
ITP-13	52	F	Left	Moderate	KRAS G12D, SMAD4, APC	Lung, pleura, mediastinum, pericard, bone, ovarium	Synchronous	Progressive disease and dead
ITP-14	67	M	Right	Mucinous	KRAS G12V, TP53	Local, lung	Synchronous	Progressive disease and alive
ITP-15	73	M	Left	Well	RAS mt	Lung	Metachronous	Progressive disease and dead

Table 1. Patients' characteristics.

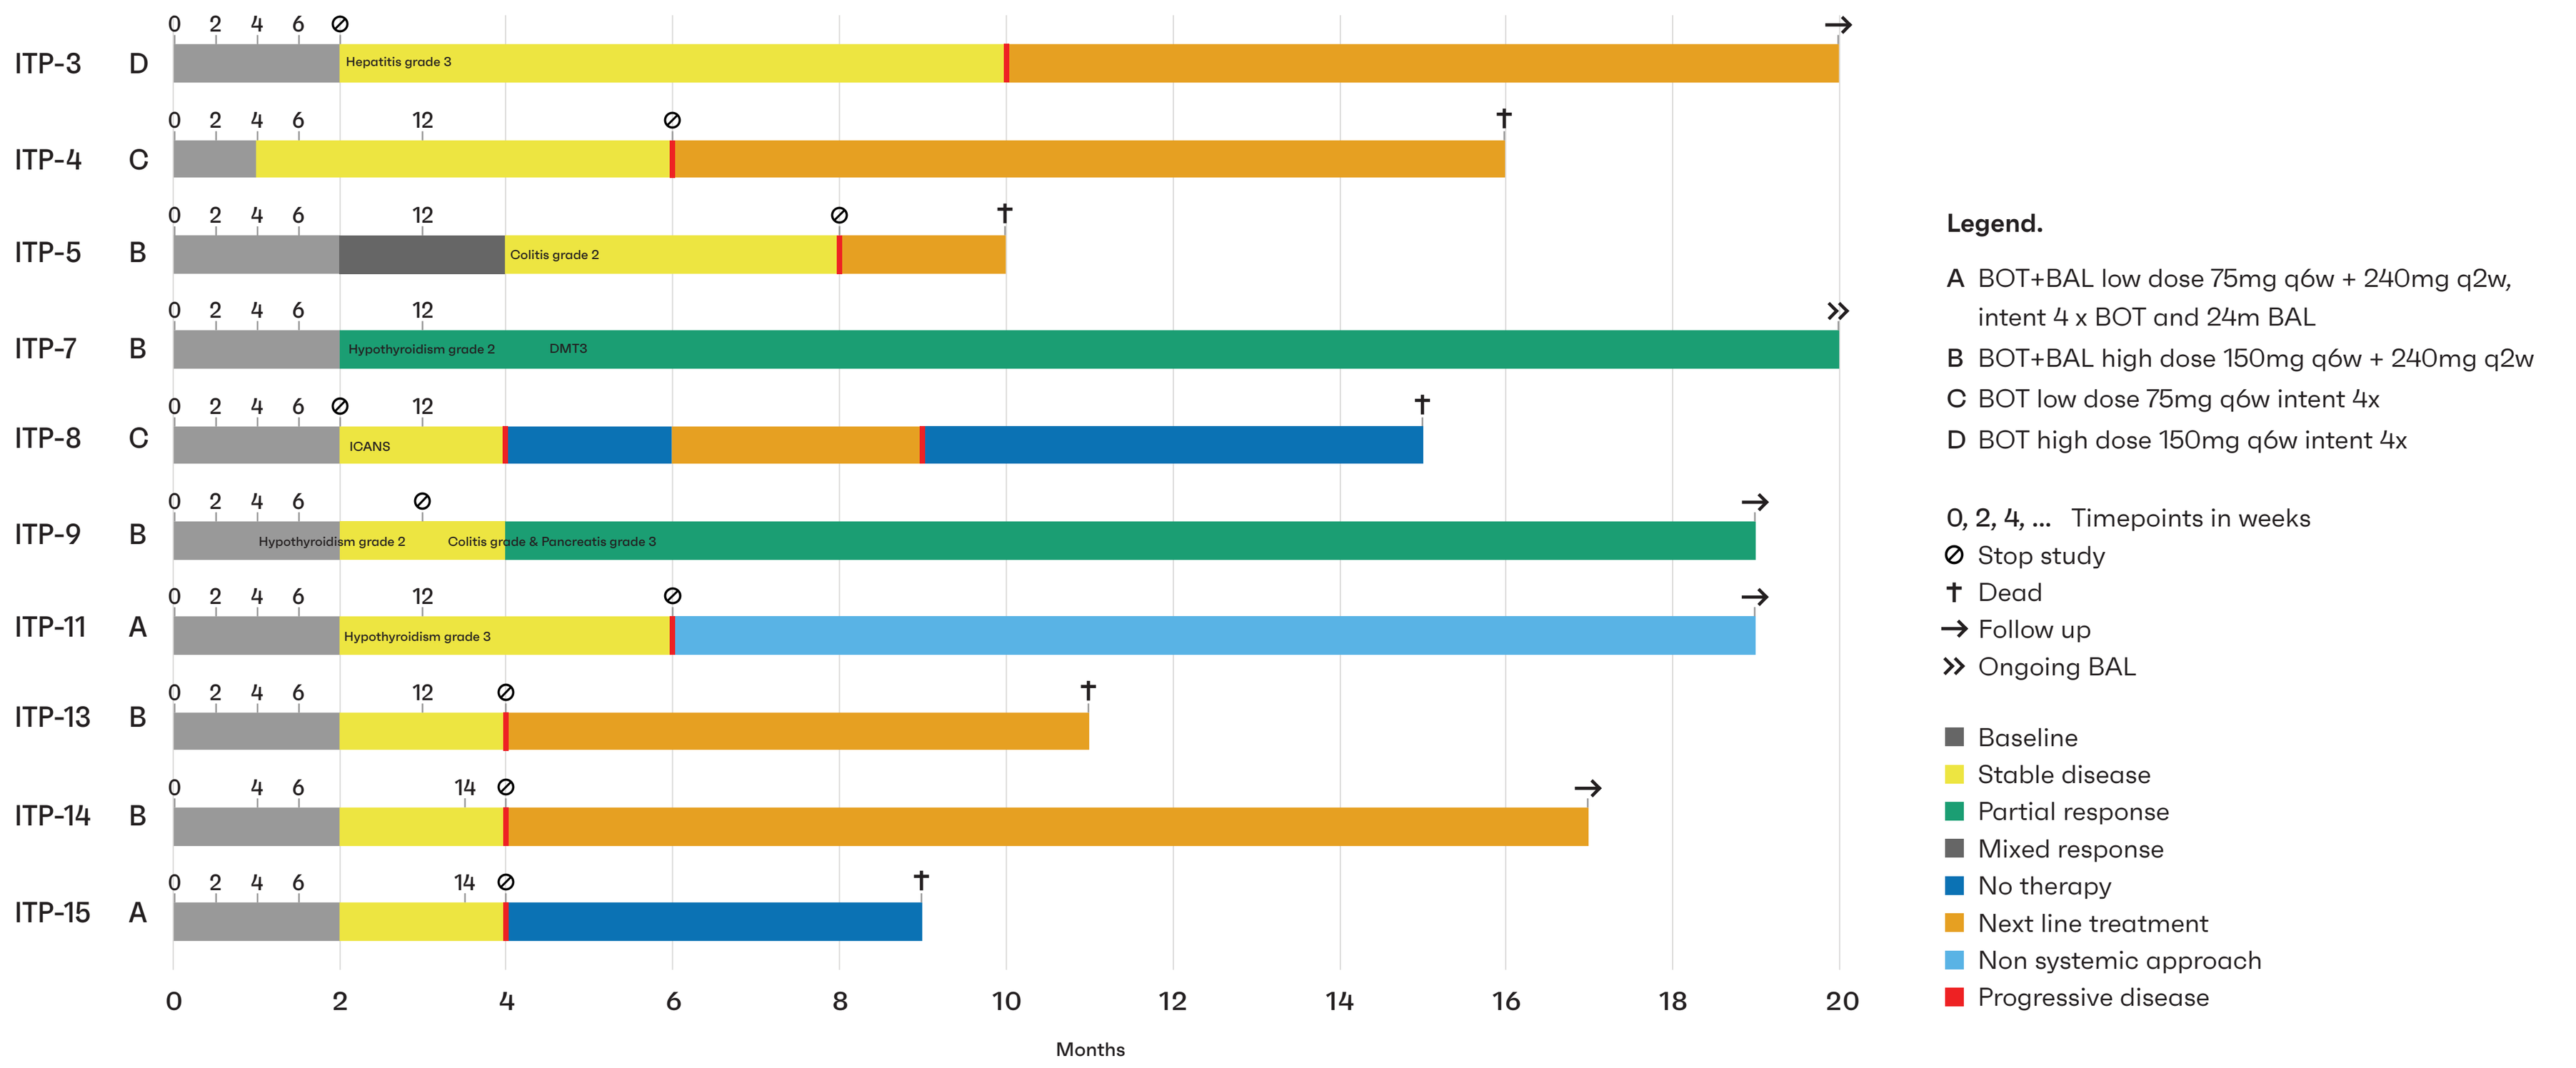
ITP-3 D
ITP-4 C
ITP-5 B
ITP-7 B
ITP-8 C
ITP-9 B
ITP-11 A
ITP-13 B
ITP-14 B
ITP-15 A
Hepatitis grade 3
Colitis grade 2
Hypothyroidism grade 2
DMT3
ICANS
Colitis grade & Pancreatitis grade 3
Hypothyroidism grade 3
Months
Legend.
A BOT+BAL low dose 75mg q6w + 240mg q2w, intent 4 x BOT and 24m BAL
B BOT+BAL high dose 150mg q6w + 240mg q2w
C BOT low dose 75mg q6w intent 4x
D BOT high dose 150mg q6w intent 4x
0, 2, 4, ... Timepoints in weeks
Stop study
Dead
Follow up
Ongoing BAL
Baseline
Stable disease
Partial response
Mixed response
No therapy
Next line treatment
Non systemic approach
Progressive disease

Figure 2. Swimmer plot. Clinical data cut off at February 2025.

Results

Therapy-induced T cell clones identified after first cycle persist throughout course of treatment.

In a partial responder (ITP-7), longitudinal analysis demonstrated that a significant fraction of T cell clones induced by the initial treatment cycle persisted and frequently expanded throughout the 12-week monitoring period.

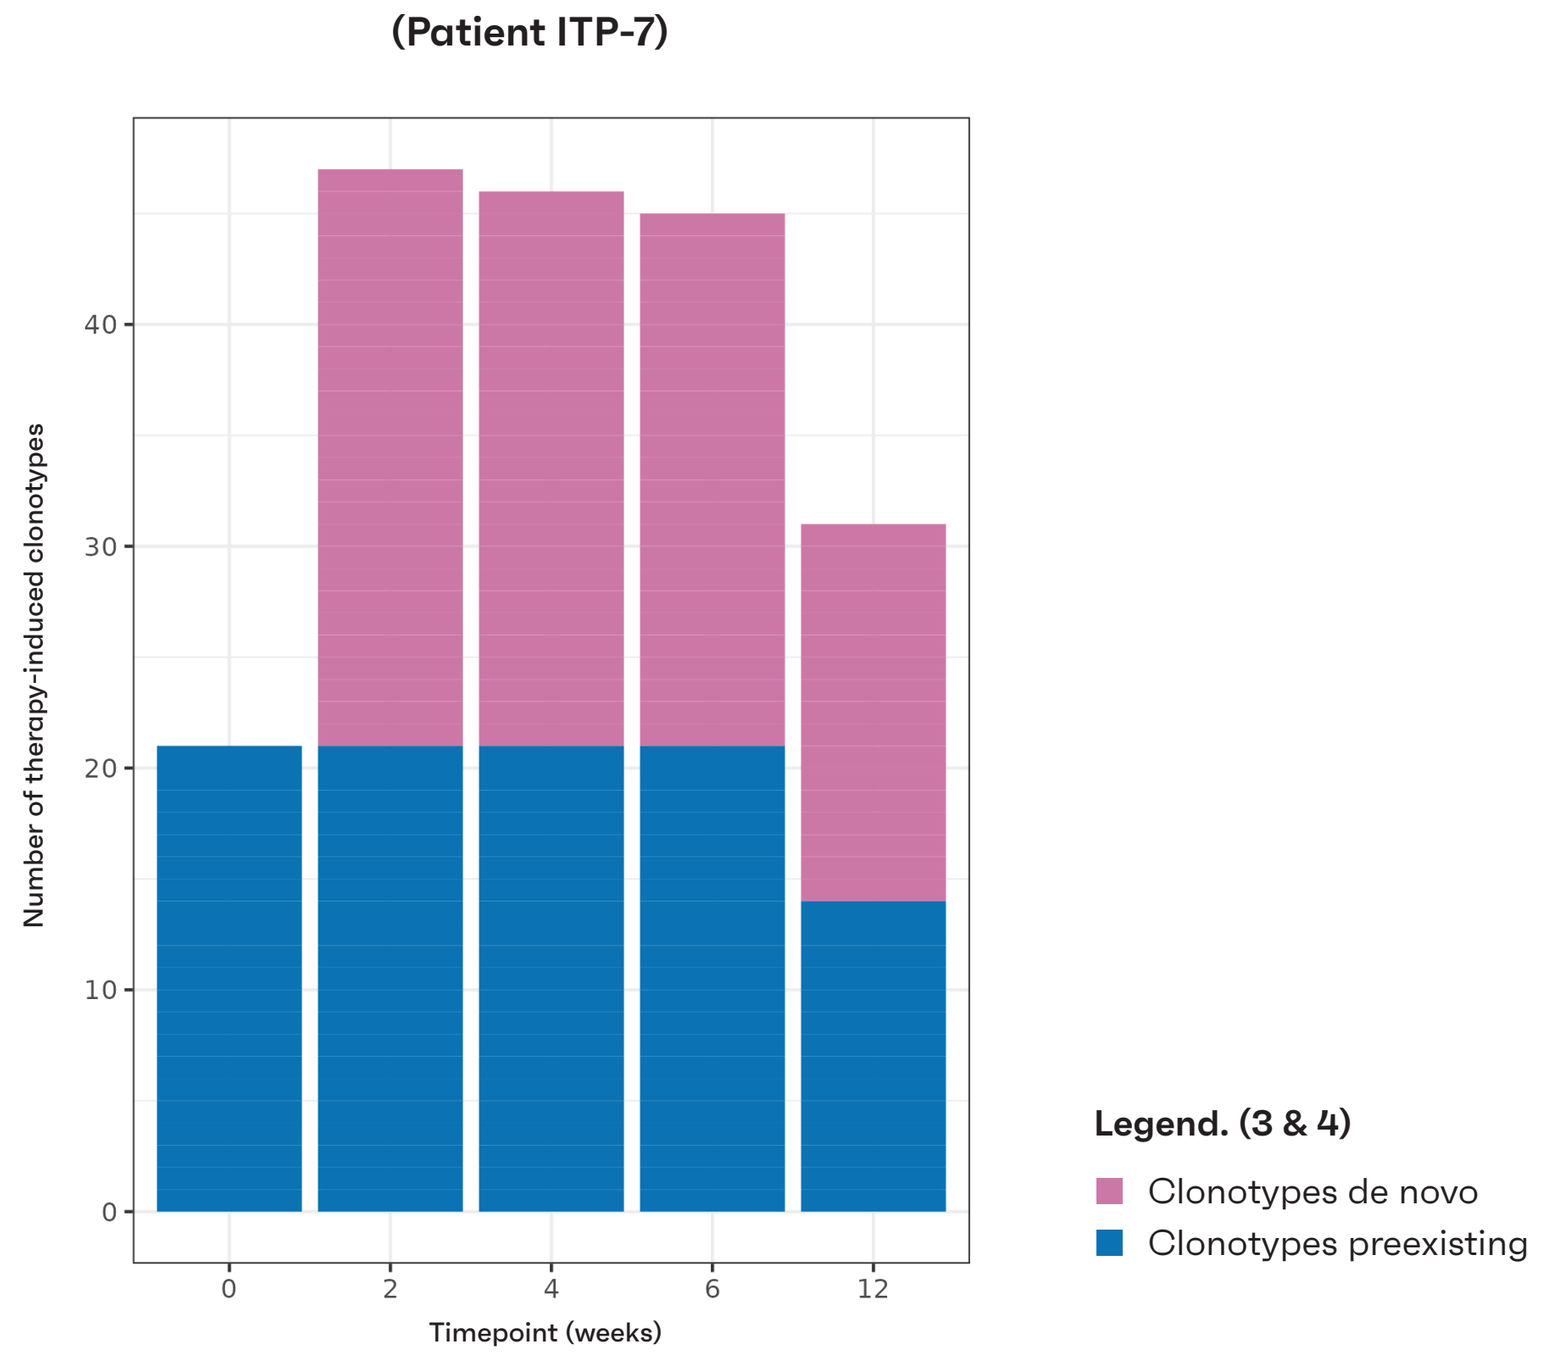
Evolution of therapy-induced clonotype presence (Patient ITP-7)
Number of therapy-induced clonotypes
Timepoint (weeks)
Legend. (3 & 4)
Clonotypes de novo
Clonotypes preexisting

Figure 3. Dynamics of therapy-induced TCR clonotype counts in a partial responder patient (ITP-7) from baseline to week 12, as identified by osTAS. The number of therapy-induced clonotypes remained relatively stable after the initial increase observed post-first cycle.

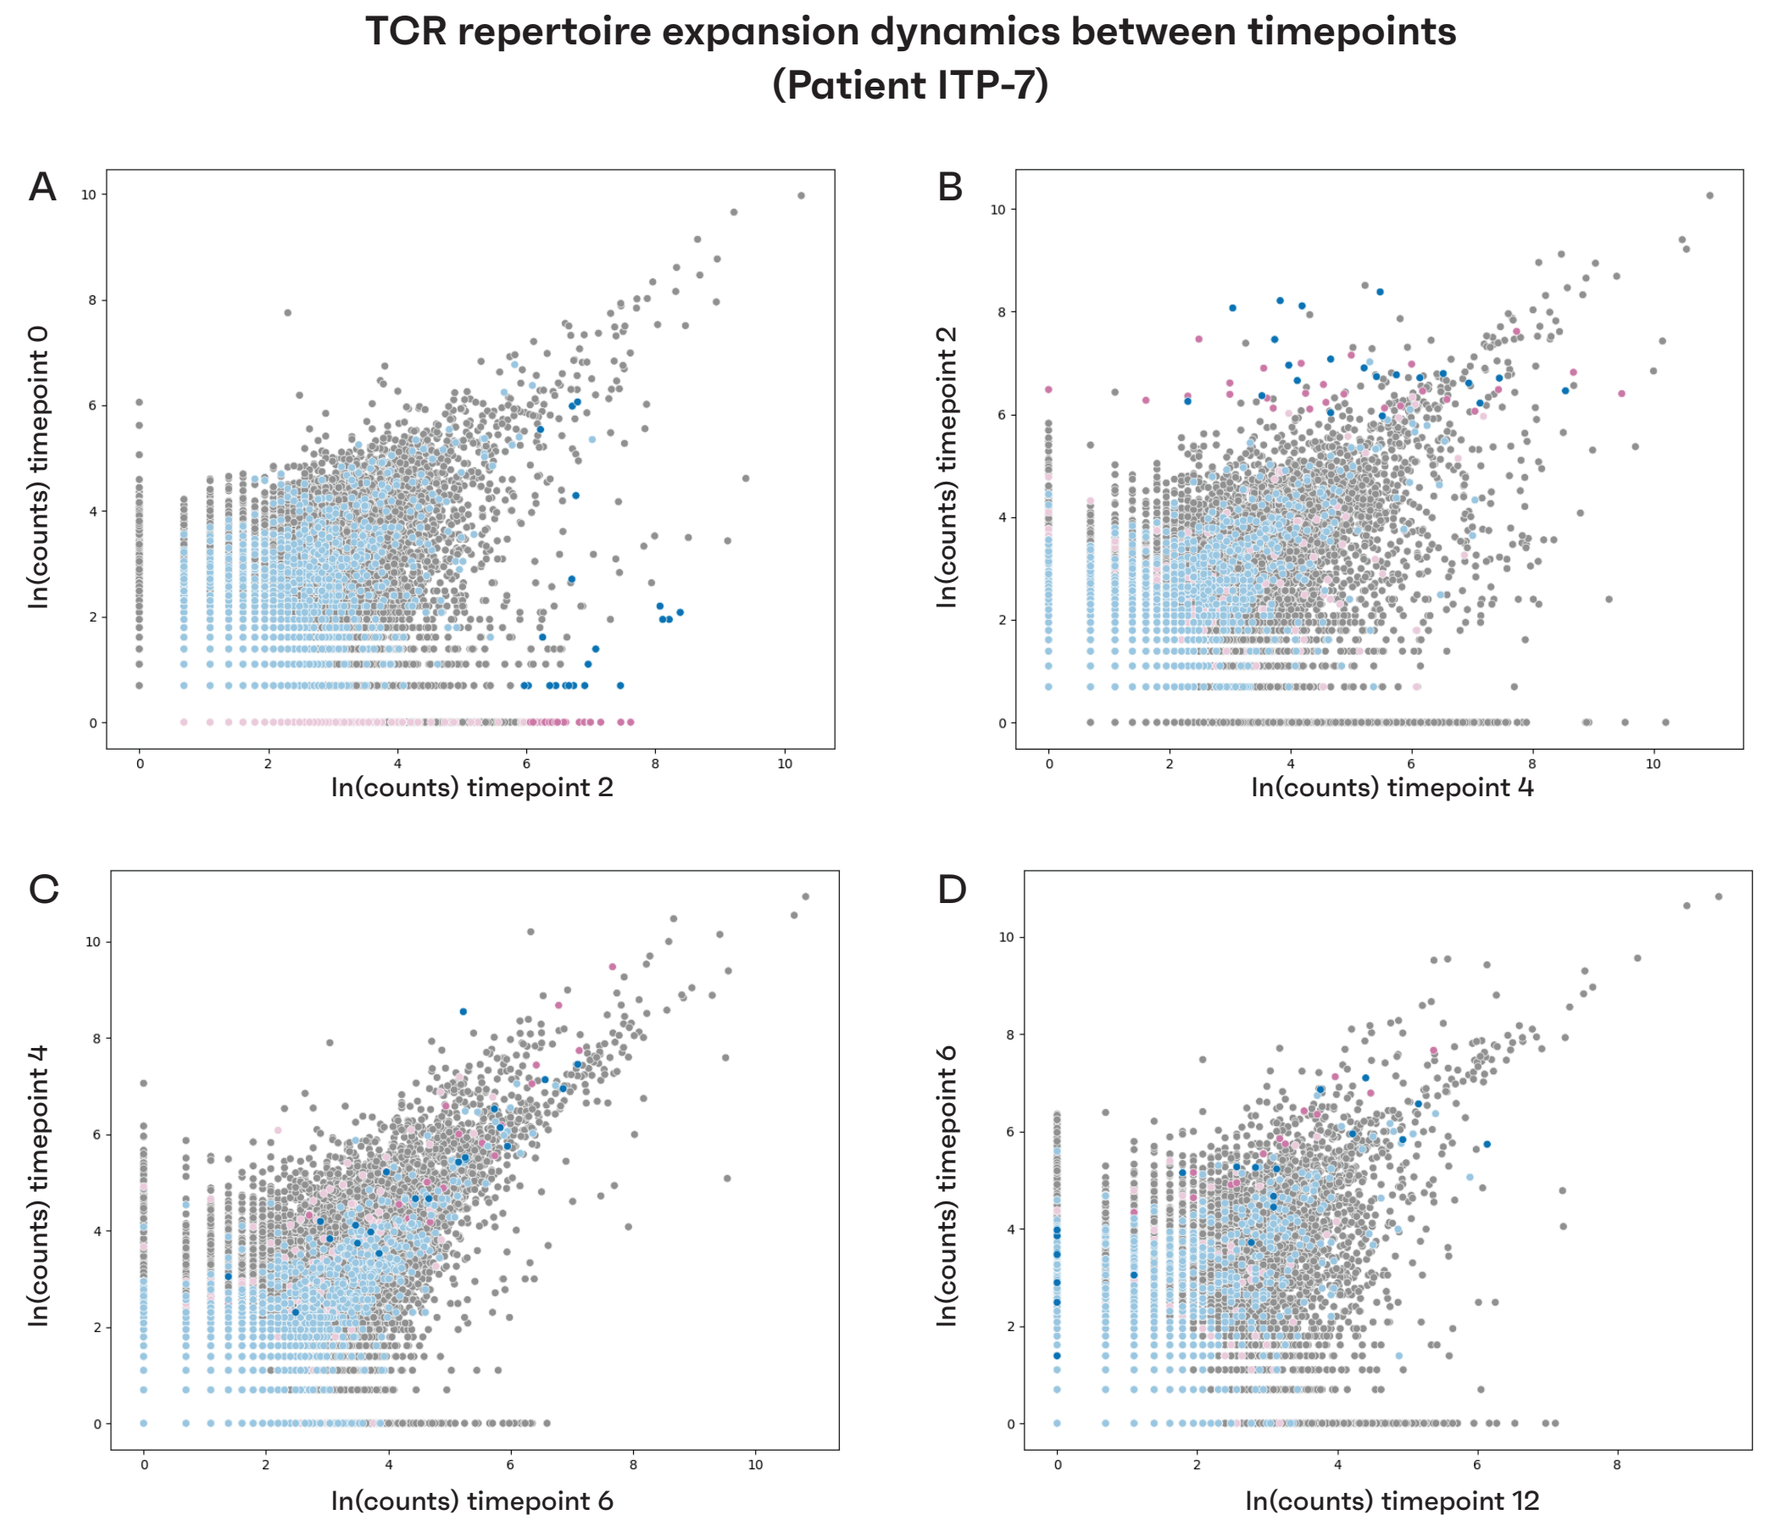
TCR repertoire expansion dynamics between timepoints (Patient ITP-7)
A
B
C
D
ln(counts) timepoint 0
ln(counts) timepoint 2
ln(counts) timepoint 4
ln(counts) timepoint 6
ln(counts) timepoint 12

Figure 4. TCR repertoire expansion dynamics in patient ITP-7, comparing consecutive timepoints. Significantly expanded TCR clonotypes after the first treatment cycle are highlighted in dark blue (pre-existing at baseline) and dark pink (de novo). Light blue and light pink dots represent clonotypes functionally clustered with these significantly expanded clones. Gray dots depict the remaining TCR repertoire. Notably, many therapy-induced clones (blue and pink) persist and often expand further in subsequent timepoints.

Expansion of treatment induced TCR clonotypes is associated with clinical response.

The T cell Activity Score, which quantifies the expansion of treatment-induced TCR clonotypes, demonstrated a clear association with clinical response categories defined prior to analysis by RECIST criteria. Patients achieving a Partial Response (PR) exhibited the highest TAS, followed by the group with Progressive Disease but Alive (PDA) showing intermediate scores, while patients with Progressive Disease and Dead (PDD) displayed the lowest TAS. This trend was particularly pronounced when considering only clusters of pre-existing therapy-induced TCRs.

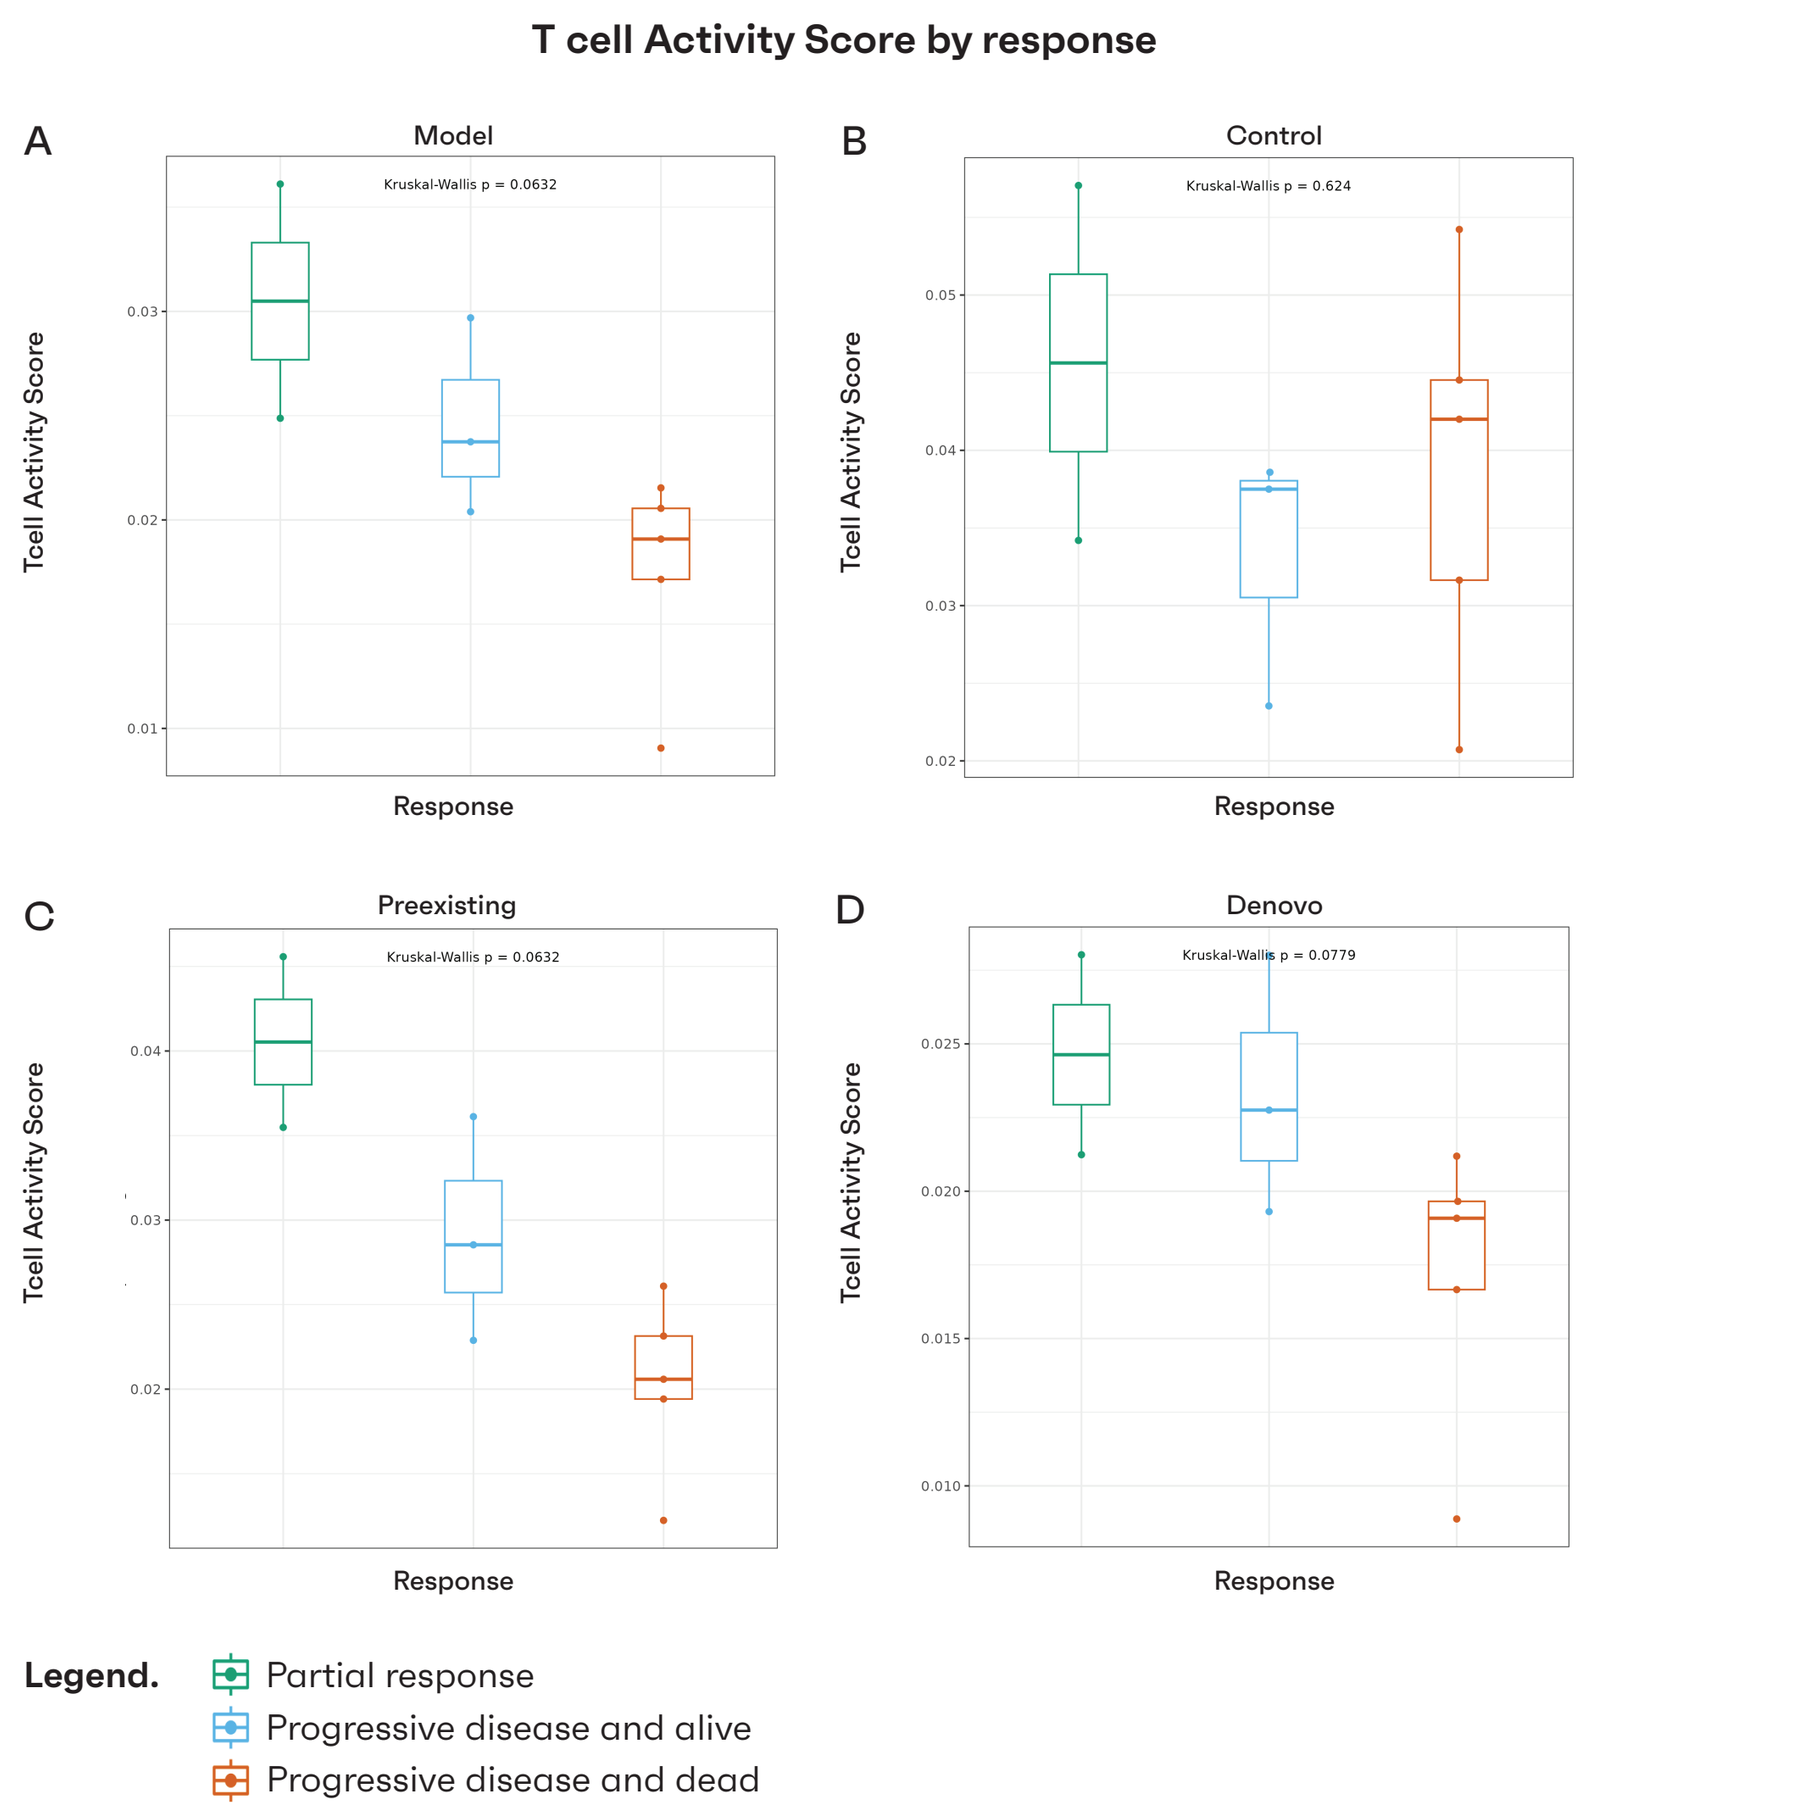
T cell Activity Score by response
A Model
Kruskal-Wallis p = 0.0632
B Control
Kruskal-Wallis p = 0.624
C Preexisting
Kruskal-Wallis p = 0.0632
D Denovo
Kruskal-Wallis p = 0.0779
Tcell Activity Score
Response
Legend.
Partial response
Progressive disease and alive
Progressive disease and dead

Figure 5. Boxplots illustrating the distribution of TAS across different response groups: PR, PDA, and PDD. Subplots show: A) Overall TAS as determined by the model, B) A control analysis using the same model parameters but on non significantly expanded clonotypes, C) TAS calculated considering only pre-existing TCRs identified as therapy-induced, and D) TAS calculated considering only de novo TCRs identified as therapy-induced.

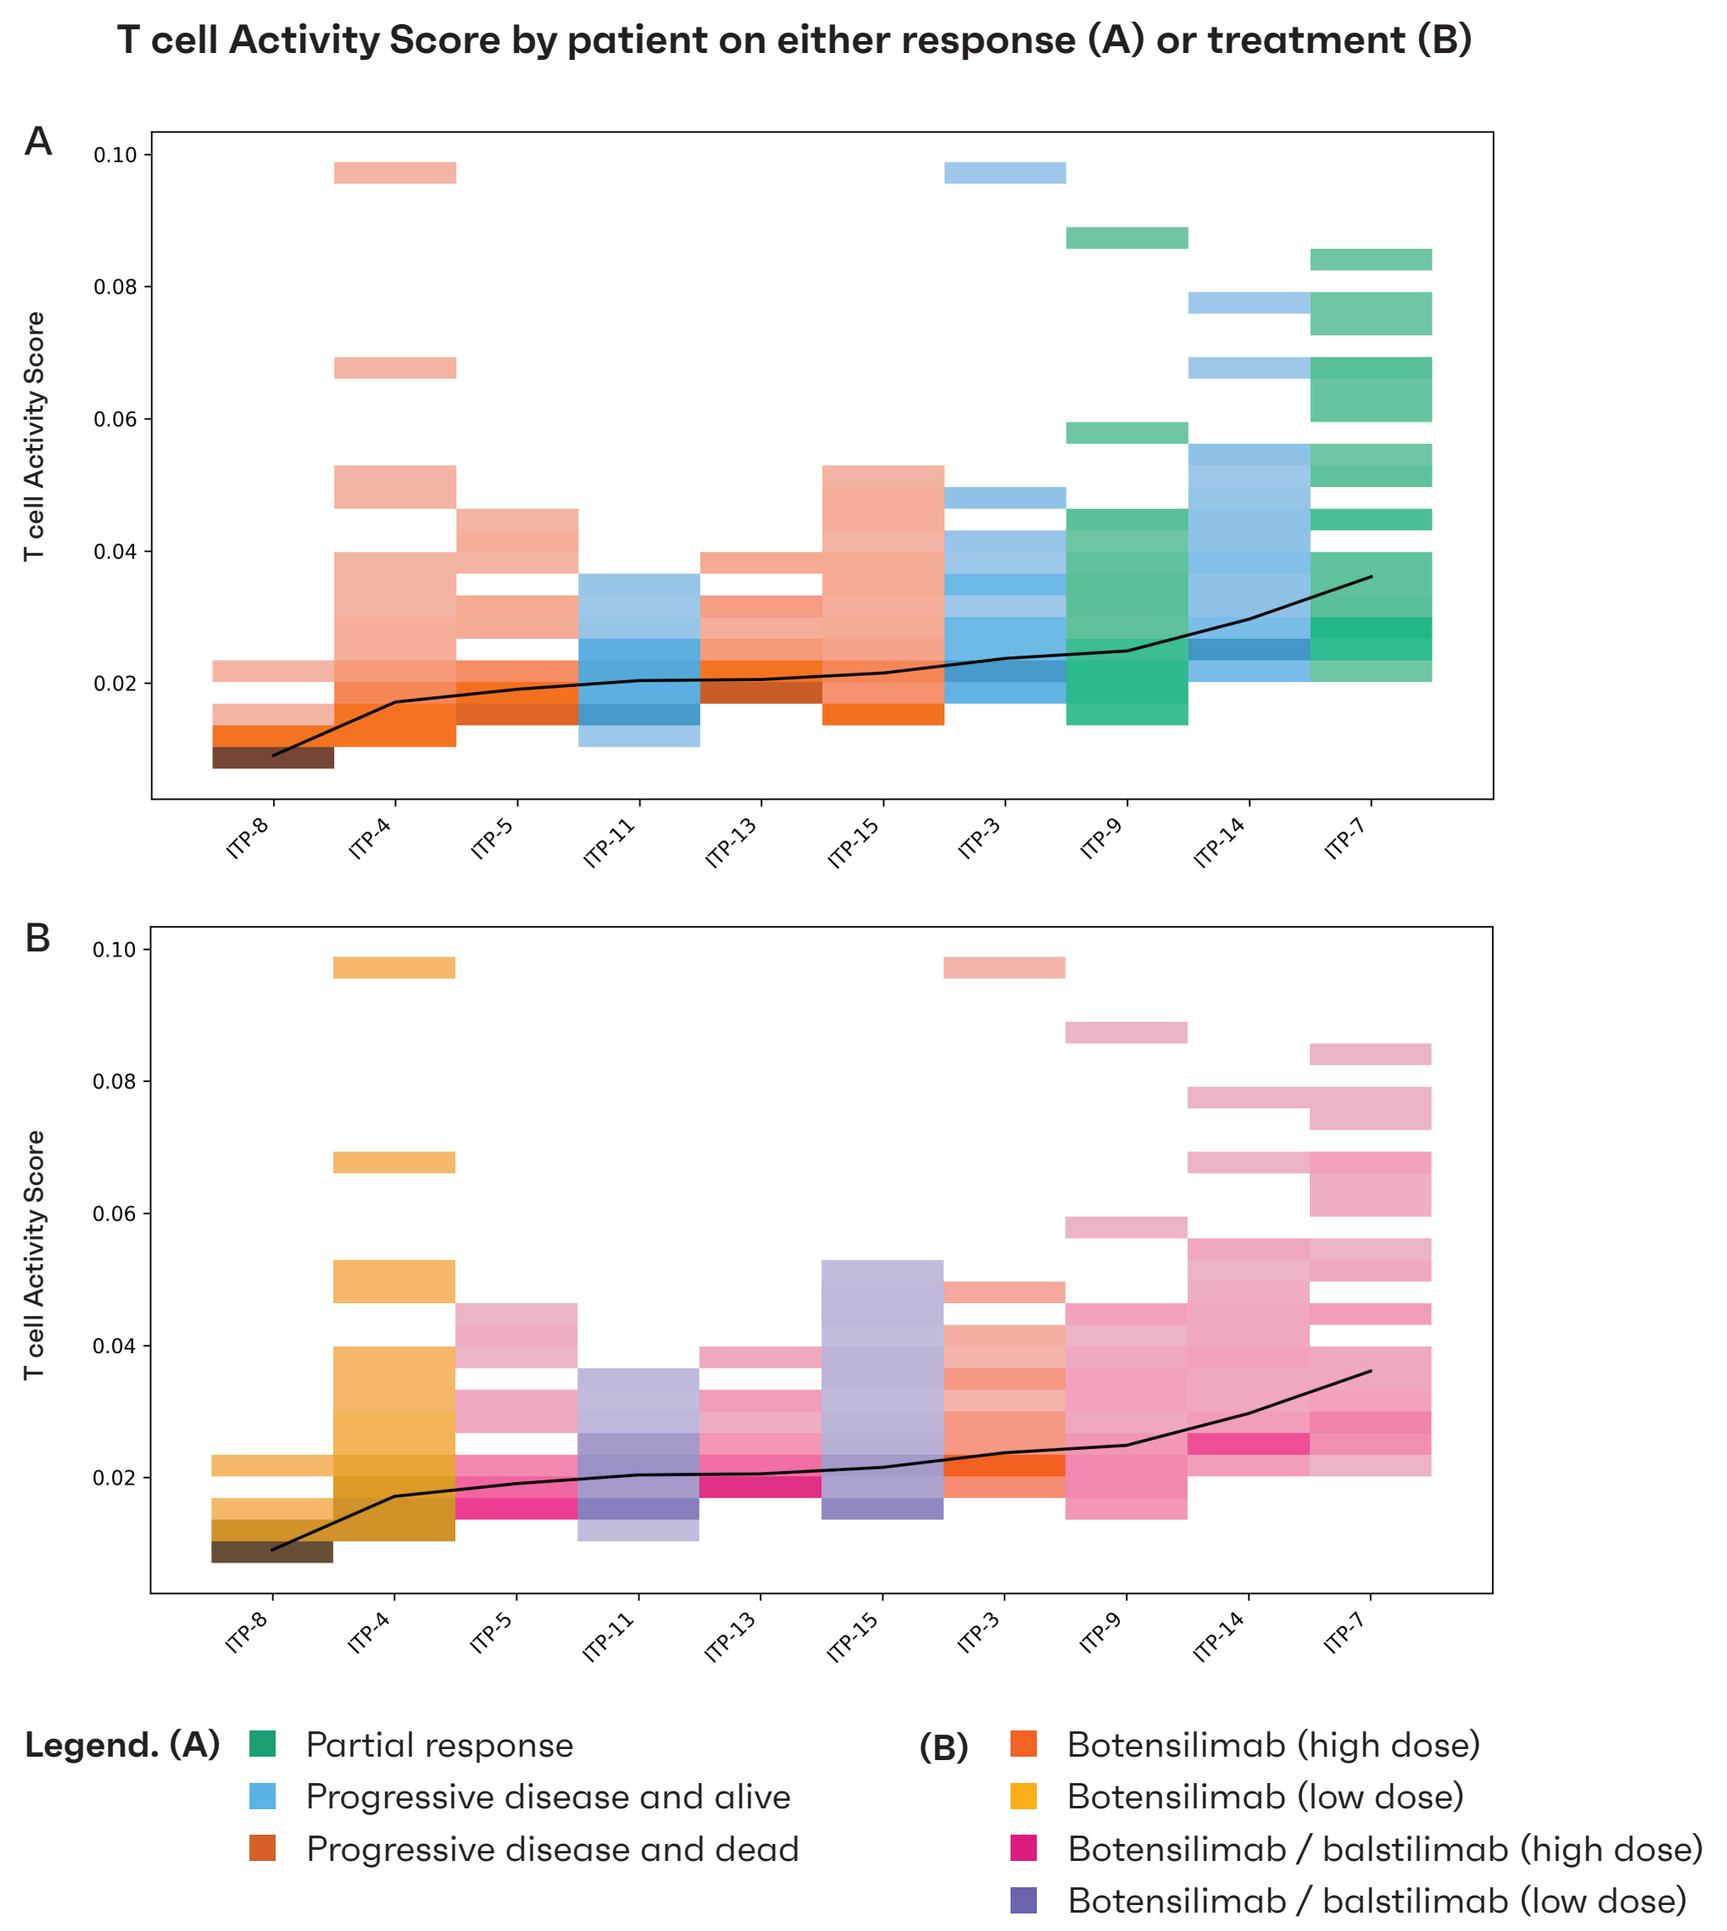
T cell Activity Score by patient on either response (A) or treatment (B)
T cell Activity Score
ITP-8
ITP-4
ITP-5
ITP-11
ITP-13
ITP-15
ITP-3
ITP-9
ITP-14
ITP-7
Legend. (A)
Partial response
Progressive disease and alive
Progressive disease and dead
(B)
Botensilimab (high dose)
Botensilimab (low dose)
Botensilimab / balstilimab (high dose)
Botensilimab / balstilimab (low dose)

Figure 6. Plot displaying individual patients ordered by their TAS. Bars are colored according to their clinical response (A) and treatment (B) arm. This visualization highlights the clustering of partial responders towards the higher end of the TAS spectrum.

Significant variations in TCR repertoire clonality dynamics after the first cycle of immunotherapy are observed between different response groups.

PR patients showed a marked increase in clonality post-first cycle, alongside sustained diversity. Progressive disease patients (PDA and PDD) exhibited less pronounced or absent clonality increases, with PDD sometimes showing increased diversity.

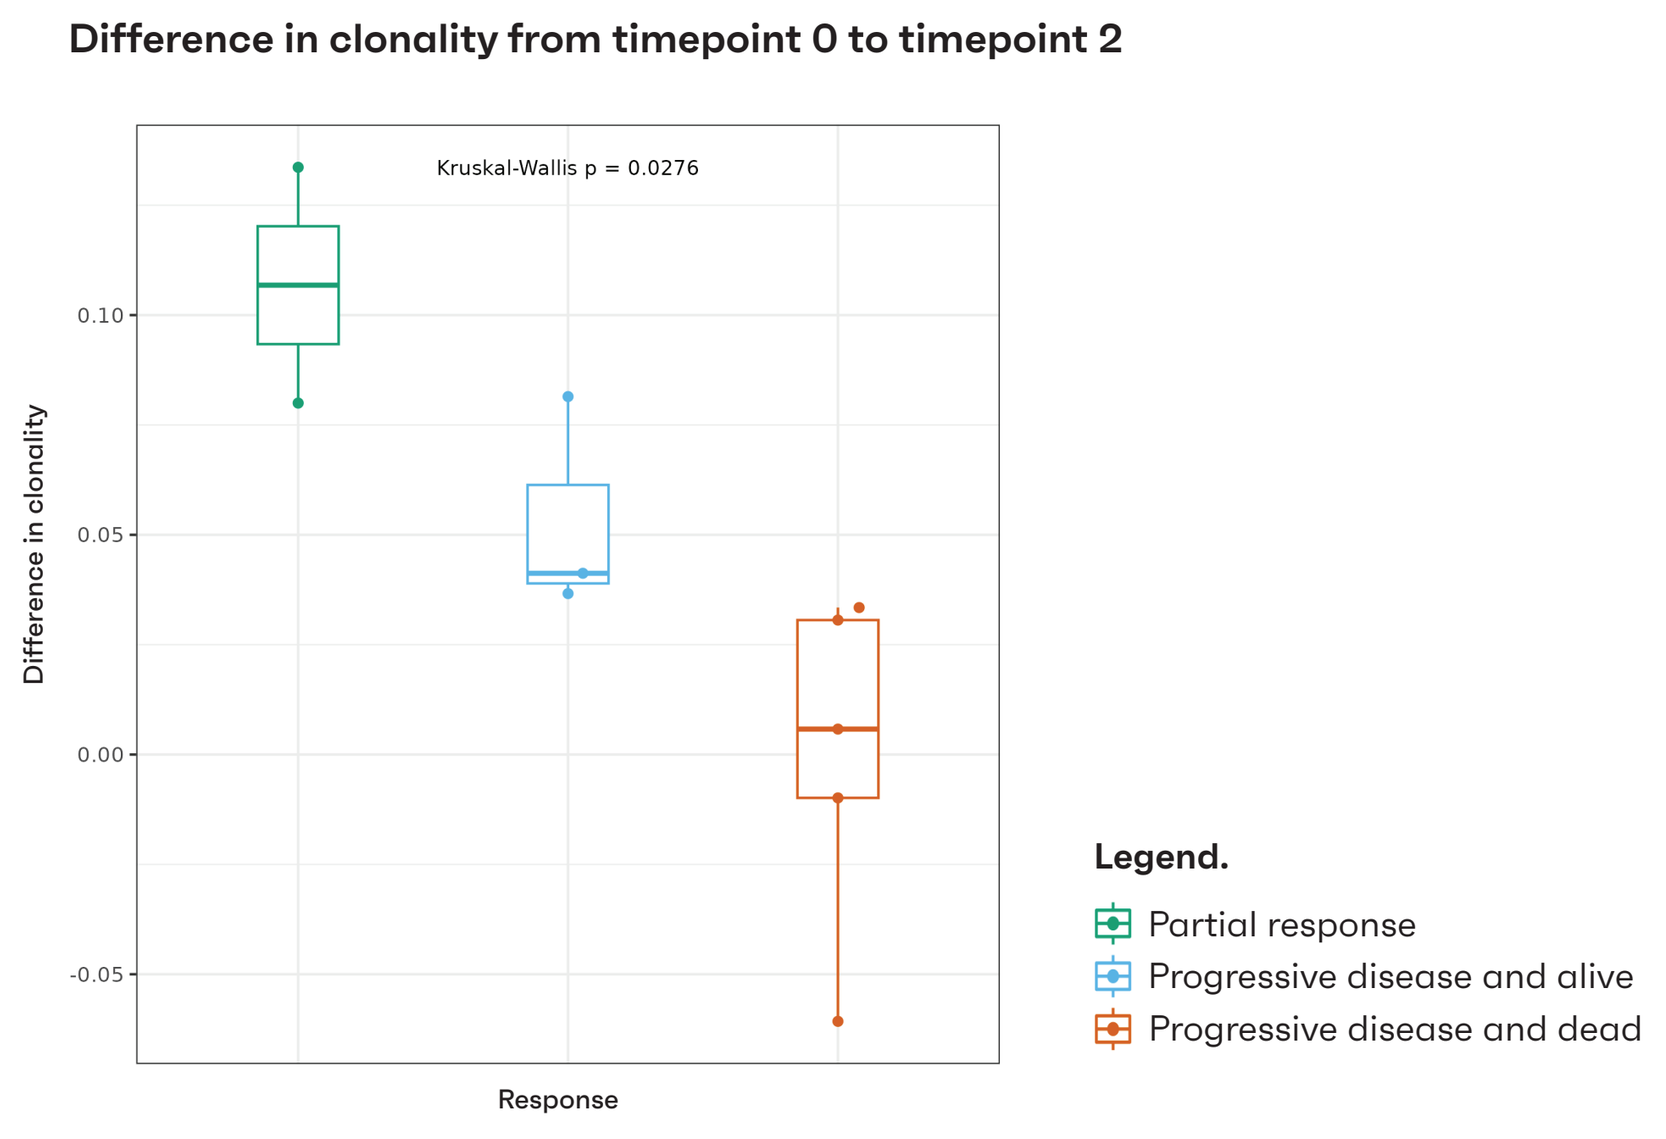
Difference in clonality from timepoint 0 to timepoint 2
Kruskal-Wallis p = 0.0276
Difference in clonality
Response
Legend.
Partial response
Progressive disease and alive
Progressive disease and dead

Figure 7. Boxplot showing the difference in TCR repertoire clonality between baseline and after the first cycle of treatment, stratified by clinical response group (PR, PDA, PDD).

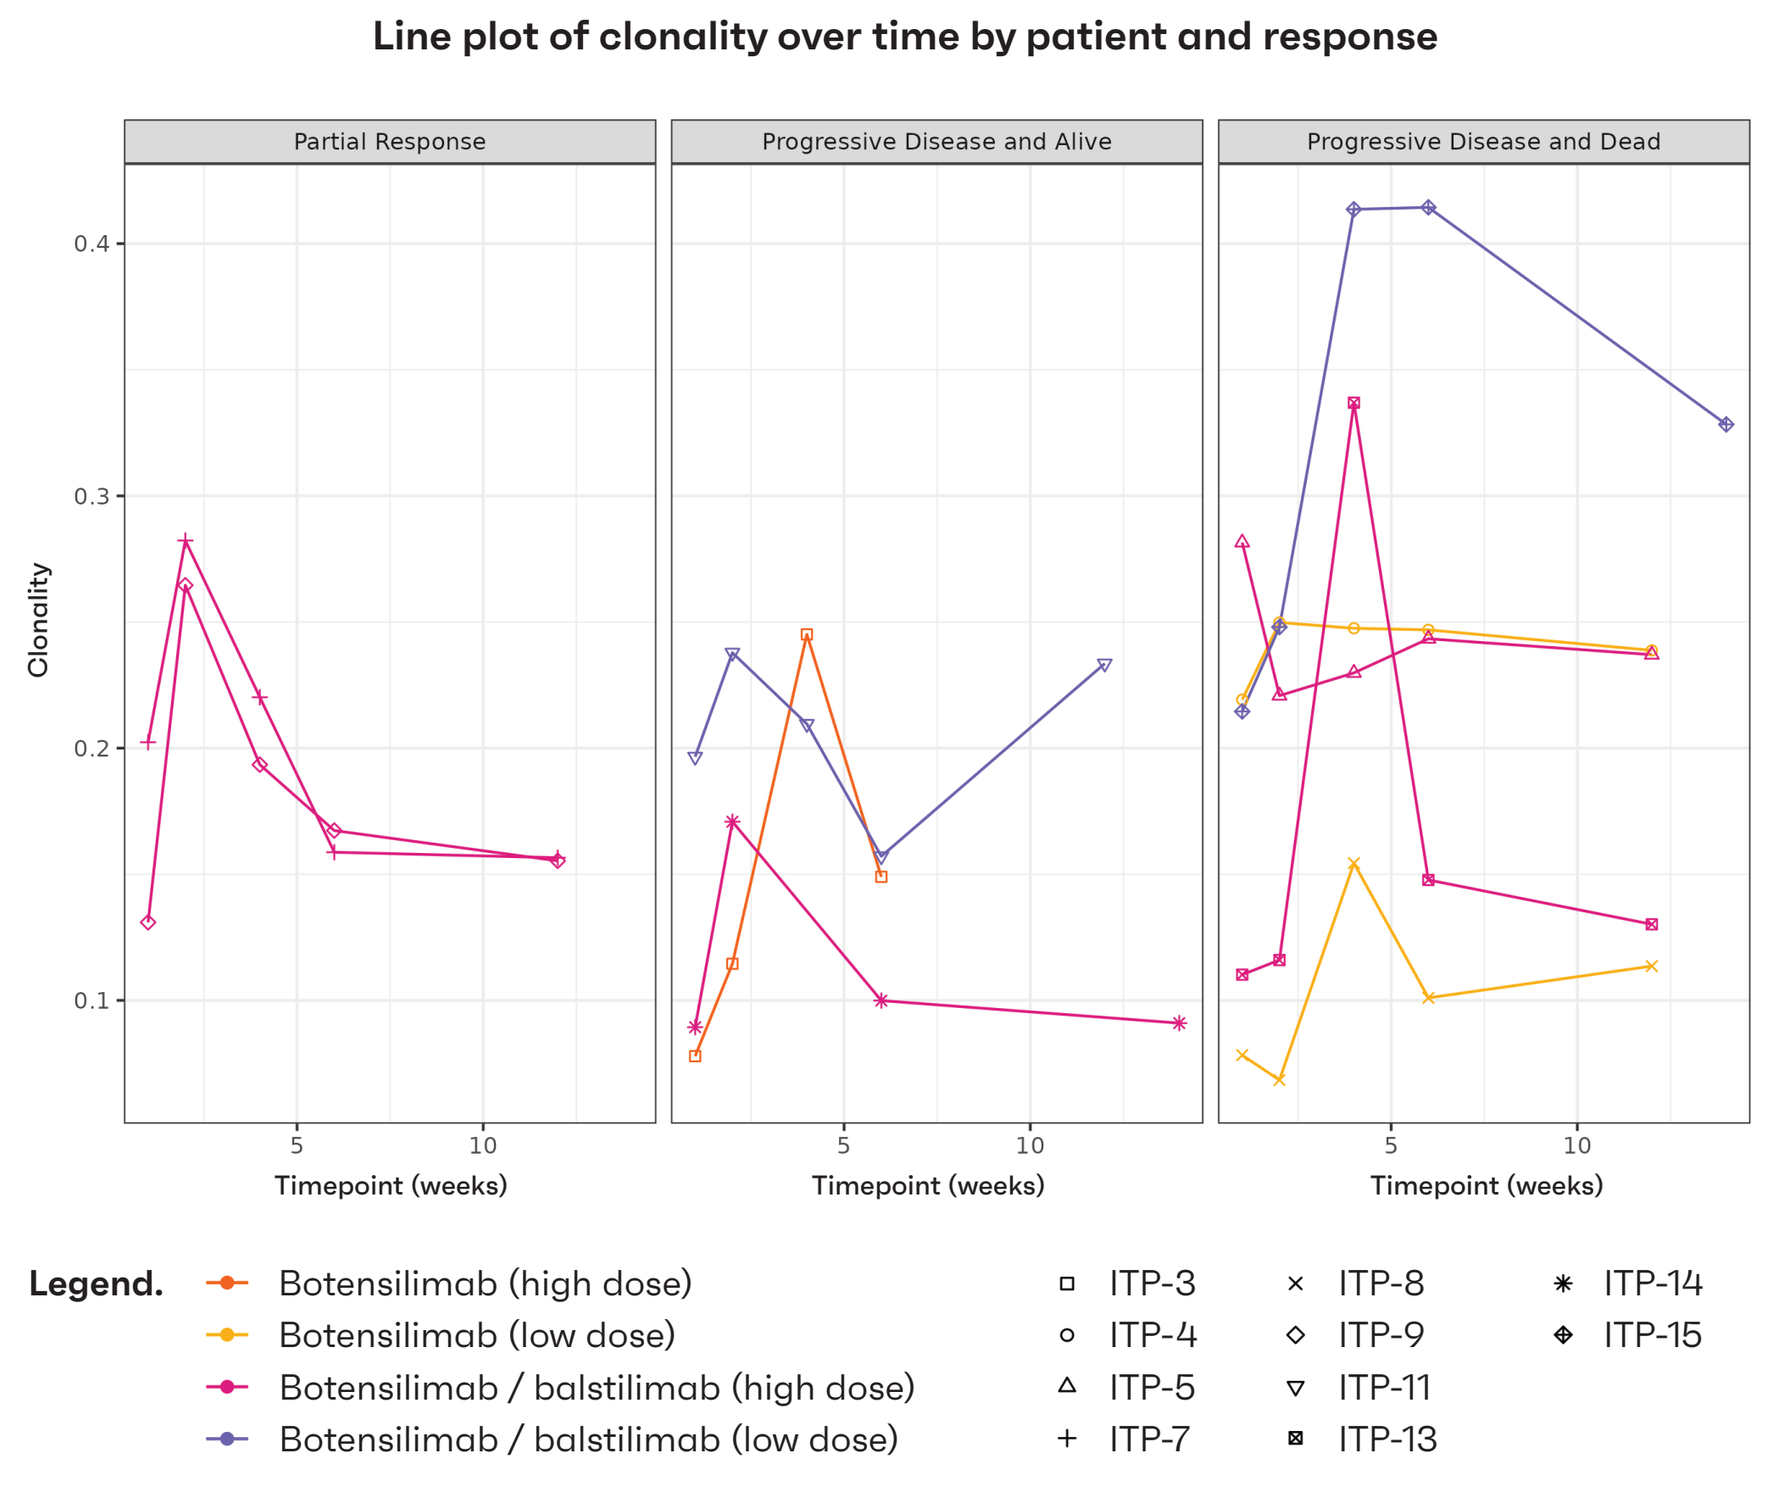
Line plot of clonality over time by patient and response
Partial Response
Progressive Disease and Alive
Progressive Disease and Dead
Clonality
Timepoint (weeks)
Legend.
Botensilimab (high dose)
Botensilimab (low dose)
Botensilimab / balstilimab (high dose)
Botensilimab / balstilimab (low dose)
ITP-3
ITP-4
ITP-5
ITP-7
ITP-8
ITP-9
ITP-11
ITP-13
ITP-14
ITP-15

Figure 8. Line plots depicting the clonality dynamics for individual patients throughout the treatment course, grouped by clinical response and colored by treatment arm.

Early T cell Activity Scores and clonal expansion following the first treatment cycle may possess prognostic value for patient stratification based on disease status and survival.

Analysis of early on-treatment changes revealed that the TAS and the degree of clonal expansion after the first treatment cycle show a positive correlation (Pearson correlation = 0.624). Notably, when plotting these two parameters against each other, distinct stratification of patient groups based on their clinical response (PR, PDA, and PDD) was observed.

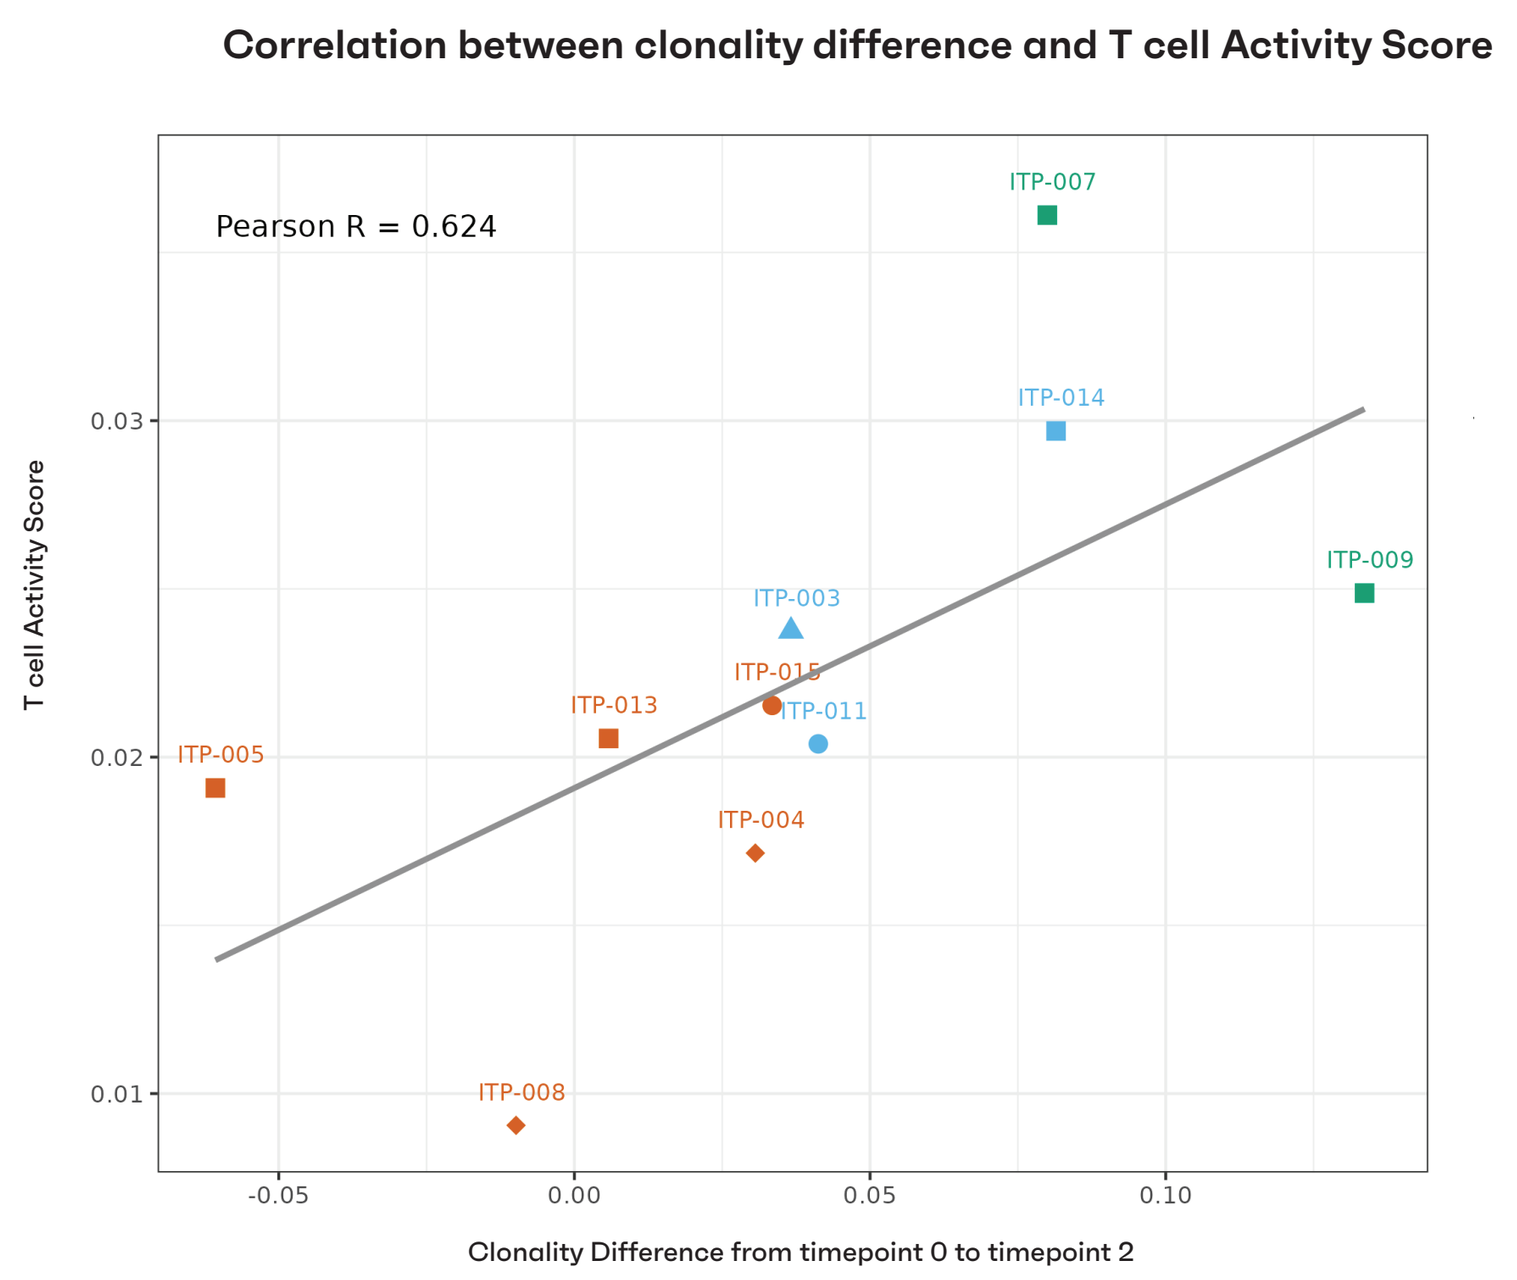
Correlation between clonality difference and T cell Activity Score
Pearson R = 0.624
ITP-007
ITP-014
ITP-009
ITP-003
ITP-015
ITP-011
ITP-013
ITP-005
ITP-004
ITP-008
T cell Activity Score
Clonality Difference from timepoint 0 to timepoint 2
Legend.
Partial response
Progressive disease and alive
Progressive disease and dead
Botensilimab (high dose)
Botensilimab (low dose)
Botensilimab / balstilimab (high dose)
Botensilimab / balstilimab (low dose)

Figure 9. Each point represents an individual patient. The Y-axis displays the TAS at week 2, and the X-axis represents the change in TCR repertoire clonality from baseline to week 2. We can visually separate patient groups according to their clinical response: PR, PDA, and PDD. A Pearson correlation coefficient of 0.624 indicates a positive association between early T cell activity and clonal expansion, with these early parameters showing potential for stratifying patients by response.

Conclusions

- TCR repertoire profiling is an emerging technique that captures therapy-induced immune responses.
- TCR metrics might explain clinical valuable responses not always captured by historical clinical oncology assessment metrics.
- TCR repertoire dynamics in the blood after the first cycle of ICI are associated with response, suggesting its potential role as a surrogate biomarker for outcome to immunotherapy.

Acknowledgments

The authors would like to thank Sofie Gielis (KU Leuven) and Juan Luis Melero Ollonarte (Omniscope) for their contribution in the final analysis of the results.

Contact

gertjan.rasschaert@uzleuven.be

Download poster

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.