January 23-25, 2025
Moscone West •
San Francisco, CA &
Online #GI25

#158. Preoperative botensilimab (BOT) with or without balstilimab (BAL) for patients with resectable, locally advanced pMMR or dMMR colon cancer: results from the UNICORN trial by GONO

Filippo Ghelardi[1], Margherita Ambrosini[1], Alessandra Raimondi[1], Giovanna Sabella[2], Riccardo Cerantola[3], Federica Palermo[1], Samantha Di Donato[4], Daniele Spada[5], Stefano Tamberi[6], Eleonora Perissinotto[3], Francesca Bergamo[3], Alessandro Passardi[7], Emiliano Tamburini[8], Riccardo Rosati[9], Andrea Sartore-Bianchi[10,11], Marcello Guaglio[12], Chiara Cremolini[13,14], Isacco Montroni[12], Sara Lonardi[3], Filippo Pietrantonio[1]

[1]Department of Medical Oncology, Fondazione IRCCS Istituto Nazionale dei Tumori Milan, Italy; [2]First Division of Pathology , Department of Pathology and Laboratory Medicine, Istituto Nazionale Tumori IRCCS Milan, Italy; [3]Medical Oncology 1, Veneto Institute of Oncology IOV - IRCCS, Padua, Italy; [4]Department of Medical Oncology, General Hospital, Prato, Italy; [5]Oncology Unit, ASST Cremona, Cremona, Italy; [6] Oncology Unit, Ravenna Hospital, AUSL Romagna, Ravenna, Italy; [7]Department of Medical Oncology, IRCCS Istituto Romagnolo per lo Studio dei Tumori "Dino Amadori", Meldola, Italy; [8]Department of Oncology and Palliative Care, Cardinale G Panico, Tricase City Hospital, Tricase, Italy; [9]Department of Surgery, San Raffaele Hospital, Milan, Italy; [10]Grande Ospedale Metropolitano Niguarda, Milan, Italy; [11]Department of Oncology and Hemato-Oncology, Università degli Studi di Milano, Milan, Italy; [12]Department of Surgery, Istituto Nazionale Tumori IRCCS Milan, Italy; [13]Unit of Medical Oncology 2, University Hospital of Pisa, Pisa, Italy; [14]Department of Translational Research and New Technologies in Medicine and Surgery, University of Pisa, Pisa, Italy.

BACKGROUND

Standard treatment for patients (pts) with localized colon cancer (CC) is surgery and adjuvant chemotherapy, where indicated.
Neoadjuvant strategies allow for earlier exposure to systemic therapy, potentially leading to tumor downstaging, micro-metastases eradication, improved survival, and may pave the way for non-operative management (NOM). Previous experience with neoadjuvant ipilimumab and nivolumab resulted in major pathological response in 20% and 95% of patients with pMMR and dMMR tumors, respectively. BOT, a novel Fc-enhanced multifunctional anti-CTLA-4 antibody plus BAL, an anti-PD-1 antibody, have demonstrated activity in proficient mismatch repair (pMMR) CC pts with durable responses in metastatic CRC and promising rates of pathological complete response (pCR) in the localized setting.

METHODS

UNICORN (NCT05845450) is a window-of-opportunity, multicohort, umbrella platform phase II trial enrolling non-metastatic, radiologically staged rT3-4 N0-2, resectable CRC pts to be treated with a short course preoperative targeted treatment according to a prespecified molecular profile assessed by immunohistochemistry and next-generation sequencing on tumor biopsy.
Pts with pMMR tumors received intravenous (IV) BOT at 1 mg/kg on day 1 (cohort 4) or IV BOT 1 mg/kg on day 1 and BAL 3 mg/kg on days 1 and 15 (cohort 5, enrolled after 4) and underwent radical surgery on day 35 $\pm$ 5. Pts with dMMR tumors received IV BOT 1 mg/kg on day 1 (cohort 6) or IV BOT 1 mg/kg on day 1 and BAL 3 mg/kg on days 1 and 15 (cohort 7, enrolled after 6) and underwent radical surgery on day 35 $\pm$ 5.
Pathological response (pR), major response (pMR) and pCR rates were defined as ≤ 50%, ≤10% and 0% residual viable tumor on surgical specimen. The primary endpoint was centrally assessed pMR rate in each cohort. According to a Fleming 1-stage design, choosing β=80% and α=5%, selecting H0=15% and H1=45%, 14 pts were enrolled in each cohort and the treatment was judged promising if ≥5/14 pMRs were observed.

RESULTS

All 56 enrolled pts completed preoperative treatment and underwent surgery. Timely surgery was performed in most pts (98%), except 1 who underwent surgery with a delay < 4 weeks due to treatment-related hyperthyroidism.

mail: **filippo.ghelardi@istitutotumori.mi.it**

Baseline characteristics (**Table 1**) were superimposable among cohorts. Expectedly, a numerically higher proportion of right-sided tumors was observed in dMMR cohorts (79%).
As depicted in **Figure 1**, among pts with pMMR disease, activity was limited for BOT monotherapy (pMR 0%, pR 43%). With BOT + BAL, pCR was 29%, pMR 36% and pR 71%.
Among those with dMMR status, BOT led to pCR, pMR and pR in 29%, 36% and 64% pts, respectively. Notably, BOT + BAL led to pCR and pMR in 93% and 100% pts.

Table 1. Patient and disease characteristics

Characteristic	Cohort 4 n=14 (%)	Cohort 5 n=14 (%)	Cohort 6 n=14 (%)	Cohort 7 n=14 (%)
Age, years: median (IQR)	69 (56-75)	65 (57-70)	63 (54-72)	66 (52-71)
Sex				
Male	6 (43)	6 (43)	7 (50)	4 (29)
Female	8 (57)	8 (57)	7 (50)	10 (71)
Primary tumor location				
Left colon	8 (57)	6 (43)	3 (21)	3 (21)
Right colon	6 (43)	8 (57)	11 (79)	11 (79)
Lynch Syndrome				
Yes	0	0	2 (14)	1 (7)
No	0	0	12 (86)	13 (93)
Unknown	14 (100)	14 (100)	0	0
Adjuvant chemo				
Yes	10 (71)	8 (57)	3 (21)	0
FP	4 (29)	1 (7)	2 (14)	0
Ox-FP	6 (42)	7 (50)	1 (7)	0
No	4 (29)	6 (43)	11 (79)	14 (100)

Table 2. Immune-mediated AEs (n=56):

Adverse event	Any grade N (%)	Grade ≥3 N (%)
Any IMAE	22 (39)	1 (2)
Flu-like syndrome	7 (13)	0
Fatigue	3 (5)	0
Pruritus	3 (5)	0
Skin rash	3 (5)	0
Thyroiditis	3 (5)	1 (2)
Diarrhea	2 (4)	0
Hepatitis	1 (2)	0
Nephritis	1 (2)	0
Myalgias	1 (2)	0
Mucositis	1 (2)	0

Adverse events (AEs) of any grade occurred in 28/56 pts (50%), and 22 (39%) were deemed immune-mediated (IMAE) by investigator assessment (**Table 2**). Any IMAE occurred in 7 (25%) and 15 (54%) pts enrolled in BOT and BOT + BAL cohorts, respectively. Serious AEs occurred in 9 pts (16%) and were treatment-related in 3 (5%).
Among patients enrolled in cohort 4, 71% received subsequent adjuvant chemotherapy, mostly oxaliplatin (Ox) and fluoropyrimidine (FP)- based. A numerically lower proportion of patients enrolled in cohort 5 (57%) received adjuvant chemotherapy, possibly as result of tumor downstaging.

Figure 1. Waterfall plot of pathological regression

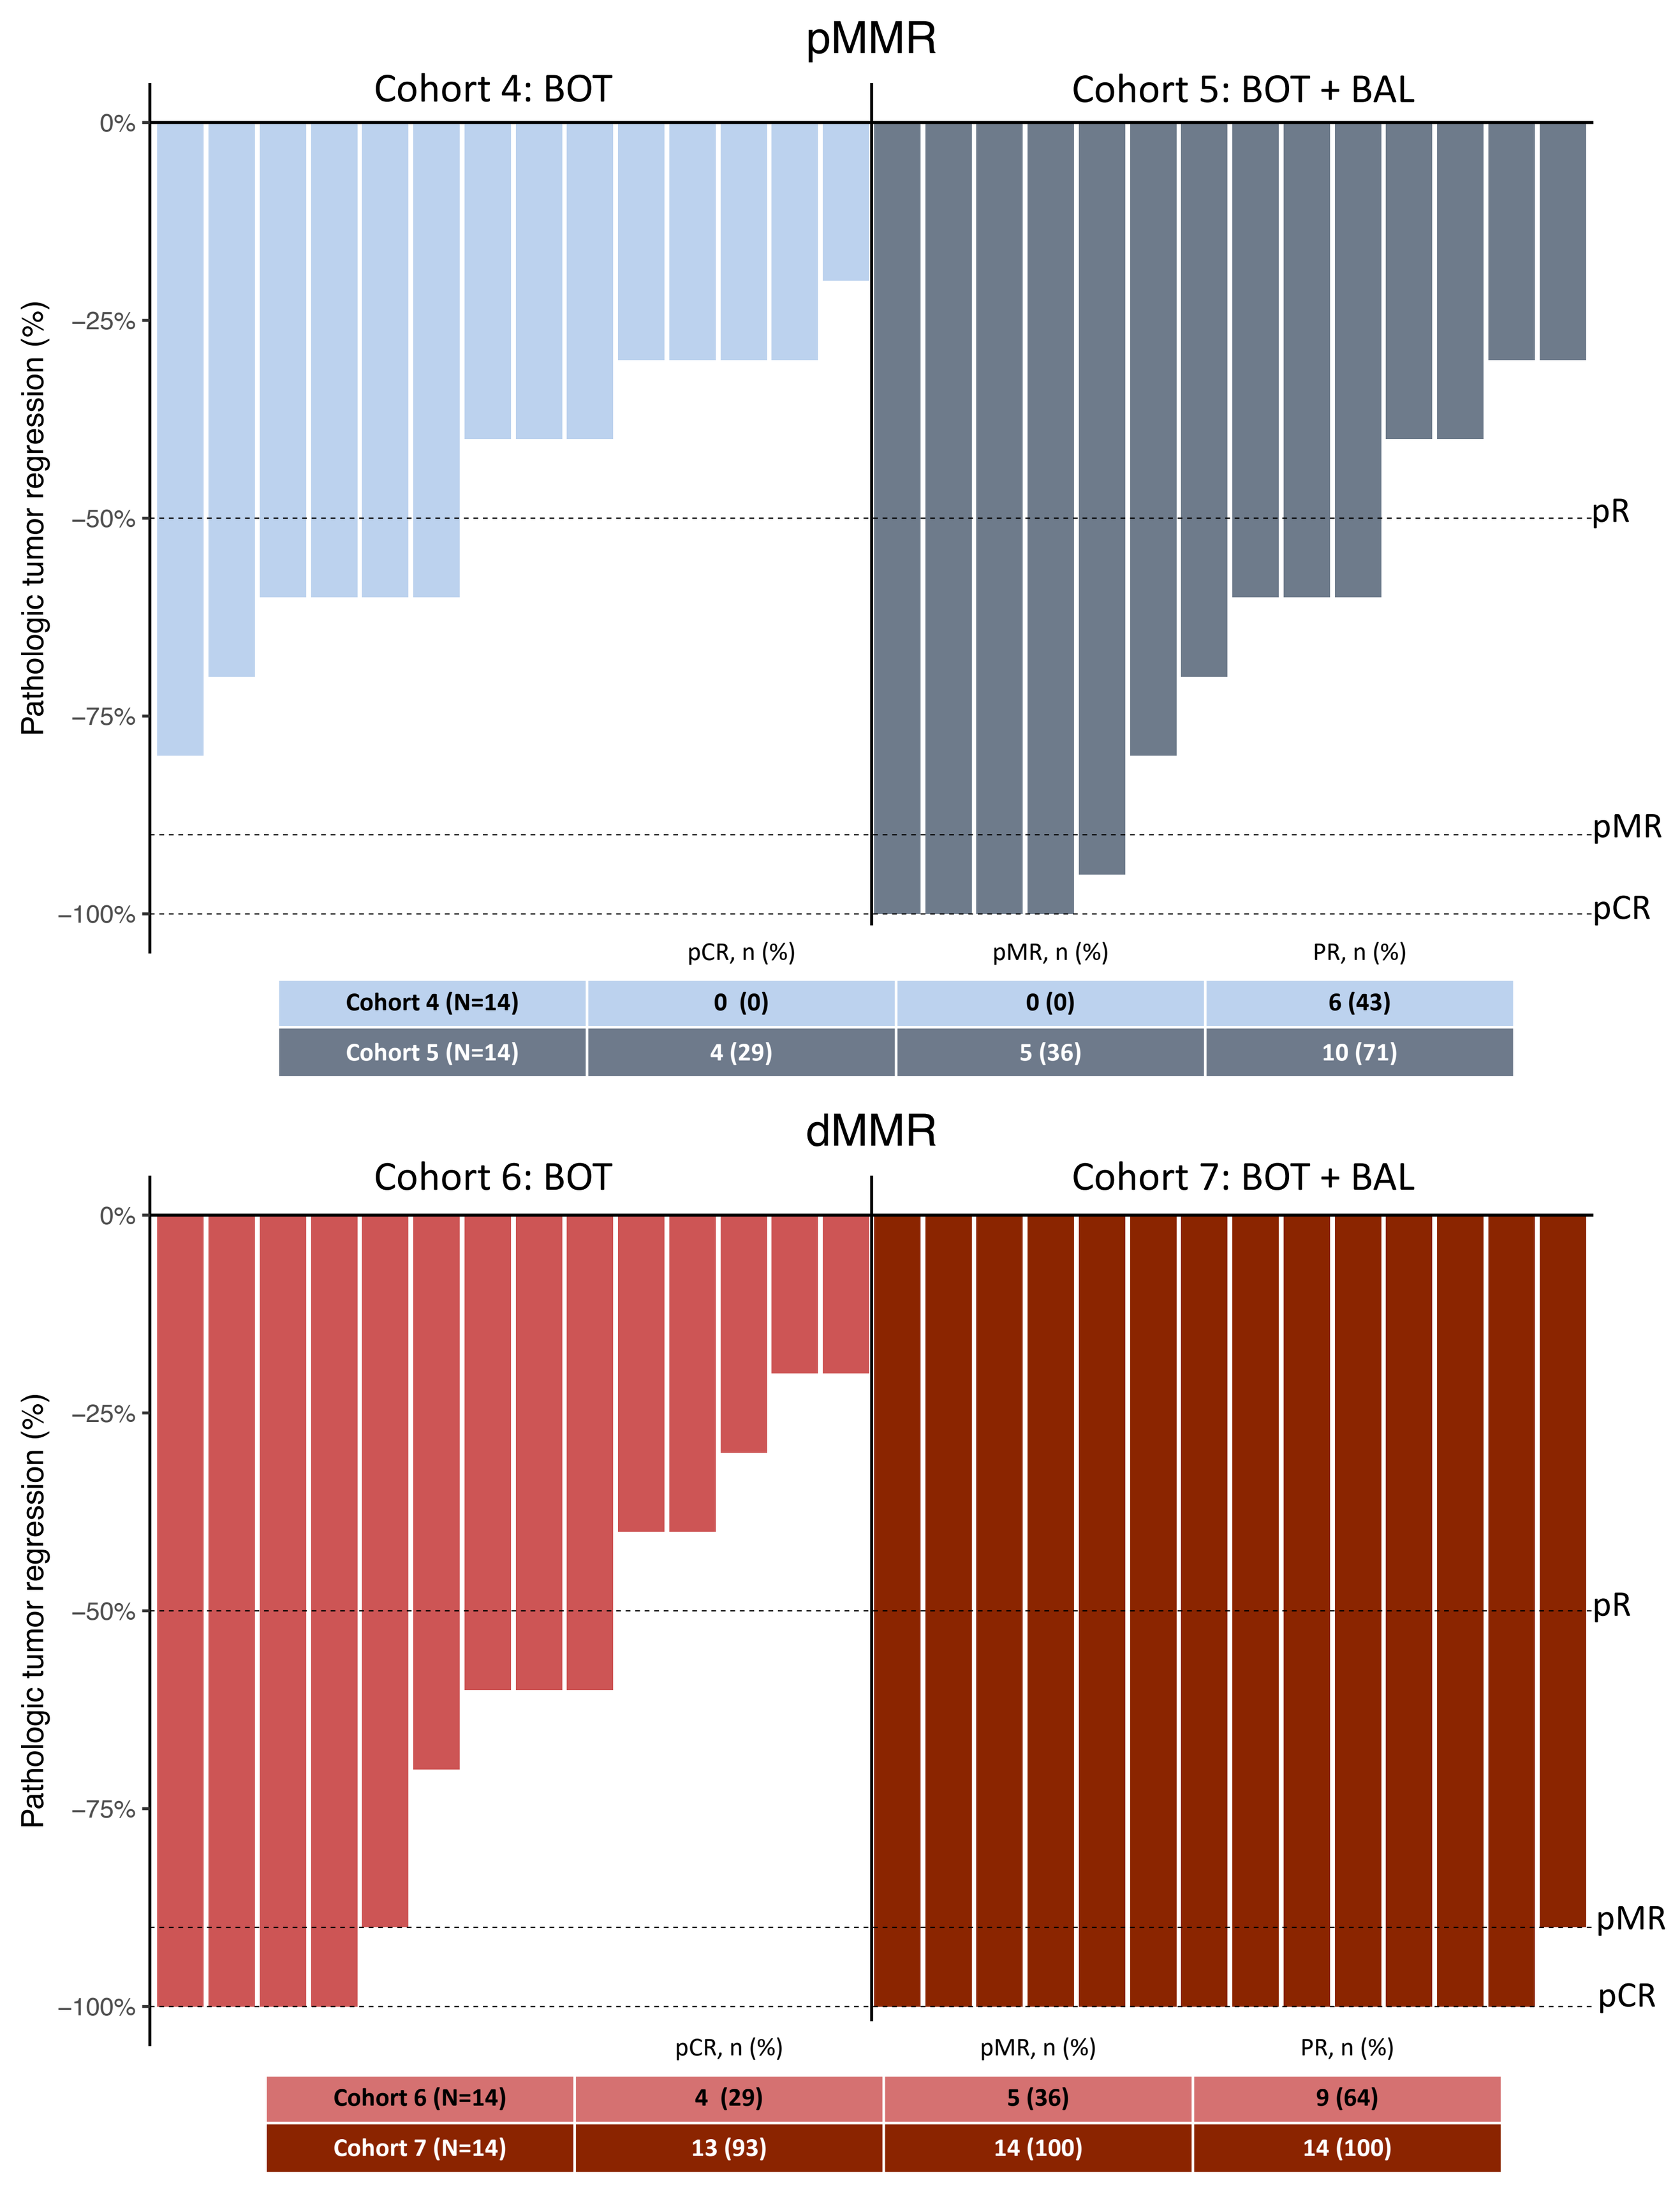

pMMR
Cohort 4: BOT
Cohort 5: BOT + BAL
Pathologic tumor regression (%)
0%
−25%
−50%
−75%
−100%
pR
pMR
pCR
pCR, n (%)
pMR, n (%)
PR, n (%)
Cohort 4 (N=14) | 0 (0) | 0 (0) | 6 (43)
Cohort 5 (N=14) | 4 (29) | 5 (36) | 10 (71)
dMMR
Cohort 6: BOT
Cohort 7: BOT + BAL
Cohort 6 (N=14) | 4 (29) | 5 (36) | 9 (64)
Cohort 7 (N=14) | 13 (93) | 14 (100) | 14 (100)

Fifty-six pts received study treatment between June 2023 and July 2024. At data cutoff date (January 2nd, 2025), median follow-up times were 13.2 (IQR 9.6-14.6), 8.6 (IQR 6.4-9.3), 11.2 (IQR 19.1-12.5), and 5.9 (IQR 5.0-7.3) months in cohorts 4 to 7, respectively. Across all cohorts, 4 progression events occurred, 1 in cohort 4 and 3 in cohort 6. No death occurred.
Specifically, as shown in **Figure 2**, 12-month PFS rate was 92% (95% CI, 79-100) in cohort 4, 70% (95% CI, 45-100) in cohort 6 and 100% in cohorts 5 and 7.

Figure 2. Kaplan-Meier curves for PFS in the four cohorts

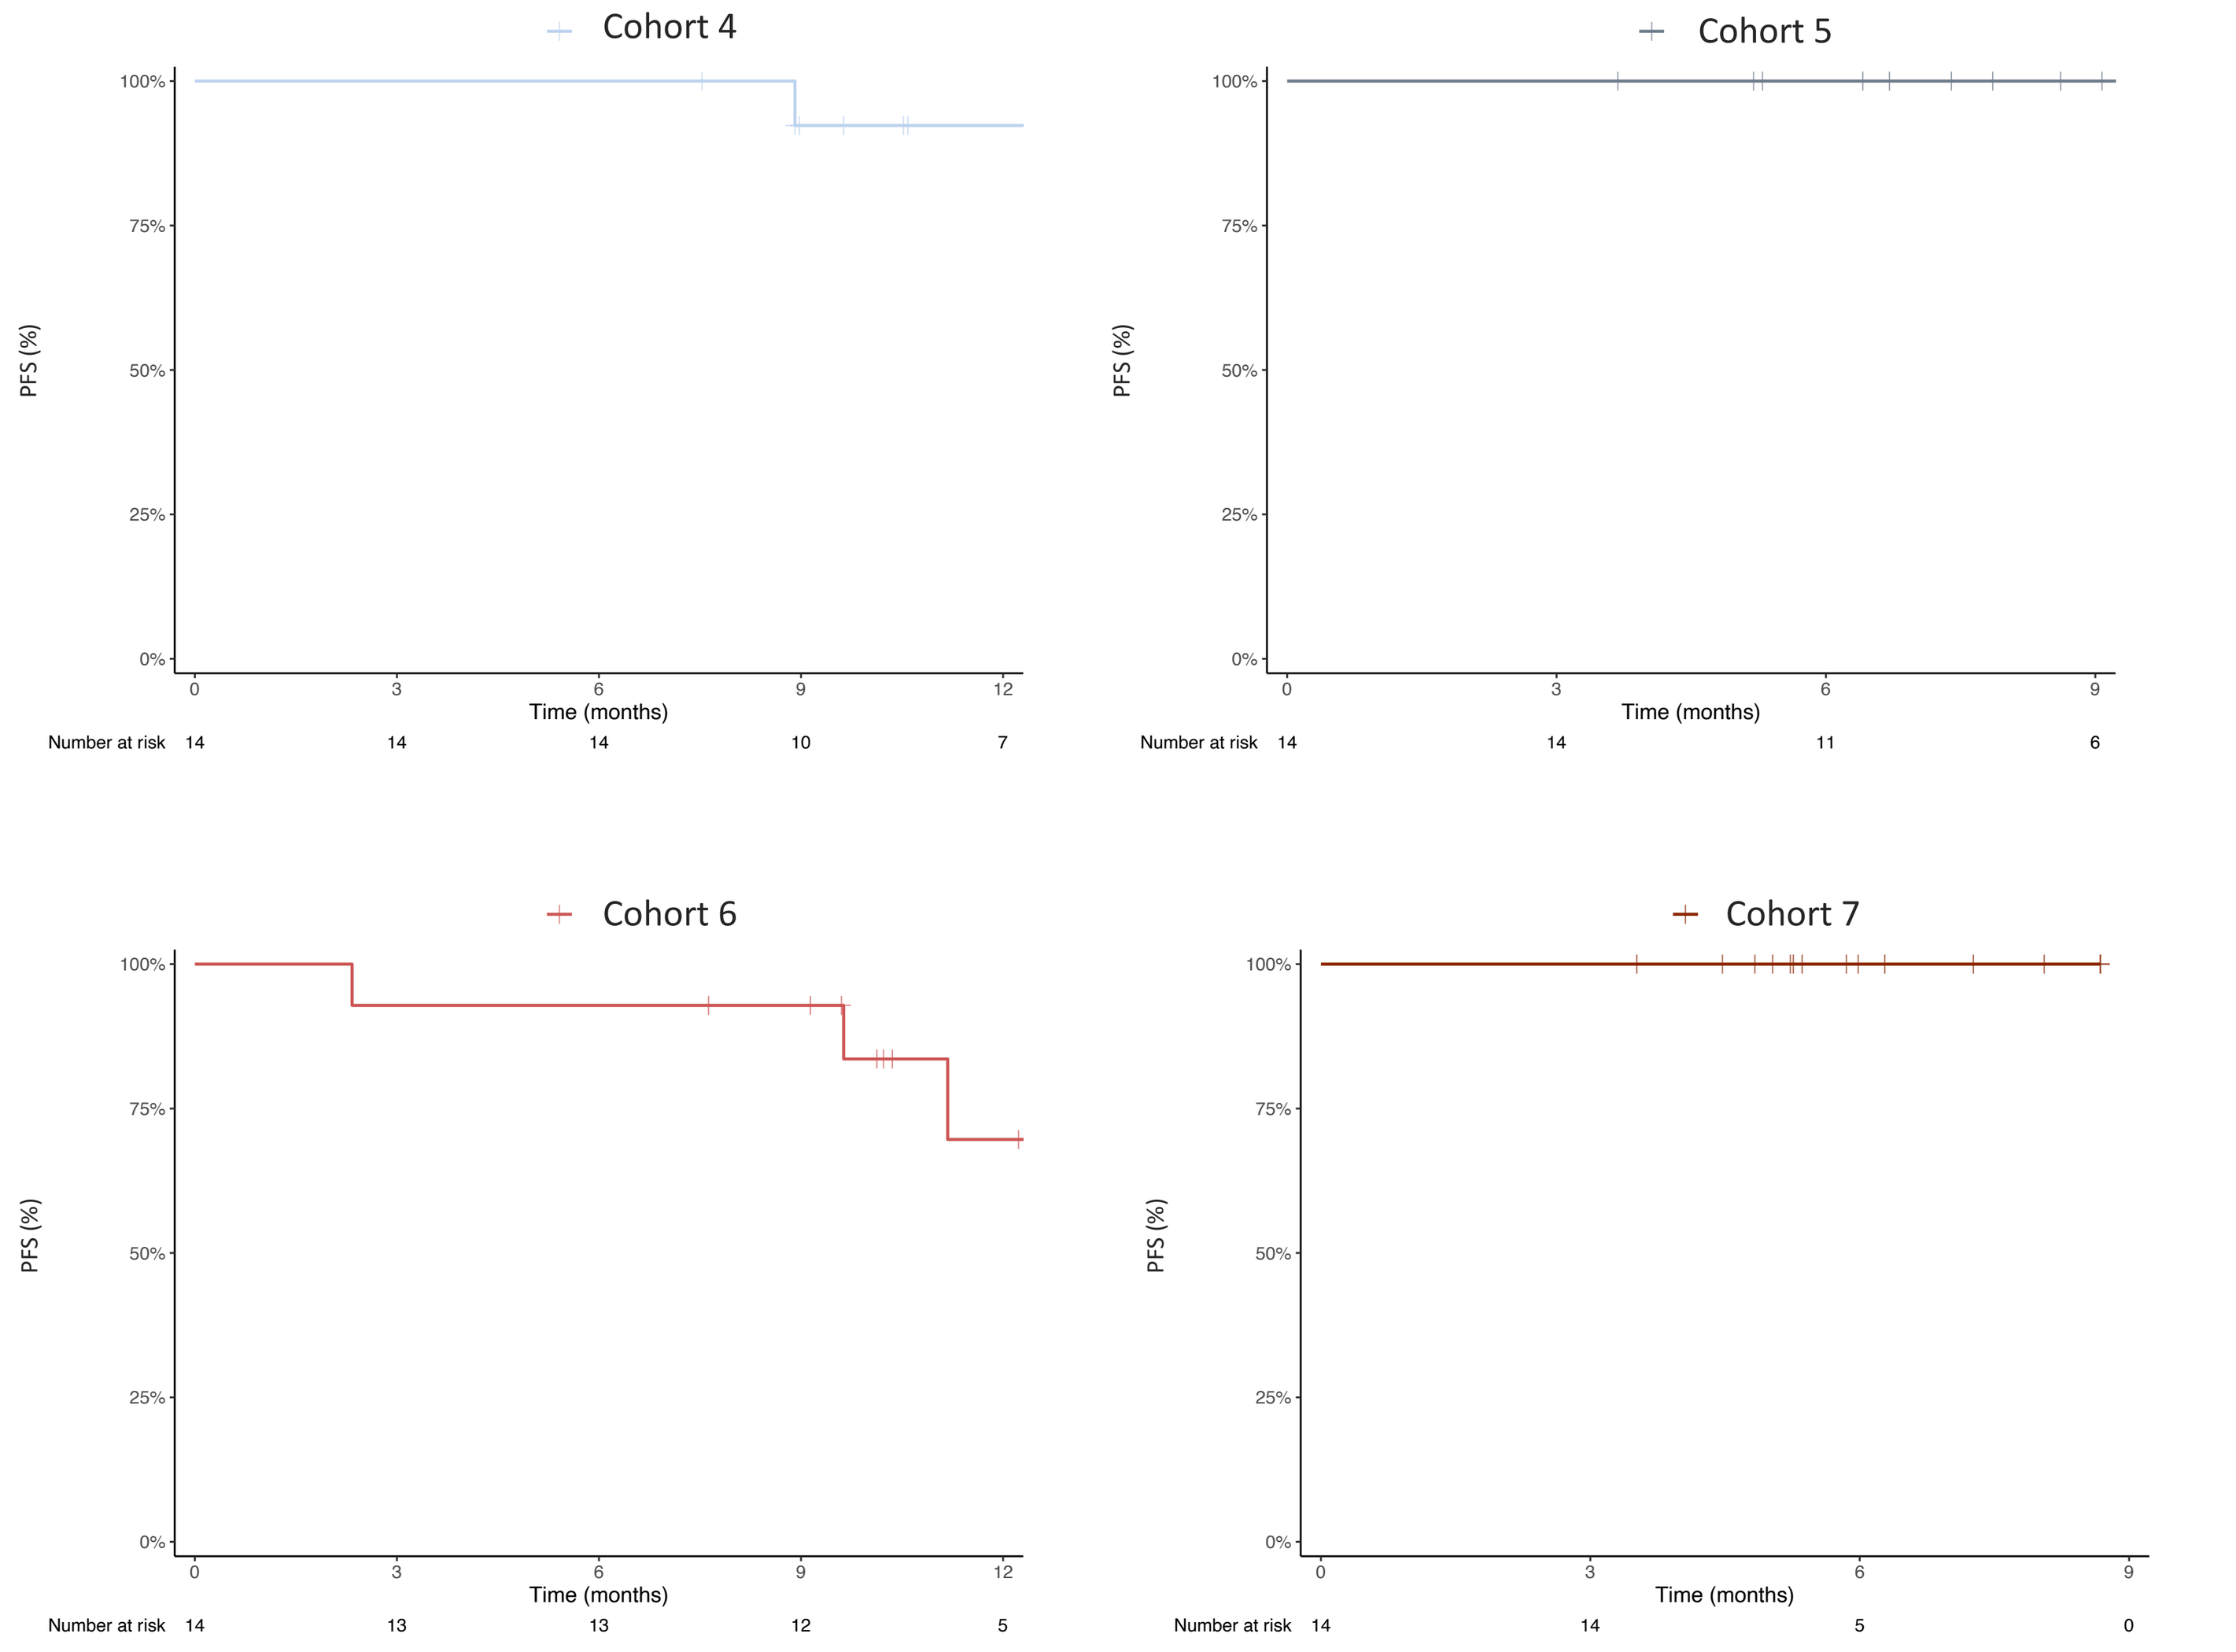

Cohort 4
Cohort 5
Cohort 6
Cohort 7
PFS (%)
Time (months)
Number at risk

CONCLUSIONS

The primary endpoint was met in all cohorts except for BOT monotherapy in pts with pMMR status, highlighting the specific contribution of BAL to the combination activity, which is in line with experience in the metastatic setting.
Despite a limited sample size, the activity of 1 neoadjuvant cycle with BOT and BAL favorably compares to ipilimumab and nivolumab in both pMMR and dMMR subgroups.
The pCR rate was remarkable in pMMR and the highest ever reported in dMMR pts, paving the way to NOM studies irrespective of MMR status.

We thank the patients, their families and caregivers. This investigator-initiated trial was partially funded by Agenus Inc, which also provided BOT and BAL supplies.