Poster #

219

Botensilimab (BOT) plus balstilimab (BAL) in microsatellite stable metastatic colorectal cancer: Assessing efficacy in non-liver metastatic sites

Marwan G. Fakih, MD[1], Andrea J. Bullock, MD, MPH[2], Benjamin L. Schlechter, MD[3], Apostolia M. Tsimberidou, MD, PhD[4], Wells Messersmith, MD[5], Agustin Pimentel, MD[6], Ani S. Balmanoukian, MD[7], Rachel E. Sanborn, MD[8], Neil H. Segal, MD, PhD[9,10], Bruno Bockorny, MD[2], Sunil Sharma, MD, FACP, MBA[11], Brian Henick, MD[12], Ian Chau, MD, FRCP[13], Joseph E. Grossman, MD[14], Jaymin M. Patel, MD[14], Wei Wu, MS[14], Benny Johnson, DO[14], Heinz-Josef Lenz, MD, FACP[15], Steven J. O'Day, MD[14,16], Anthony B. El-Khoueiry, MD[15]

[1]City of Hope Comprehensive Cancer Center, Duarte, CA, USA, [2]Beth Israel Deaconess Medical Center, Boston, MA, USA, [3]Dana-Farber Cancer Institute, Boston, MA, USA, [4]The University of Texas MD Anderson Cancer Center, Houston, TX, USA, [5]University of Colorado Cancer Center, Aurora, CO, USA, [6]Sylvester Comprehensive Cancer Center, University of Miami, Miami, FL, USA, [7]The Angeles Clinic and Research Institute, Los Angeles, CA, USA, [8]Earle A. Chiles Research Institute, Providence Cancer Institute, Portland, OR, USA, [9]Memorial Sloan Kettering Cancer Center, New York, NY, USA,[10]Weill Medical College at Cornell University, New York, USA, [11]HonorHealth Research Institute, Scottsdale, AZ, USA, [12]Columbia University Irving Medical Center, New York, NY, USA, [13]Royal Marsden Hospital, London, UK, [14]Agenus Inc., Lexington, MA, USA, [15]University of Southern California Norris Comprehensive Cancer Center, Los Angeles, CA, USA,[16]Providence Saint John's Cancer Institute, Santa Monica, CA, USA

BACKGROUND

- Botensilimab (BOT) is an Fc-enhanced, multifunctional anti-CTLA-4 antibody with differentiated mechanisms of action, designed to extend therapy to cold/poorly immunogenic solid tumors including microsatellite stable metastatic colorectal cancer (MSS mCRC; **Figure 1**)[1,2]
- Responses to novel immunotherapy (I-O) combinations in MSS mCRC have typically been restricted to non-liver metastatic (NLM) populations. Within this subgroup, responses outside of metastatic sites such as the lungs and lymph nodes are rare[3,4]
- We sought to determine whether BOT and balstilimab (BAL; anti–PD-1) could confer responses in NLM sites outside of the lungs and lymph nodes

Botensilimab Mechanism of Action

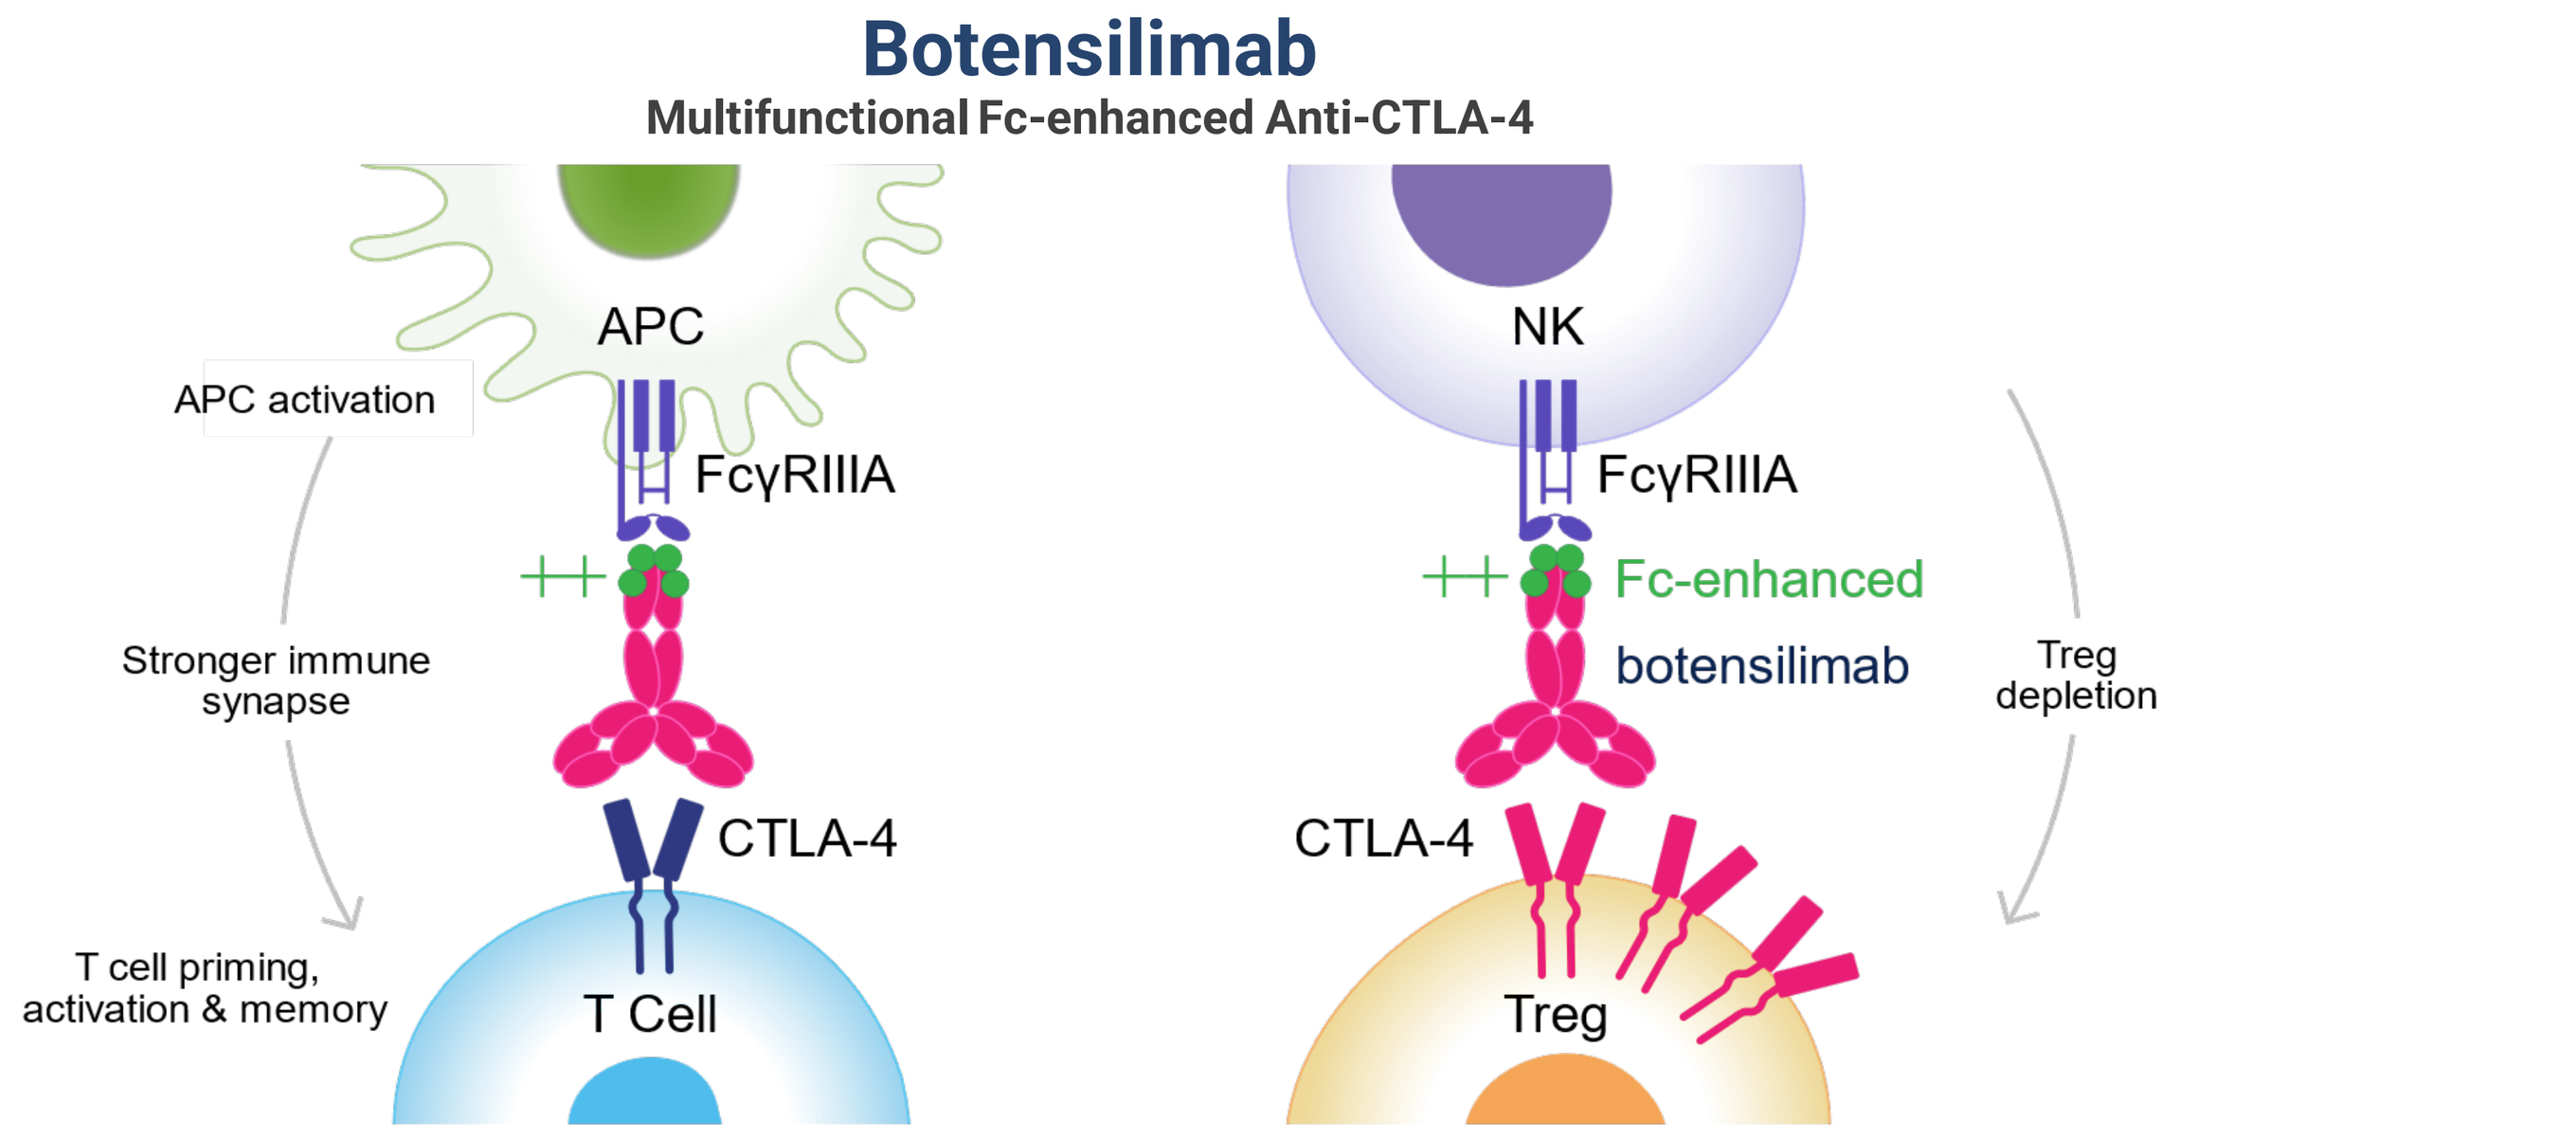
Botensilimab
Multifunctional Fc-enhanced Anti-CTLA-4
APC
NK
APC activation
FcγRIIIA
FcγRIIIA
Fc-enhanced
botensilimab
Stronger immune synapse
Treg depletion
CTLA-4
CTLA-4
T cell priming, activation & memory
T Cell
Treg

Figure 1. A novel innate and adaptive immune activator. BOT promotes enhanced T cell priming and expansion, T cell activation and memory formation, activation of antigen presenting cells, and reduction of intratumoral regulatory T cells, while improving safety through a reduction in complement-mediated toxicities[1,2]

C-800-01 Study Design: NLM MSS mCRC Cohort (N=77)

NCT0386027: First-in-human trial of **BOT ± BAL** in patients with advanced cancer[5]

Key Eligibility

- Relapsed/refractory mCRC
- MSS by local assessment
- Prior I-O allowed

Study Endpoints

- Efficacy: ORR, DCR, DOR, PFS, OS
- Safety: AEs

Treatment *(up to 2 years)*

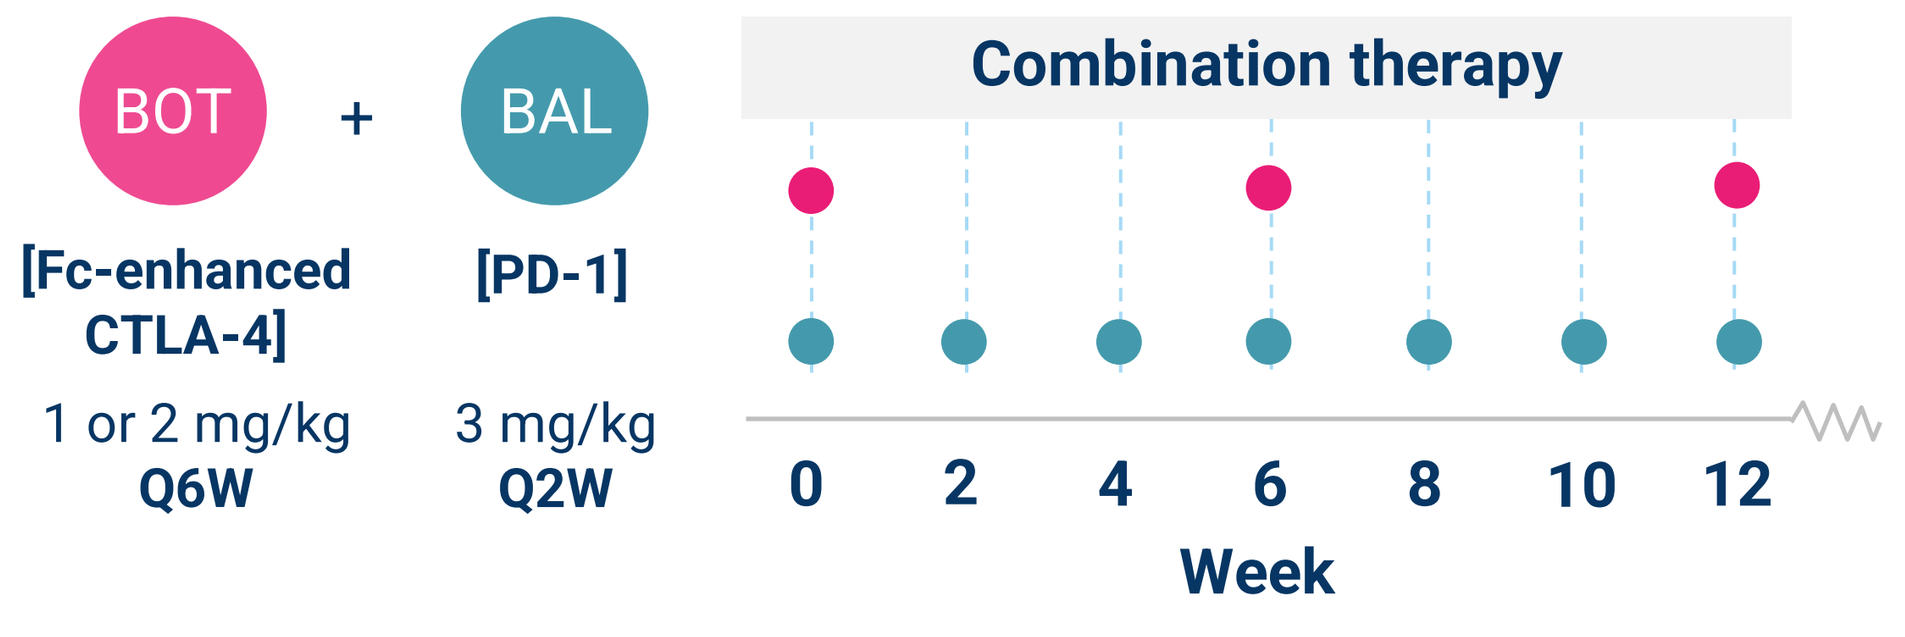
BOT
+
BAL
[Fc-enhanced CTLA-4]
[PD-1]
1 or 2 mg/kg Q6W
3 mg/kg Q2W
Combination therapy
0
2
4
6
8
10
12
Week

Patient Population

- *Safety Analysis Population* – **77** patients with NLM MSS mCRC treated with 1 or 2 mg/kg BOT Q6W plus 3 mg/kg BAL Q2W
- *Efficacy Evaluable Population* – **70** of these patients with ≥1 post-baseline 6-week imaging scan

References: 1. Chand et al. Submitted manuscript. 2. Bullock A et al. *Ann Oncol* 2023;34:S178–9. 3. Fakih M et al. *EClinicalMedicine* 2023; 58:101917. 4. Fakih M et al. *JAMA Oncol* 2023; 9: 627-634. 5. https://clinicaltrials.gov/ct2/show/NCT03860272.

Abbreviations: AE, adverse event; APC, antigen presenting cell; bal, balstilimab; bot, botensilimab; CR, complete response; CT, computed tomography; CTLA-4, cytotoxic T-lymphocyte associated protein 4; DCR, disease control rate (CR, PR, or SD ≥6 weeks); DOR, duration of response; ECOG, Eastern Cooperative Oncology Group; Fc, fragment crystallizable; FcγR, fragment crystallizable gamma receptor; H&E, hematoxylin and eosin; I-O, immunotherapy; mCRC, metastatic colorectal cancer; MSS, microsatellite stable; MRI, magnetic resonance imaging; NK, natural killer; NLM, no active liver metastases; NR, not reached; ORR, objective response rate; OS, overall survival; PFS, progression free survival; PD, progressive disease; PD-1, programmed cell death protein 1 ; PR, partial response; PS, performance status; Q[X]W, every X weeks ; SD, stable disease; TMB, tumor mutational burden; Treg, regulatory T cell.

Correspondence: mfakih@coh.org

RESULTS

Baseline Characteristics

	N=77
Median Age, years (range)	56 (36–82)
Sex, n (%)	
Male	37 (48%)
Female	40 (52%)
Race, n (%)	
White	59 (77%)
Black	1 (1%)
Asian	10 (13%)
Other or not specified	7 (9%)
Ethnicity, n (%)	
Hispanic or Latino	13 (17%)
Not Hispanic or Latino	61 (79%)
Not specified	3 (4%)
ECOG PS, n (%)	
0	32 (42%)
1	45 (58%)
Prior Lines of Therapy, n (%)	
Median (range)	4 (2–6)
≥3	56 (73%)
Prior PD-(L)1/CTLA-4, n (%)	16 (21%)
Prior regorafenib, n (%)	17 (22%)
Prior trifluridine/tipiracil, n (%)	12 (16%)
Prior bevacizumab, n (%)	65 (84%)

	N=77
TMB >10, Mut/Mb, n/N (%)[a]	2/39 (5%)
RAS mutation, n/N (%)	47/72 (65%)
BRAF mutation, n/N (%)	2/39 (5%)
Time from metastatic disease to first dose of study drug, months	
Median (range)	36.7 (0.2–179.4)
≥18 months, n (%)	59 (77%)
<18 months, n (%)	18 (23%)
Location of metastases, n (%)	
Lungs	62 (81%)
Peritoneal disease	33 (43%)
Lymph nodes	32 (42%)
Soft tissue	15 (19%)
Other[b]	18 (23%)
Bone	3 (4%)
Brain	2 (3%)
Metastatic sites, n (%)	
1 organ	28 (36%)
Lung only	22 (29%)
Lymph nodes only	2 (3%)
Peritoneal disease only	4 (5%)
≥2 organs	49 (64%)

Data cutoff: 01-MAR-2024
[a]Only two patients had a TMB >10 Mut/Mb and <13.
[b]Other organ involvement includes adrenal glands, bone, brain, kidney, pleura, retroperitoneum, and spleen.

Table 1. Baseline demographics and patient characteristics.

Efficacy

	Number of patients	ORR, % (n/nn)	DCR, % (n/nn)	Median OS, months (95% CI)
NLM MSS mCRC (efficacy evaluable population)	70	26% (18/70)	80% (56/70)	**NR** (20.7–NR)
NLM MSS mCRC (safety analysis population)	77	23% (18/77)	73% (56/77)	**21.2** (16.5–NR)
Any lung involvement	62	26% (16/62)	74% (46/62)	**21.2** (20.7–NR)
Lung only	22	18% (4/22)	82% (18/22)	**NR** (13.3–NR)
Any peritoneal involvement	33	18% (6/33)	67% (22/33)	**20.7** (6.3–NR)
Any soft tissue involvement	15	27% (4/15)	73% (11/15)	**20.7** (3.6–NR)
Any other organ involvement	18	33% (6/18)	72% (13/18)	**20.9** (6.3–NR)

Data cutoff: 01-MAR-2024
Median follow-up for NLM MSS mCRC (safety analysis population) patients (N=77) was 13.6 months (range, 0.6–41.8 months), with 11/18 responses ongoing/censored.

Table 2. Efficacy outcomes by sites of metastatic disease in patients with NLM MSS mCRC.

- **Across different NLM sites**:
 - Response rates ranged from **18–33%**
 - Disease control rate ranged from **67–82%**
 - Median OS remained consistent and ranged from **20.7 months to not reached (NR)**

Case Study: Radiologic Growth of an Occult Brain Metastasis With a Pathologic Response and No Recurrence

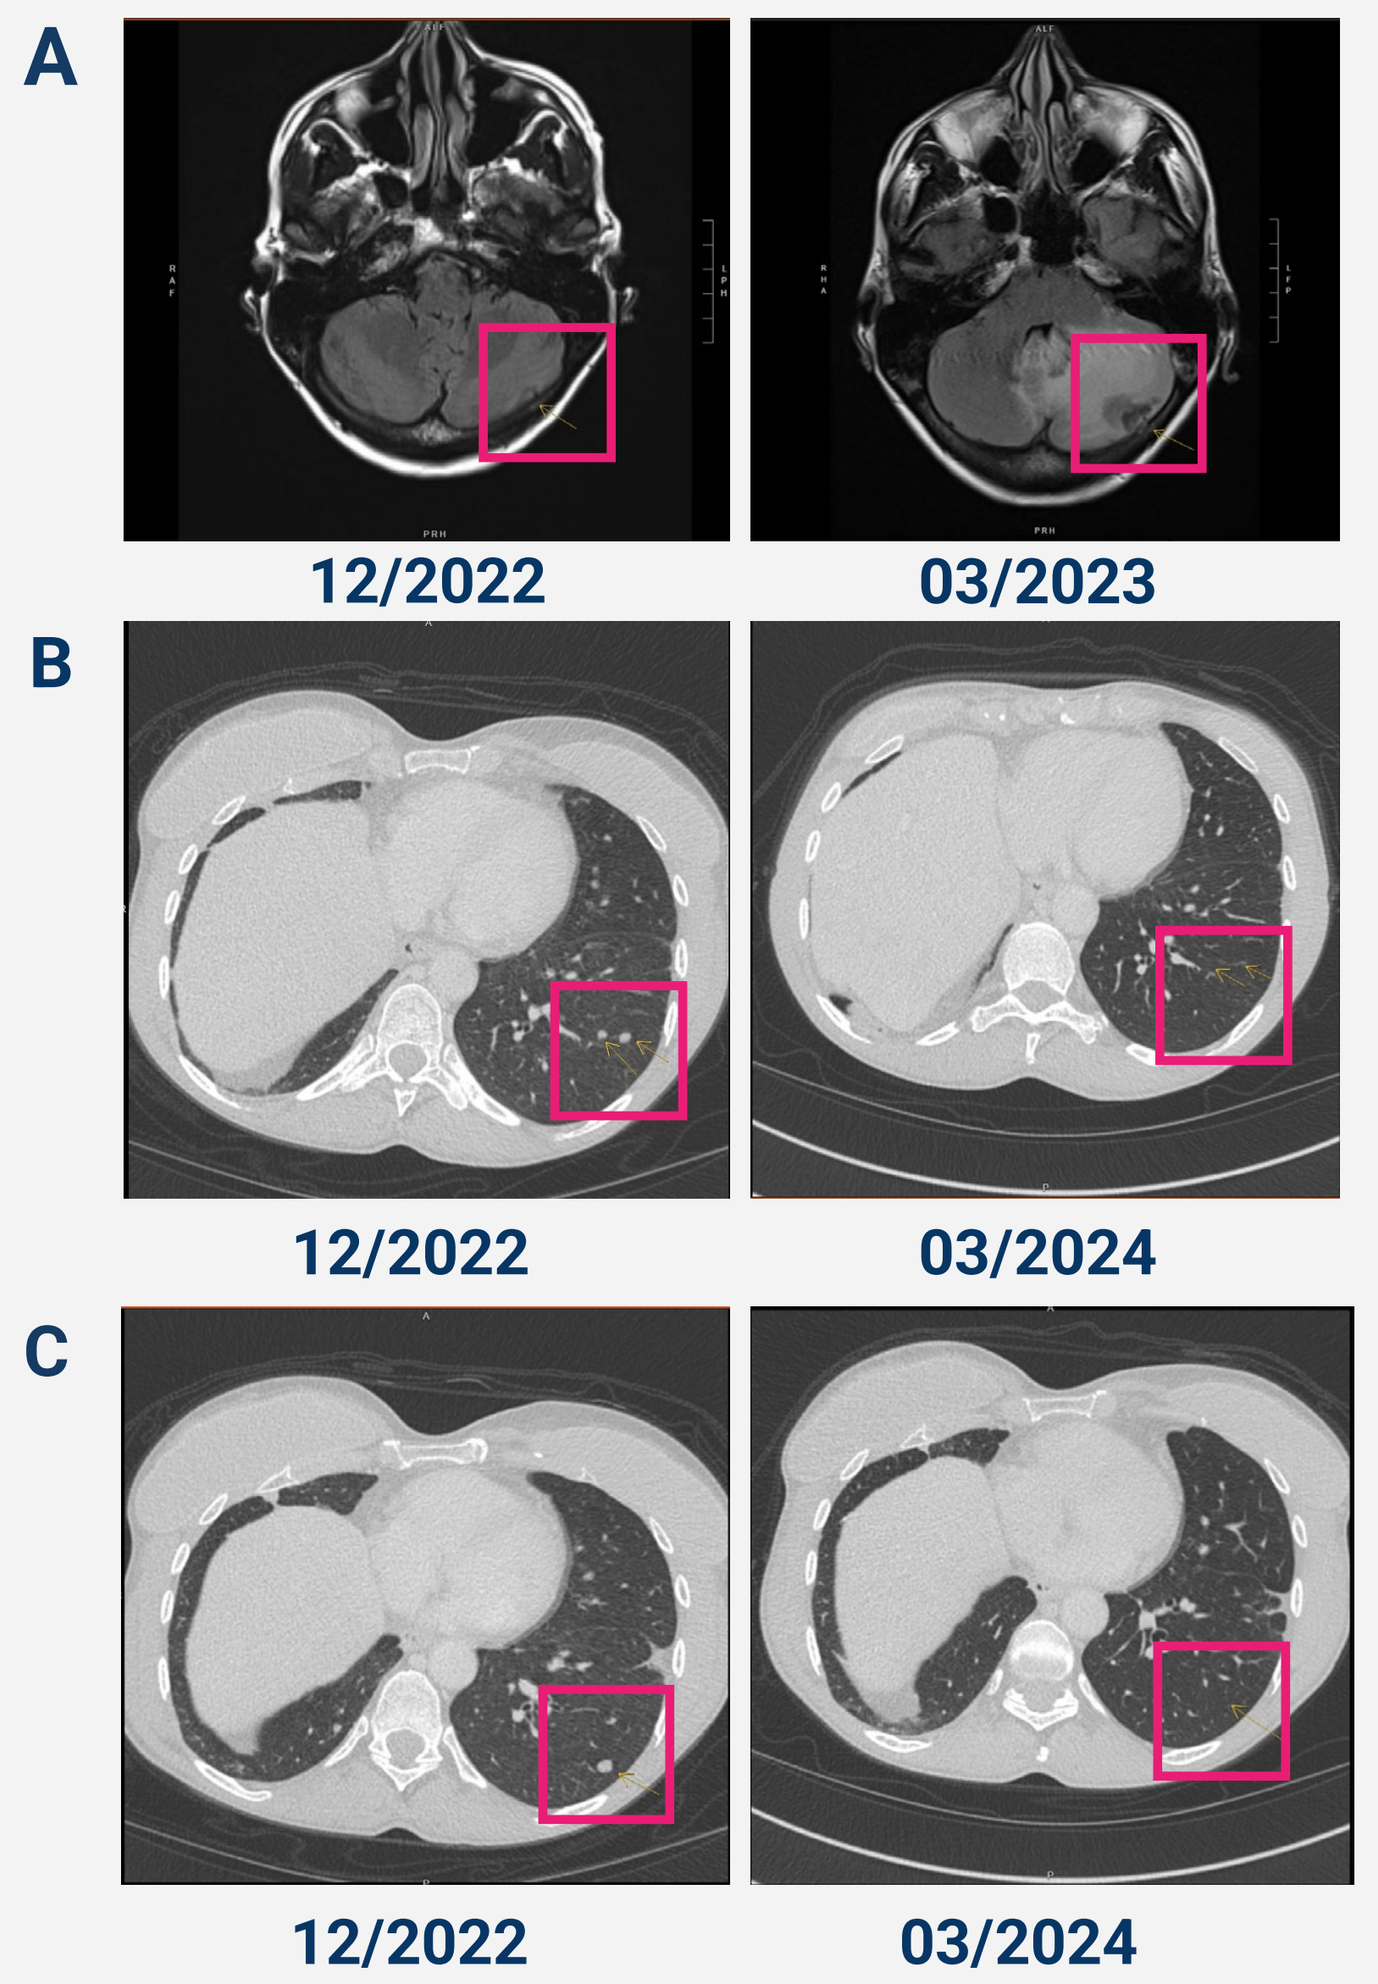
A
12/2022
03/2023
B
12/2022
03/2024
C
12/2022
03/2024

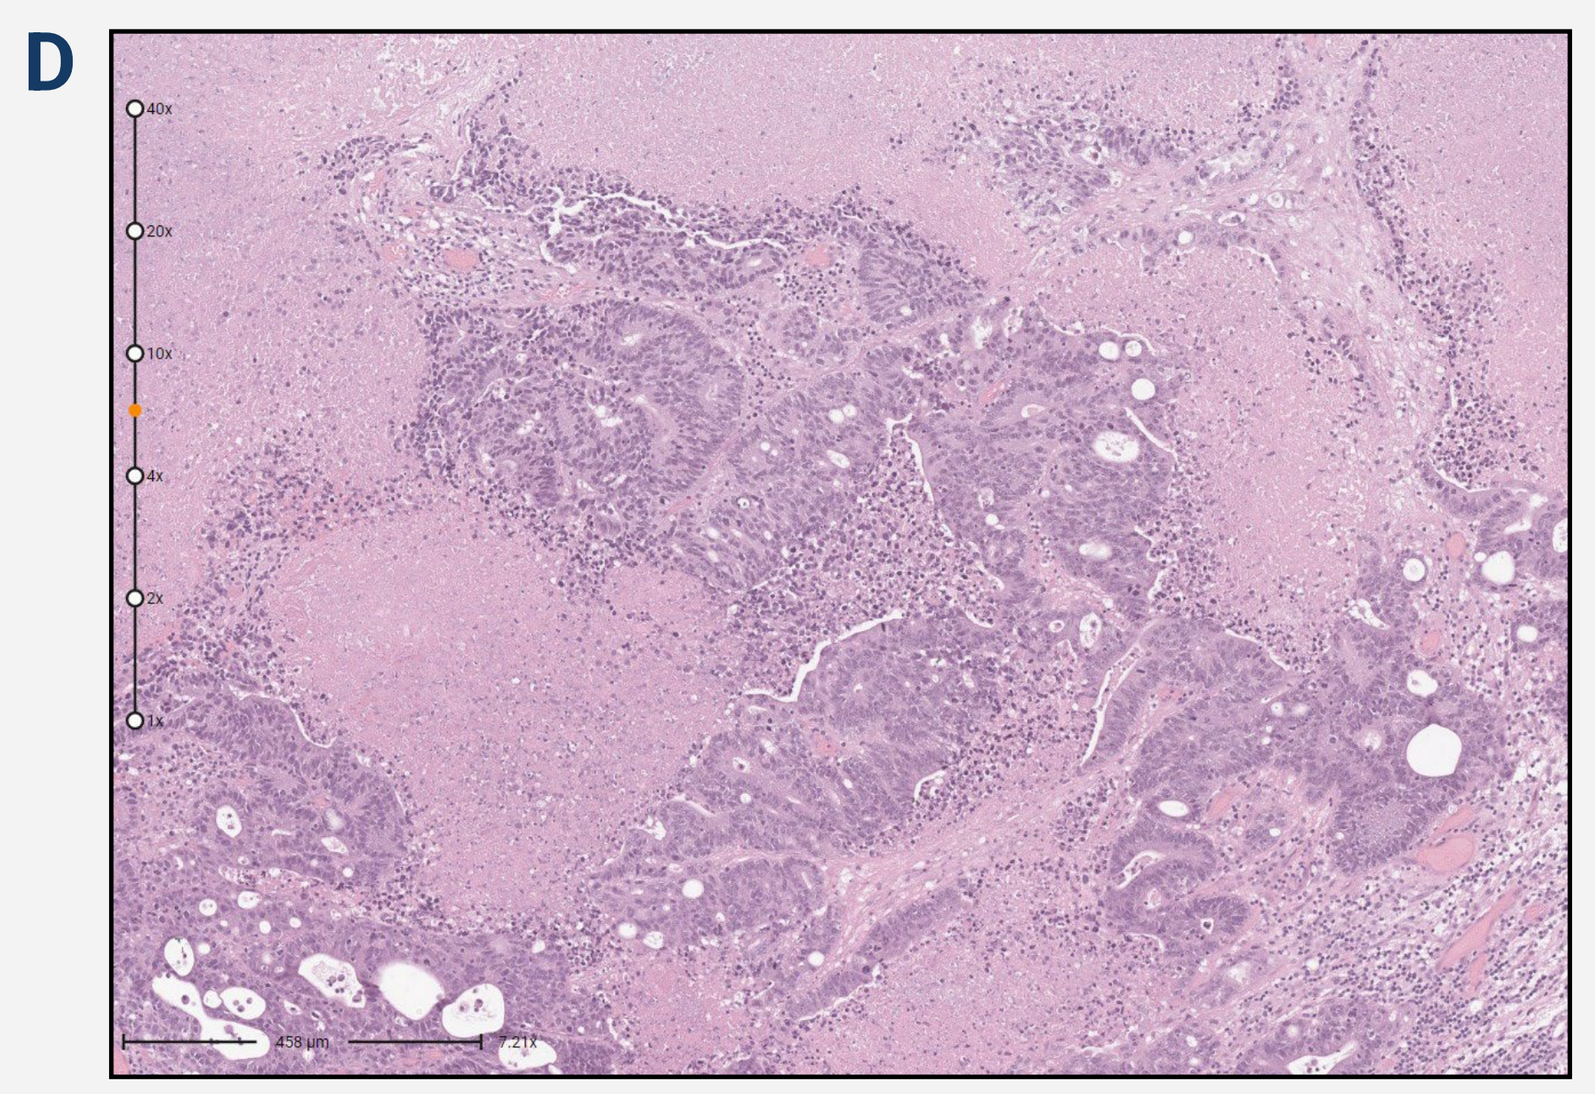
D

Figure 2. Patient scans at baseline and follow-up in a patient with an occult brain metastasis. (A) Screening (left, 12/2022) and an urgent MRI showing growth of an occult brain metastasis ~3 months later (right, 03/2023). (B-C) Chest CT at screening (left, 12/2022) versus follow-up 2 years later (right, 03/2024) showing resolution of target lesions in the lung. (D) H&E stain of the resected brain metastasis showing necrosis and lymphocytic infiltration consistent with an immune response, alongside scant residual cancer cells. As of 4/2024, this patient was still under observation, with an ongoing PR in lung lesions.

Safety Overview

- No new safety signals
- Safety in CRC consistent across tumor types in study
- No treatment-related deaths

CONCLUSIONS

- In patients with NLM MSS mCRC, response rates were comparable across different sites of metastatic disease, including historically poor prognostic sites such as the peritoneum, soft tissue, pleura and brain
- This broader clinical activity outside the lungs and lymph nodes has not previously been reported with other IO and IO/non-IO combinations
- A global randomized phase 2 trial of BOT ± BAL (versus standard of care) in MSS mCRC is fully enrolled (NCT05608044) and a global phase 3 trial is planned

Acknowledgments

- Agenus Inc. funded and is the sponsor of this study.
- The authors would like to thank the patients and their families for participating in the C-800-01 study, as well as the trial coordinators and investigators for their contributions.

Disclosures

MGF: consulting fees from AstraZeneca, Bayer, Bristol Myers Squibb, Incyte, Mirati, Pfizer, and Taiho, and data safety monitoring/advisory board for Bayer Corp, Eisai, Entos, Janssen, Merck, Mirati Therapeutics, Nouscom, Seattle Genetics, and Xenthera. AJB: research funding from Agenus Inc., AstraZeneca, Geistlich Pharma, Oncomatryx Biopharma , and Panavance Therapeutics, travel support from Agenus Inc., and consulting/advisory fees from Exelixis, Merck, and Oncolytics. WM: consulting/advisory Role at QED Therapeutics. RES: research funding from AstraZeneca and Merck, consulting fees from Amgen, AstraZeneca, Daiichi Sankyo, GE HealthCare, G1 Therapeutics, Gilead Sciences, GlaxoSmithKline, Janssen, Regeneron, and Sanofi Aventis, honoraria from Binay Foundation, EMD Serono, GameOn!, Illumina, OncLive, and Targeted Oncology. NHS: consulting fees from ABL Bio, Agenus Inc., AstraZeneca, Boehringer Ingelheim, GSK, Novartis, Numab, Puretech, Revitope, and Roche/Genentech, support for attending meetings and/or travel from AstraZeneca and Regeneron, and research funding from Agenus Inc., AstraZeneca, Bristol-Myers Squibb, Immunocore, Merck, Pfizer, Puretech, Regeneron, and Roche/Genentech. BB: research funding from Agenus Inc, and NanoView Biosciences, consulting fees from BioLineRx, received support for attending meetings and/or travel from Agenus Inc. and Erytech Pharma; data safety monitoring board for Blueprint Medicines and Enlivex Therapeutics. SS: honoraria from Array BioPharma, consulting/advisory role for Barricade Therapeutics, Celularity, Dracen Pharmaceuticals, and Elevar Therapeutics, research funding from Aadi Bioscience Inc., Amal Therapeutics, Celgene, Dracen Pharmaceuticals, HonorHealth, Inhibrx, Merck, Nektar Therapeutics, Novartis, Plexxikon, Sirnaomics, Syndax Pharmaceuticals, Takeda, Tesaro, Toray Industries, and Zai Lab, stock and other ownership interests with Barricade Therapeutics, Beta Cat Pharmaceuticals, ConverGene, Elevar Therapeutics, HLB, Salarius Pharmaceuticals, and Stingray Therapeutics. BH: honoraria from OncLive/MJH Life Sciences, DAVA Oncology, consulting/advisory role for AstraZeneca, IDEAYA Biosciences, Jazz Pharmaceuticals, Sorrento Therapeutics, Genentech/Roche, and Regeneron. IC: Honoria from Bristol-Myers Squibb, Eisai, Lilly, Novartis, Roche/Genentech, SERVIER, consulting/advisory role for Astellas Pharma, AstraZeneca, BioNTech SE, Bristol-Myers Squibb, Daiichi Sankyo/Astra Zeneca, Eisai, GlaxoSmithKline, Lilly, Merck Serono, MSD Oncology, OncXerna Therapeutics, Roche/Genentech, Seagen, SERVIER, Sotio, Taiho Oncology, Turning Point Therapeutics, travel and expenses from Bristol-Myers Squibb, and SERVIER. H-JL: consulting fees from Janssen and Pfizer, honoraria from AstraZeneca, Bristol-Myers Squibb, Genentech, Merck, Mirati, and Regeneron. ABE: honoraria/consulting/advisory role for Agenus Inc., AstraZeneca/MedImmune, Bayer, Bristol Myers Squibb, Eisai, EMD Serono, Exelixis, Gilead Sciences, Merck, QED Therapeutics, Qurient, Roche/Genentech, Senti Biosciences, Servier Laboratories, and Tallac Therapeutics, research funding from Astex Pharmaceuticals, AstraZeneca, and Fulgent Genetics. JEG, JP, WW, BJ, and SJO: Employees of Agenus Inc. with stock/stock options. JP and JEG: Leadership position at Agenus Inc. SJO: Officer at Agenus Inc. The other authors declare no competing interests (AP, ASB, and BLS).