P291

agenus

Agenus Inc. or subsidiary thereof (current or former employee), Lexington, MA 02421, USA

Driving T cell dysfunction in vitro for rational immunotherapy design

Cailin E. Joyce, Matthew Hancock, Thomas Horn, Simarjot Pabla, Benjamin Duckless, Andrew Basinski, Dhan Chand, Jeremy D. Waight, Mariana Manrique, Tyler J. Curiel, Nicholas S. Wilson, Alex Duncan, Jennifer S. Buell, David A. Savitsky, Lukasz Swiech, John C. Castle

Background

Immune checkpoint blockade (ICB) elicits durable responses in some cancer patients, but novel targets and combination approaches are needed to address resistance and broaden clinical benefit. Agenus is addressing this need with our Virtual Systems for Immuno-Oncology (VISION) platform. VISION is based on a collection of *in vitro* ecosystems which we deeply and systematically interrogate to discover novel targets, optimize our therapies and design smarter clinical trials. Here, we present an illustrative example of a VISION system modeling T cell dysfunction in a tumor microenvironment (TME)-like context for development of novel approaches to ICB resistance.

Agenus VISION platform supports smart, streamlined drug discovery and development

a VISION

CAPABILITIES

In vitro ecosystems
- T cell dysfunction
- T cell priming
- Immunosuppression
- T cell infiltration

Systemic perturbatinos
- Antibodies
- Multispecifics
- Fc fusions
- Gene editing

Computational framework
- In-house algorithms
- Integration with clinical data
- Growing AI capabilities

APPLICATIONS

Target discovery
- T cell targets
- Myeloid targets
- TME conditioners
- Cancer cell targets

Therapeutic optimization
- Biologics
- Cell therapies
- Small molecules

Smart trials
- Optimized combinations
- Predictive biomarkers
- Molecular indications

Methods

We developed a long-term human co-culture system comprised of primary T cells and cancer cells that enables controlled differentiation of naïve T cells to effector, memory and dysfunctional states. We longitudinally monitored T cell effector functions, protein and RNA expression across states and single cells. Finally, we challenged the system with anti-PD-1 antibody to uncover biomarkers and mechanisms of therapeutic resistance.

Results

T cells in our system become activated and then gradually progress to a terminally dysfunctional state driven by multiple cancer antigen exposures. T cell cytotoxicity is maintained over several antigen exposures before sharply decreasing whereas cytokine secretion begins to decrease with only one prior antigen exposure. The expression of known T cell regulators and novel factors is altered over the time course, with known factors reflecting previous observations *in vivo*. Anti-PD-1 antibody prolongs cytotoxic capacity but T cells eventually fail to respond. Single cell mapping in the presence of anti-anti-PD-1 antibody reveals an expanded population of T cells that co-expresses PD-1, TIGIT and activation markers. Consistent with this, the combination of PD-1 and TIGIT blockade enhances cytotoxicity relative to monotherapies.

Conclusions

These findings demonstrate the potential of VISION systems to deeply interrogate response and resistance to current and next-generation I-O therapies. In this case, we have applied a physiologically-relevant model of intratumoral T cell dysfunction to define a predictive biomarker signature for anti-PD-1 response and discover potential combination partners predicted to enhance durability of anti-PD-1 responses. With a growing repertoire of virtual systems and capabilities, VISION is poised to advance Agenus' multi-faceted approach to fighting cancer with immunotherapy.

Disclosures

Cailin E. Joyce, Matthew Hancock, Thomas Horn, Simarjot Pabla, Benjamin Duckless, Andrew Basinski, Dhan Chand, Jeremy D. Waight, Mariana Manrique, Tyler J. Curiel, Nicholas S. Wilson, Alex Duncan, Jennifer S. Buell, David A. Savitsky, Lukasz Swiech, John C. Castle – an: Agenus Inc.: current or former employment/consultancy and stock ownership

Correspondence

Cailin E Joyce cailin.joyce@agenusbio.com, John Castle john.castle@agenusbio.com

Presented at The Society for Immunotherapy of Cancer 34th Annual Conference (SITC), November 6-10, 2019, in National Harbor, Maryland United States

Figure 1. Chronic cancer antigen exposure drives CD8+ T cell dysfunction in vitro

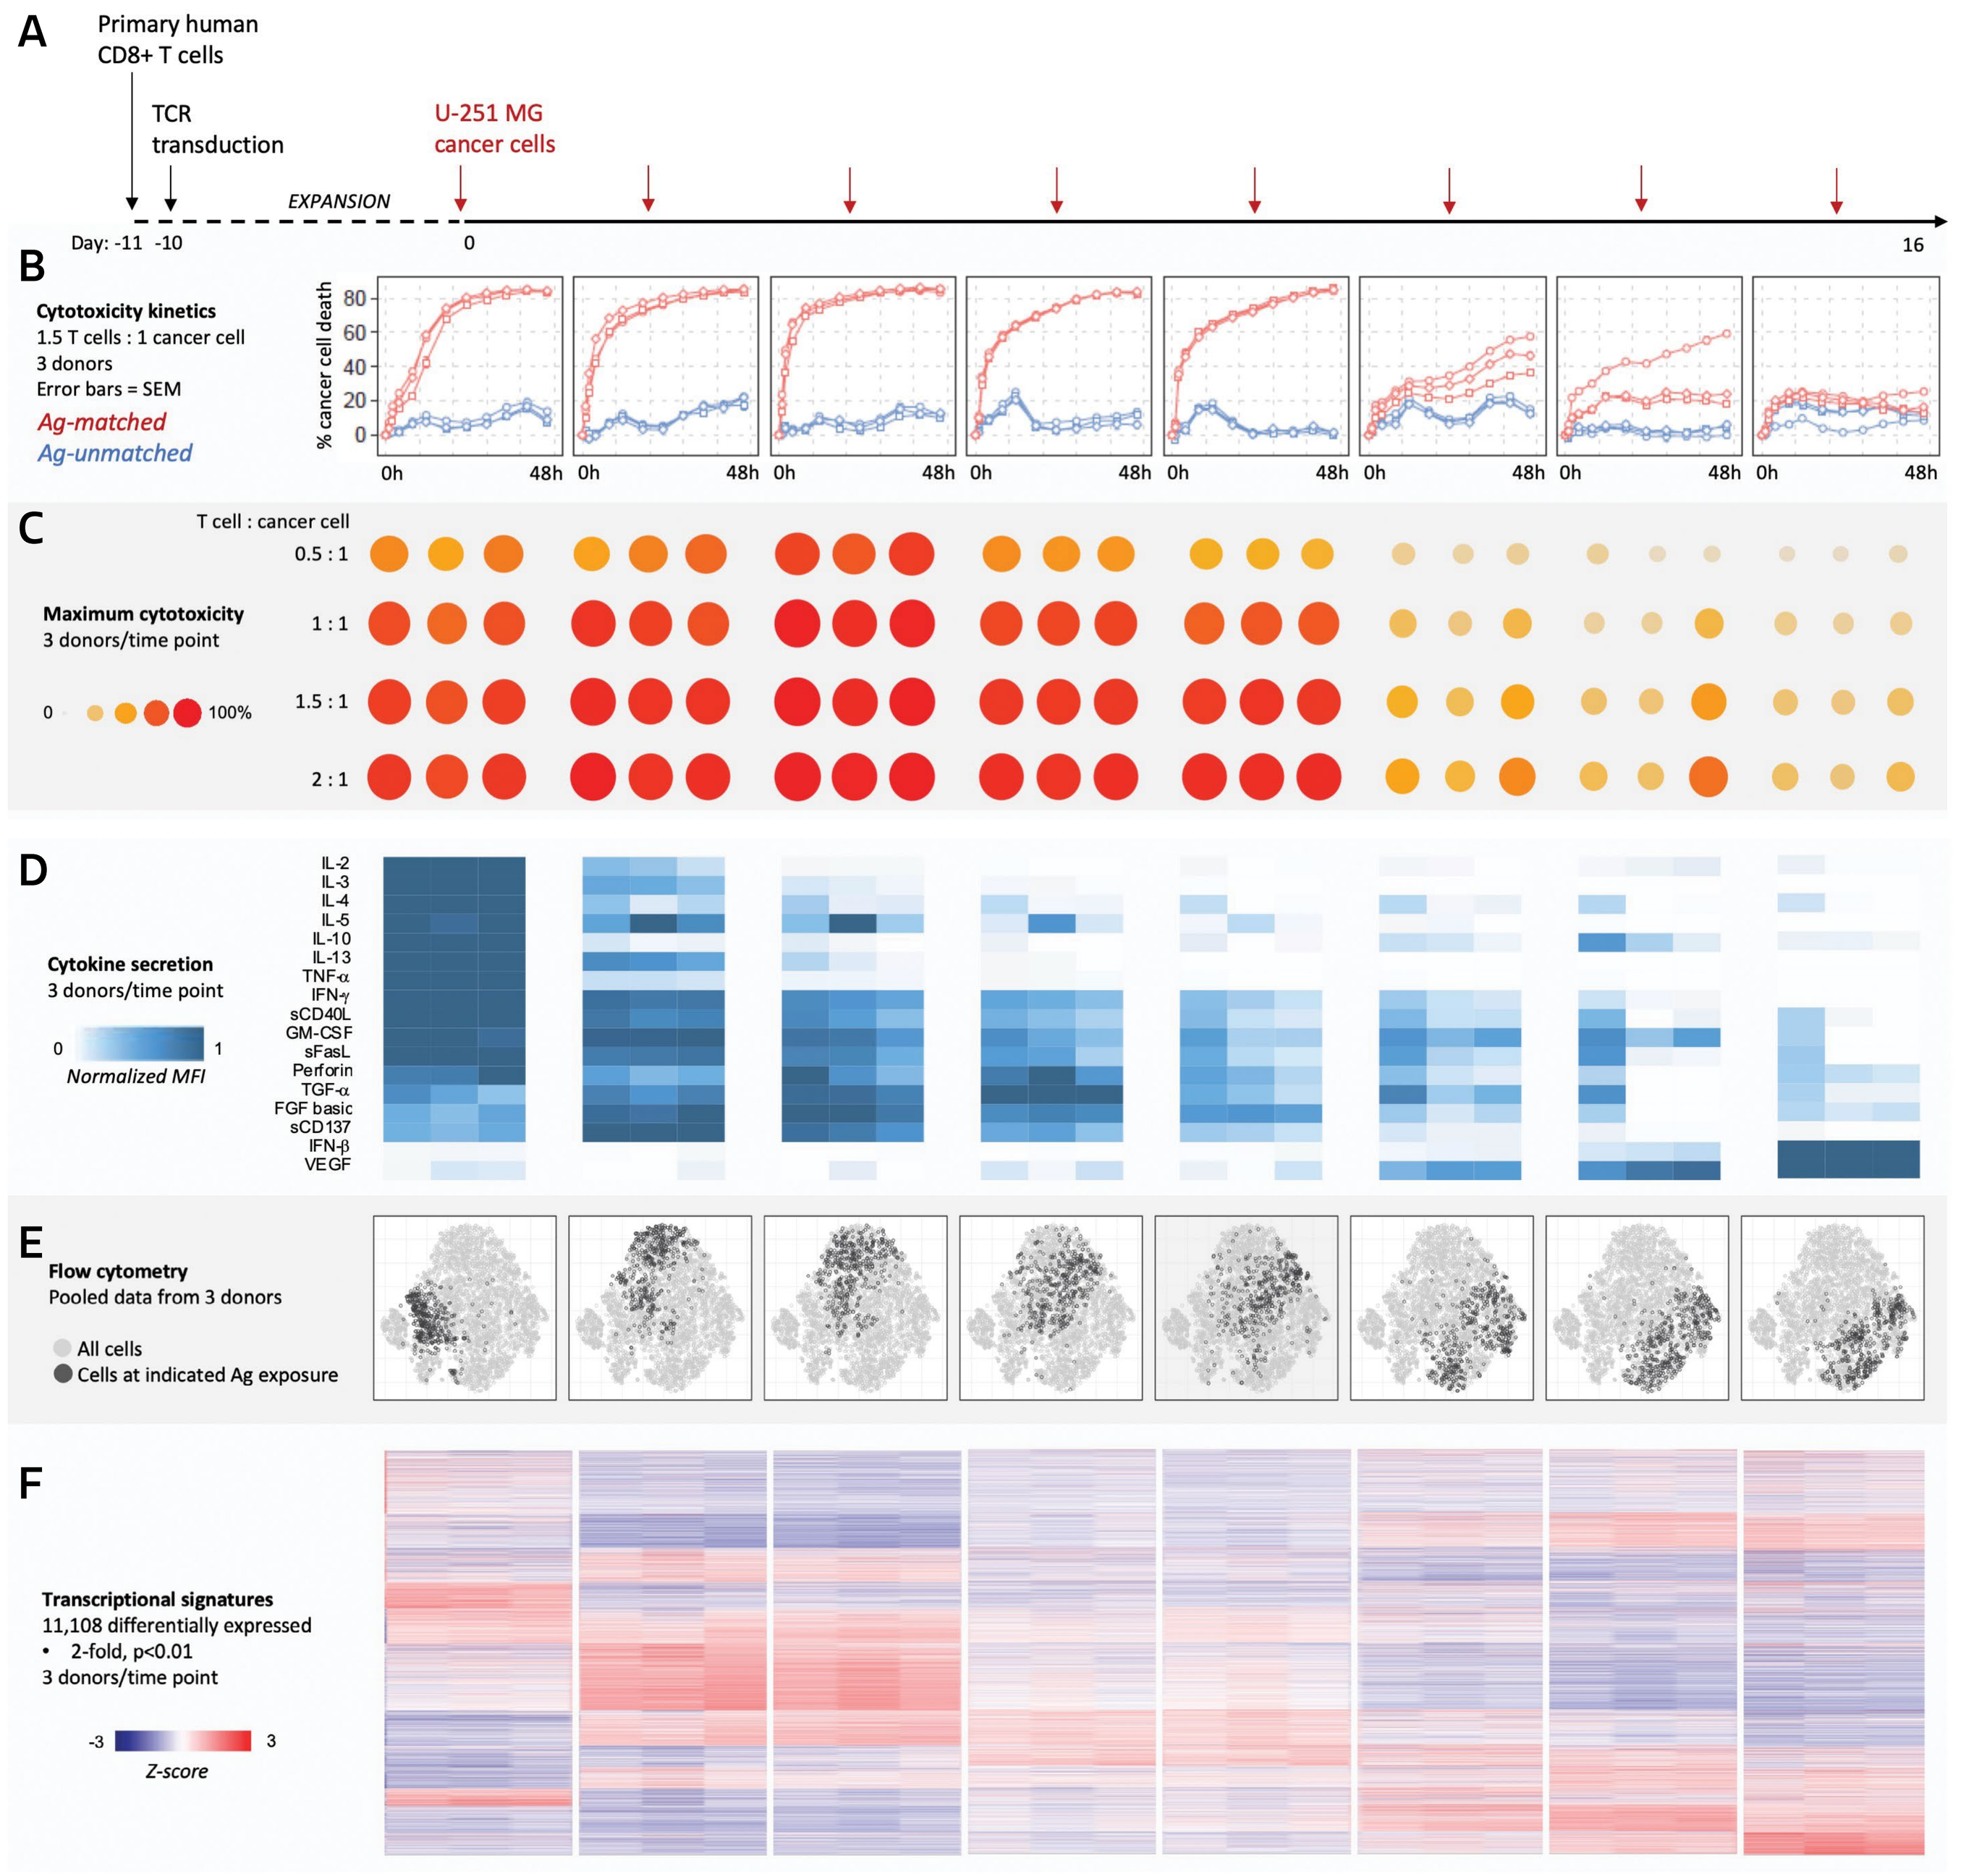
A
Primary human CD8+ T cells
TCR transduction
U-251 MG cancer cells
EXPANSION
Day: -11 -10
0
16
B
Cytotoxicity kinetics
1.5 T cells : 1 cancer cell
3 donors
Error bars = SEM
Ag-matched
Ag-unmatched
% cancer cell death
C
T cell : cancer cell
Maximum cytotoxicity
3 donors/time point
0.5 : 1
1 : 1
1.5 : 1
2 : 1
100%
D
Cytokine secretion
3 donors/time point
Normalized MFI
IL-2
IL-3
IL-4
IL-5
IL-10
IL-13
TNF-α
IFN-γ
sCD40L
GM-CSF
sFasL
Perforin
TGF-α
FGF basic
sCD137
IFN-β
VEGF
E
Flow cytometry
Pooled data from 3 donors
All cells
Cells at indicated Ag exposure
F
Transcriptional signatures
11,108 differentially expressed
2-fold, p<0.01
3 donors/time point
Z-score

A) Method for driving T cell dysfunction in vitro. *T cell preparation: PBMCs were isolated from three healthy donors on a Ficoll gradient and rested overnight. CD8+ T cells were isolated on magnetic beads, stimulated with CD3/CD28, transduced with NY-ESO-1 lentivirus, and expanded. TCR expression was confirmed by flow cytometry. Cancer cell preparation: U251 MG cell lines were transduced with lentivirus encoding a fusion protein of b2-microgobulin, HLA2-A2 and NY-ESO-1 or MART-1 peptide and selected on blasticidin. Antigen expression was confirmed with an NFAT reporter system in Jurkat T cells. For co-culture: A fixed number of irradiated U251 MG cells were cultured for 24h. CD8+ T cells were added to achieve the desired cancer:T cell ratio. The co-culture was monitored every 24h. If cancer cells were depleted, CD8+ T cells were transferred to a fresh well of irradiated U251 MG cells and the remainder were collected for downstream analyses. If cancer cells were not depleted, media was changed and reassessed the following day.* **B) Cytotoxicity kinetics** *as monitored by live cell fluorescence microscopy. Cancer cells were stained with CellTrace Far Red and incubated overnight. CD8+ T cells were added to achieve the desired cancer:T cell ratio. Whole-well imaging in the Cy5 channel was assessed in confocal mode using an ImageXpress Micro Confocal High-Content Imaging System. Image analysis was performed in MetaXpress software. Briefly, intensity and object size thresholds were applied,and the total cell area above these thresholds was calculated and normalized against the 0h time point.* **C) Maximum cytotoxicity** *across four T cell : cancer cell ratios, as calculated by area under the curve in 'B'.* **D) Secreted effector molecules.** *T cells were exposed to fresh antigen for 24h and supernatants were analyzed using the Human XL Cytokine Discovery Kit, Human Magnetic Luminex Assay or Human CD8+ T-Cell Magnetic Bead Panel. Analysis was performed using FlexMap 3D software. Briefly, mean fluorescence intensity (MFI) for each analyte was normalized to the maximum MFI observed over the course of the assay, and the resulting data was scaled between 0 and 1.* **E) tSNE visualization of flow cytometry measurements** *for twelve proteins. Assay details in Figure 2 legend.* **F) RNA expression** *for 11,108 differentially expressed genes. Assay details in Figure 3 legend.*

Figure 2. Chronic cancer antigen exposure drives expression of activation and dysfunction markers on CD8+ T cells

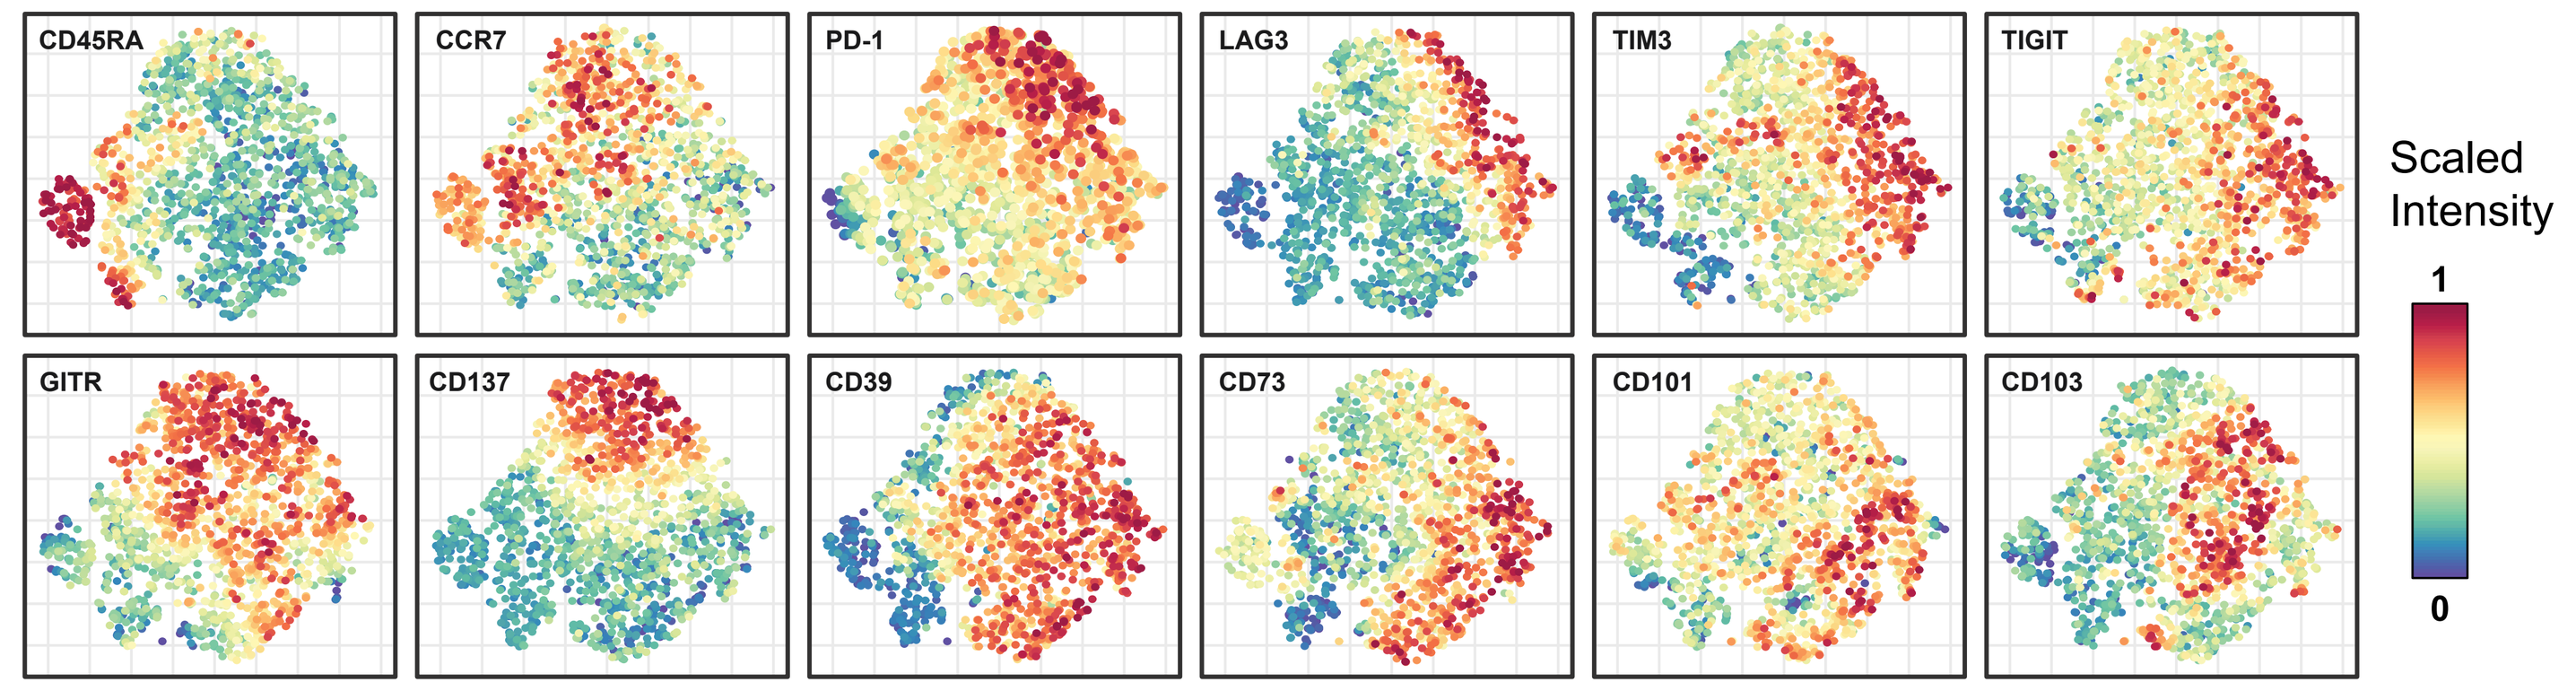
CD45RA
CCR7
PD-1
LAG3
TIM3
TIGIT
GITR
CD137
CD39
CD73
CD101
CD103
Scaled Intensity
1
0

Cells were washed, stained with 1) Near IR Live/Dead dye and 2) a mix of antibodies against CD3, CD8 and the markers listed below or isotype controls, and finally fixed in 1.6% paraformaldehyde. To accommodate integrated analysis of different time points all samples were processed and run in parallel. Data were analyzed in R using a custom workflow and functions from flowCore and flowDensity packages. Briefly, we gated on viable CD3+CD8+ cells, calculated a scaled mean fluorescence intensity and transformed using the hyperbolic arcsine to accommodate for negative values. Transformed data was used for k-means clustering (k=8) and t-Distributed Stochastic Neighbor Embedding (t-SNE) visualization (perplexity=20). Transformed expression data was further scaled between 0 and 1 for each marker.

Figure 3. Chronic cancer antigen exposure drives transcriptional signatures of activation and dysfunction in CD8+ T cells

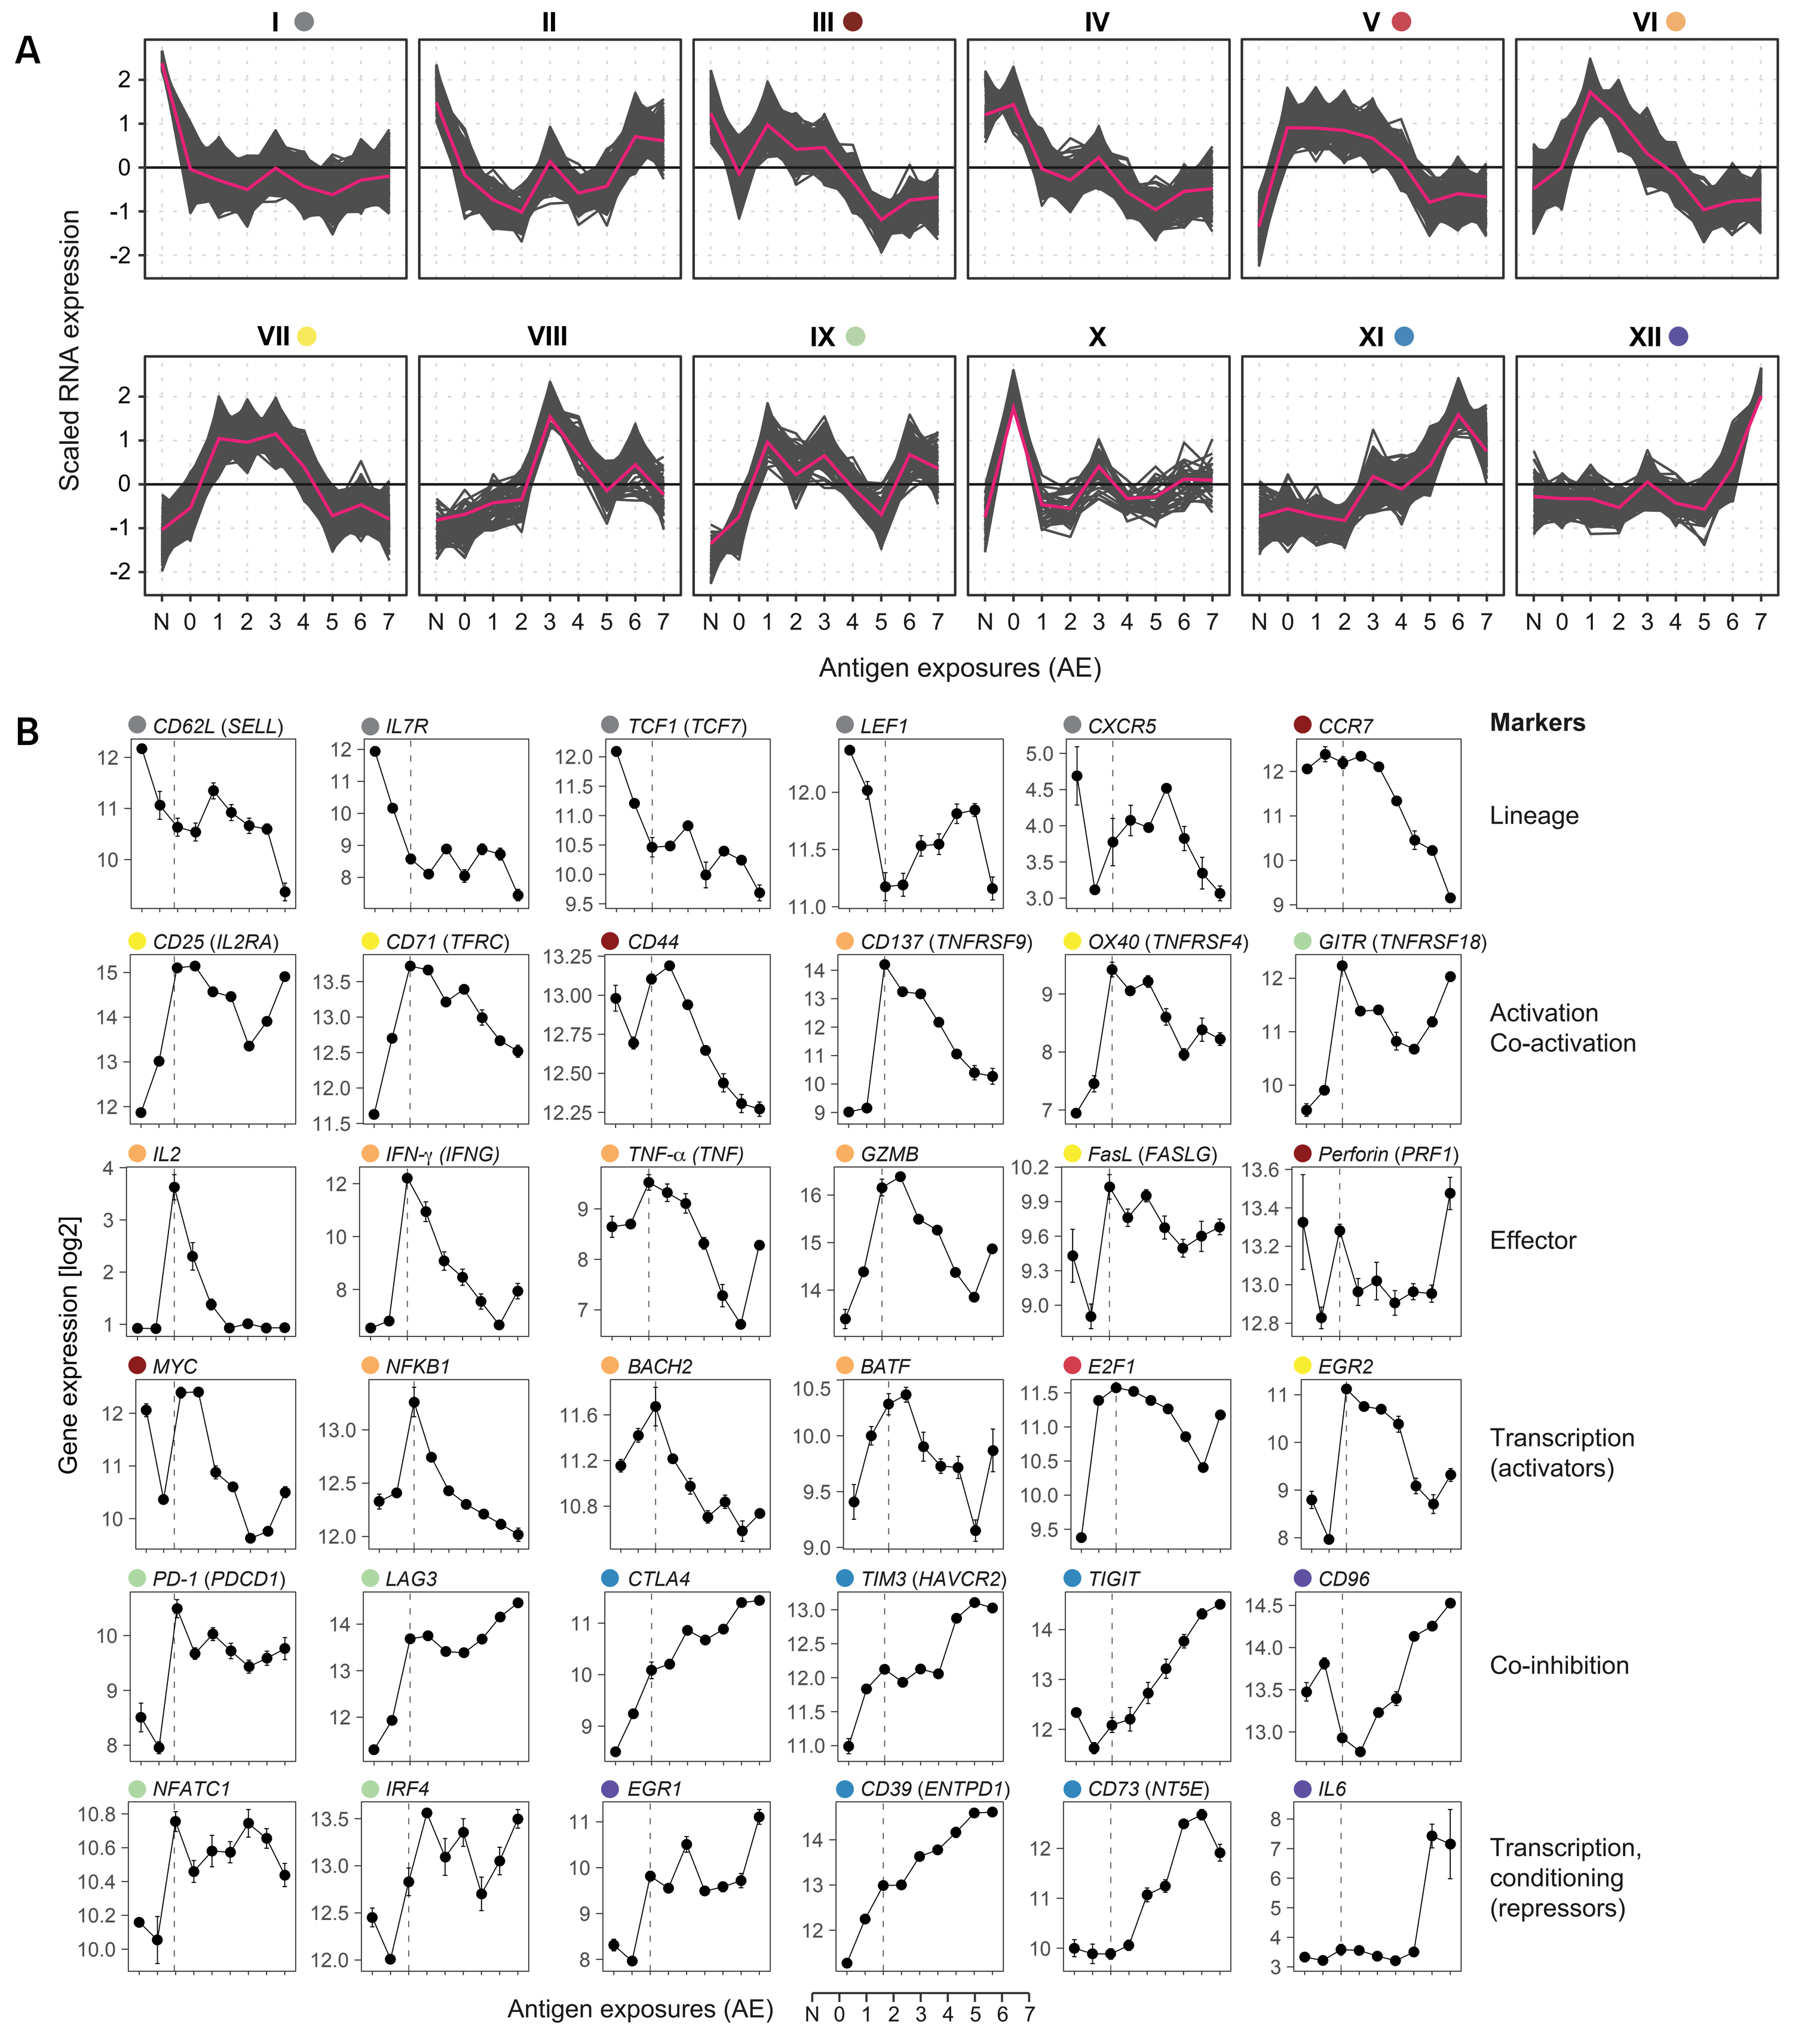
A
I
II
III
IV
V
VI
VII
VIII
IX
X
XI
XII
Scaled RNA expression
N 0 1 2 3 4 5 6 7
Antigen exposures (AE)
B
CD62L (SELL)
IL7R
TCF1 (TCF7)
LEF1
CXCR5
CCR7
Markers
Lineage
CD25 (IL2RA)
CD71 (TFRC)
CD44
CD137 (TNFRSF9)
OX40 (TNFRSF4)
GITR (TNFRSF18)
Activation
Co-activation
IL2
IFN-γ (IFNG)
TNF-α (TNF)
GZMB
FasL (FASLG)
Perforin (PRF1)
Effector
MYC
NFKB1
BACH2
BATF
E2F1
EGR2
Transcription
(activators)
PD-1 (PDCD1)
LAG3
CTLA4
TIM3 (HAVCR2)
TIGIT
CD96
Co-inhibition
NFATC1
IRF4
EGR1
CD39 (ENTPD1)
CD73 (NT5E)
IL6
Transcription,
conditioning
(repressors)
Gene expression [log2]
Antigen exposures (AE)
N 0 1 2 3 4 5 6 7

cDNA libraries were prepared using the TruSeq RNA Library Prep Kit v2 and sequenced on a NextSeq 550 (76 bp paired end run) generating an average of 20 million paired reads per sample. Reads were mapped against NCBI Human Reference Genome Build GRCh37 (hg19) using the STAR aligner in BaseSpace (Illumina). Data normalization and differential gene expression analysis was performed using DESeq2 functions in R. p-values were adjusted using the Benjamini-Hochberg method. K-means clustering was performed in R on scaled count data (k=12 based on correlation analysis). A) **K-means cluster profiles**, *with mean cluster expression shown in red and individual genes in grey. B)* **Normalized log2 RNA expression** *for selected genes. Error bars = SEM across three donors.*

Figure 4. Chronic cancer antigen exposure ultimately drives CD8+ T cells to a anti-PD-1 refractory state

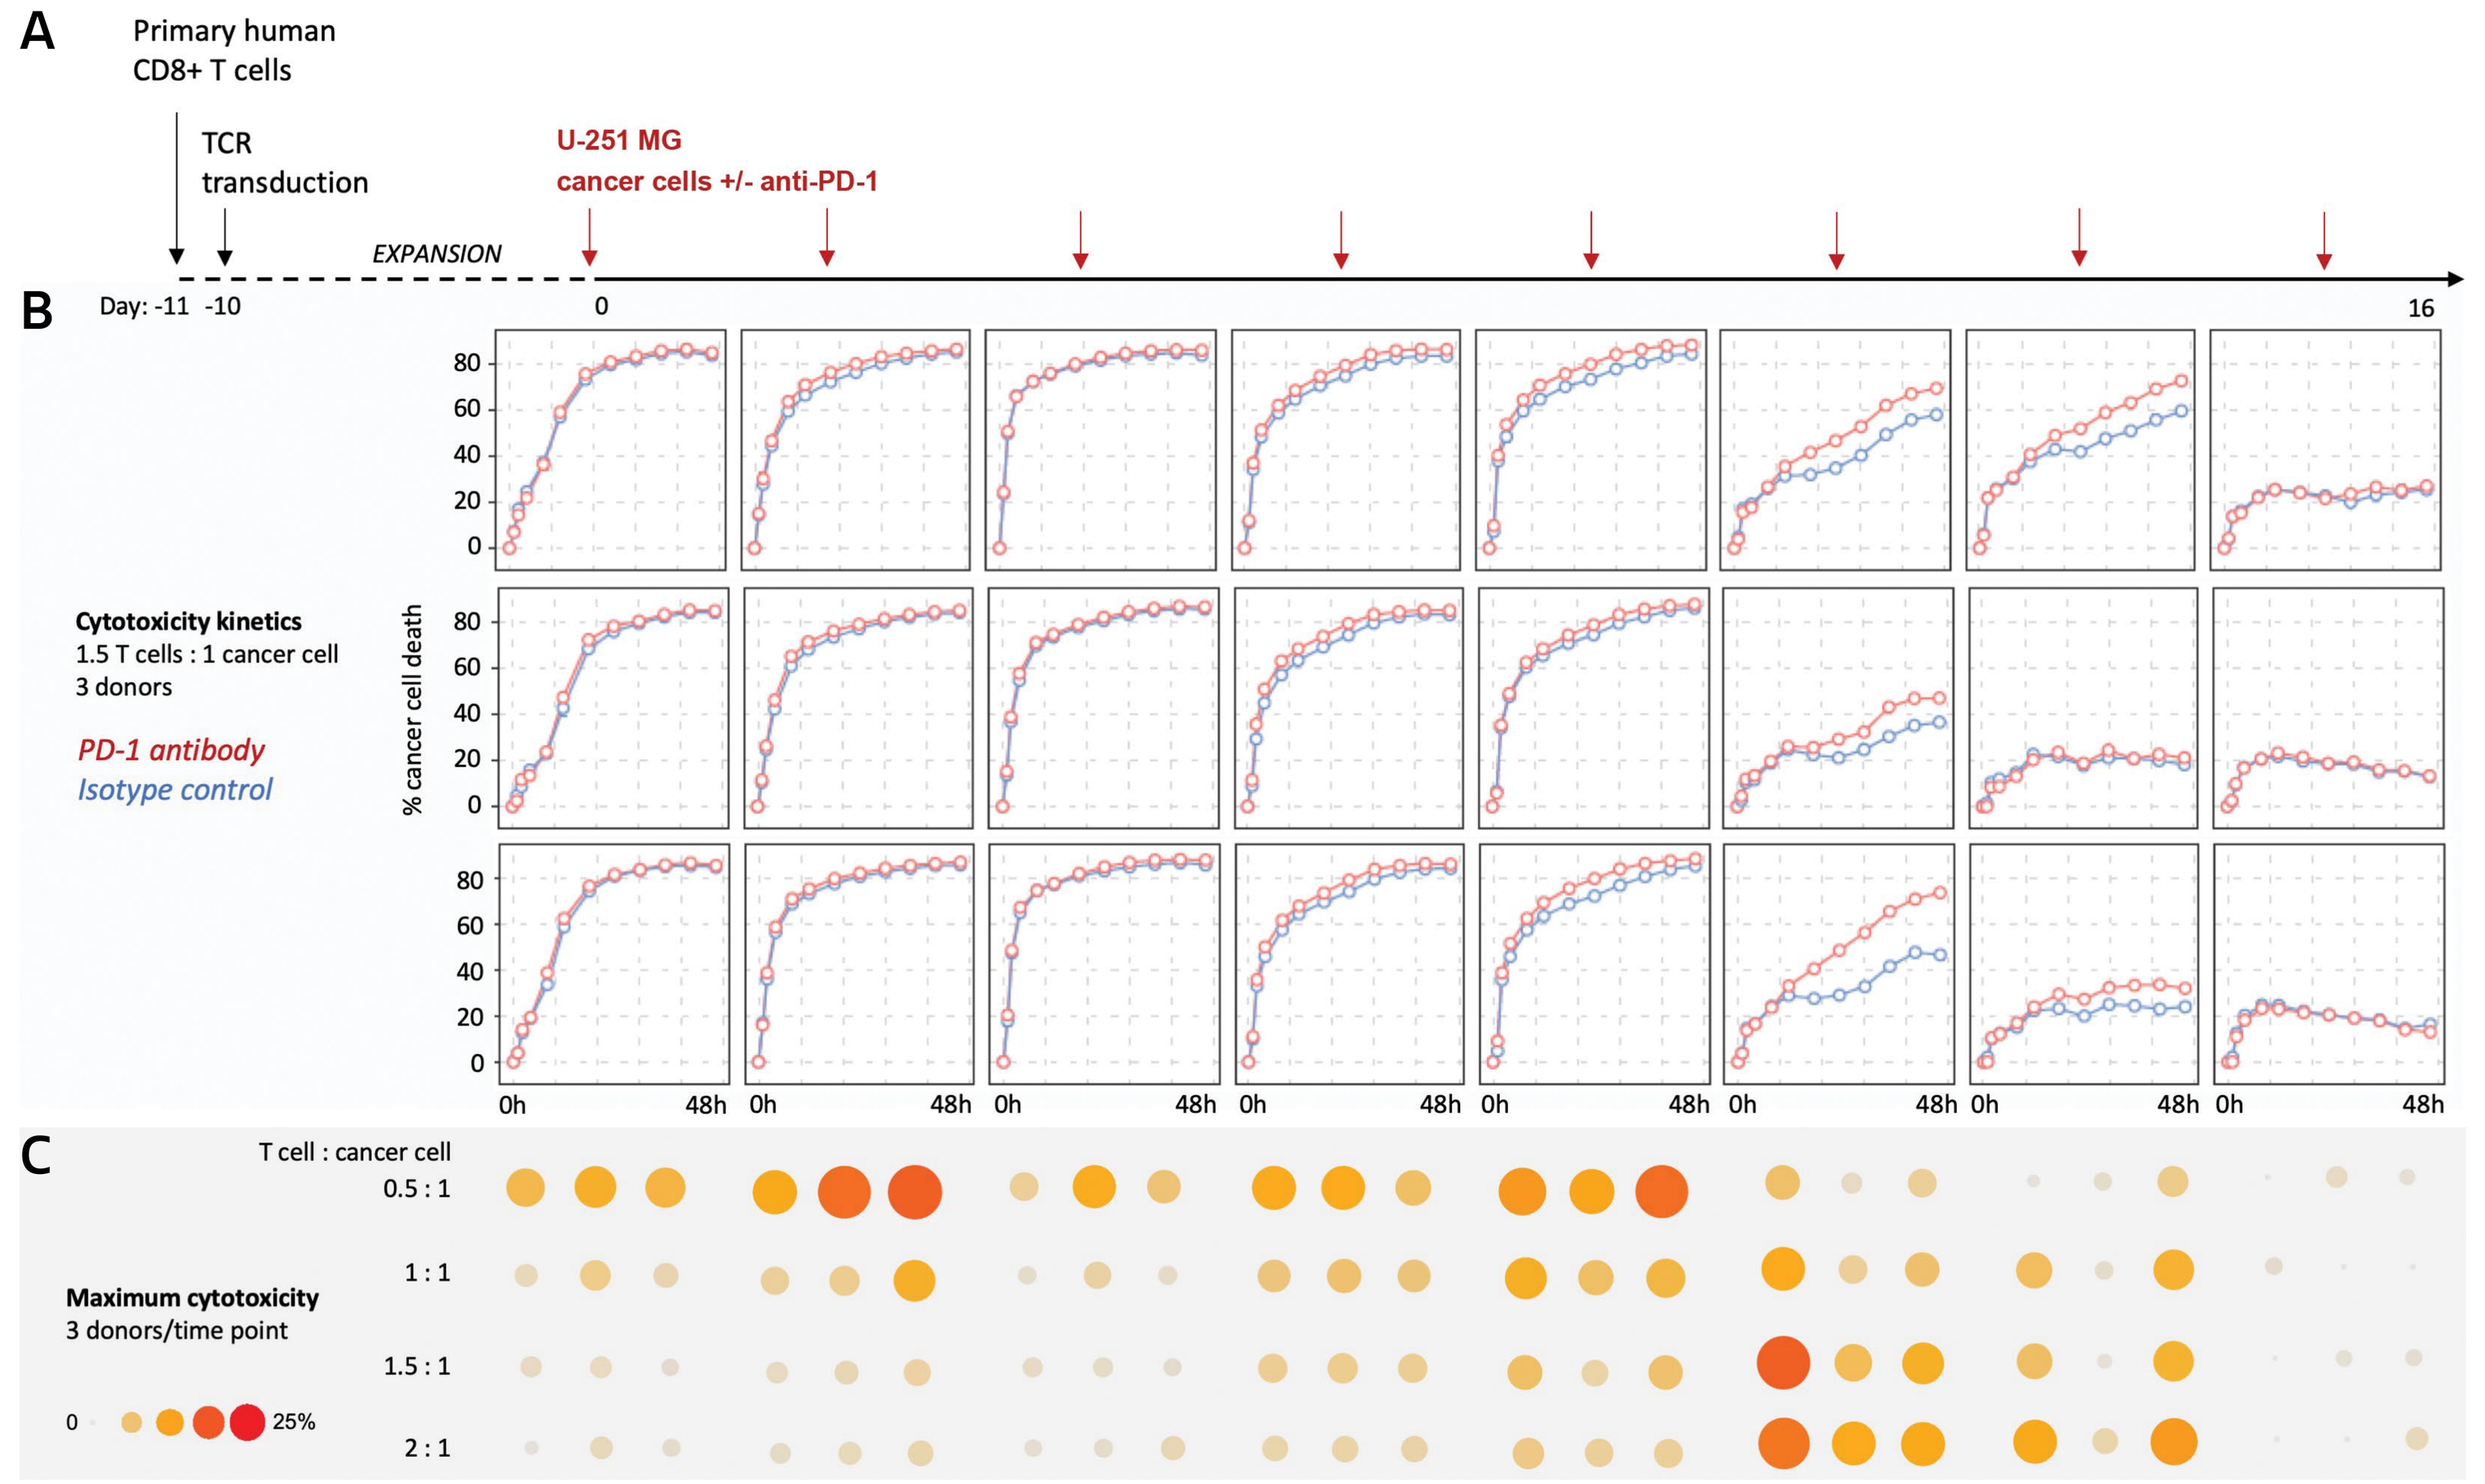
A
Primary human CD8+ T cells
TCR transduction
U-251 MG cancer cells +/- anti-PD-1
EXPANSION
Day: -11 -10
0
16
B
Cytotoxicity kinetics
1.5 T cells : 1 cancer cell
3 donors
PD-1 antibody
Isotype control
% cancer cell death
0h
48h
C
T cell : cancer cell
Maximum cytotoxicity
3 donors/time point
0.5 : 1
1 : 1
1.5 : 1
2 : 1
25%

A) Method for evaluating the effect of anti-PD1 antibody on T cell dysfunction in vitro. *Co-cultures were supplemented with 5ug/mL Nivolumab or an in-house IgG4 isotype control antibody. Additional assay details in Figure 1A legend.* **B) Cytotoxicity kinetics** *and* **C) maximum cytotoxicity** *across four T cell : cancer cell ratios, in the presence or absence of PD-1 antibody. Assay details in Figure 1B,C legend.*

Figure 5. A gene expression signature associated with the anti-PD-1 refractory state predicts anti-PD-1 response in melanoma patients

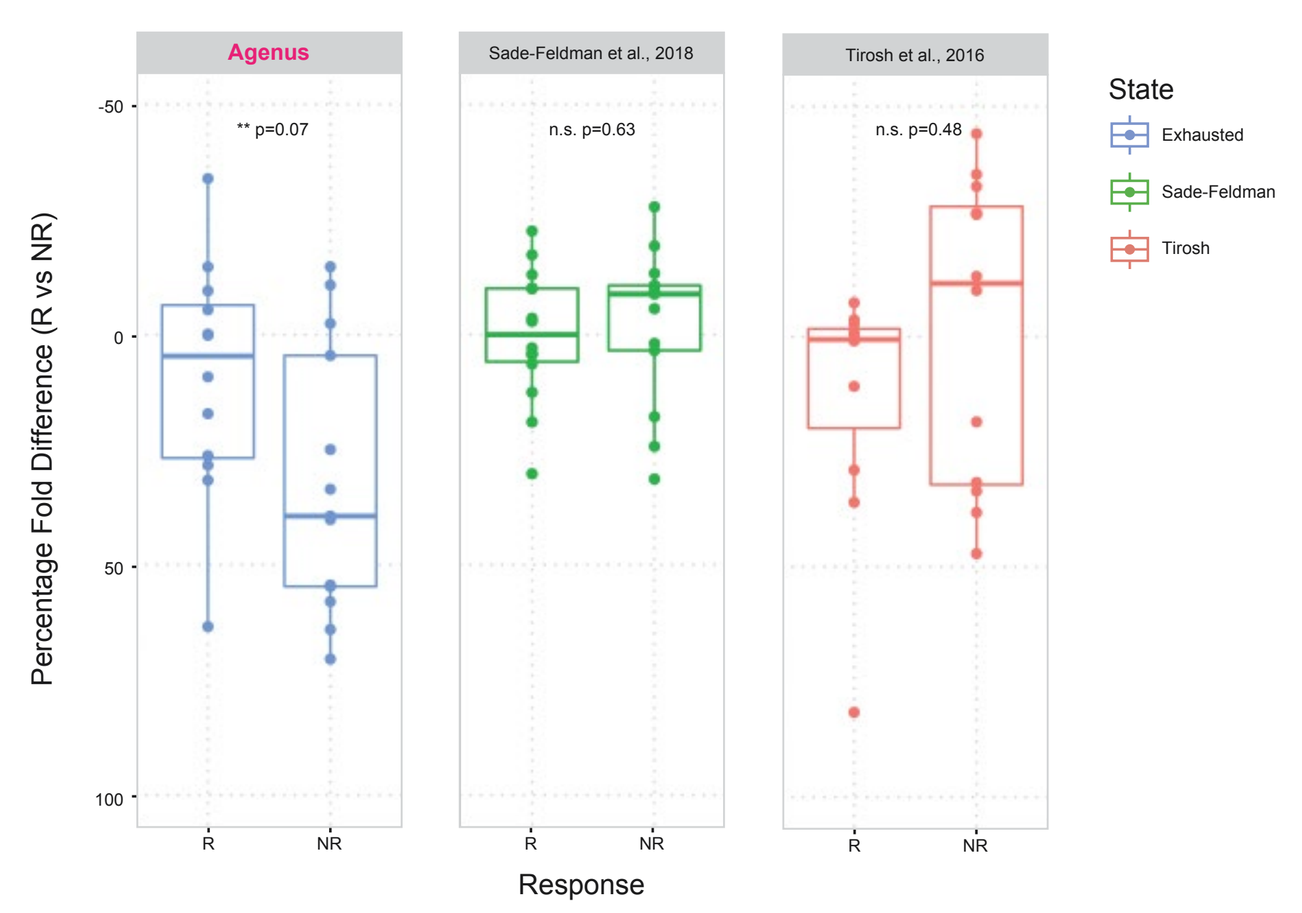
Agenus
Sade-Feldman et al., 2018
Tirosh et al., 2016
** p=0.07
n.s. p=0.63
n.s. p=0.48
State
Exhausted
Sade-Feldman
Tirosh
Percentage Fold Difference (R vs NR)
R
NR
Response

Summarized expression of Agenus dysfunction state signature (n=231 genes) in metastatic melanoma patients treated with anti-PD-1 antibody[1] compared with published T cell exhaustion signatures derived from single cell RNA-sequencing of melanoma tumor infiltrating lymphocytes[2,3]. (n=1088 genes and 28 genes, respectively). Statistical significance was assessed by two-proportion Z test.

Figure 6. Deep profiling of anti-PD-1 responsive T cell subsets identifies combination approaches to enhance durability of anti-PD-1 responses

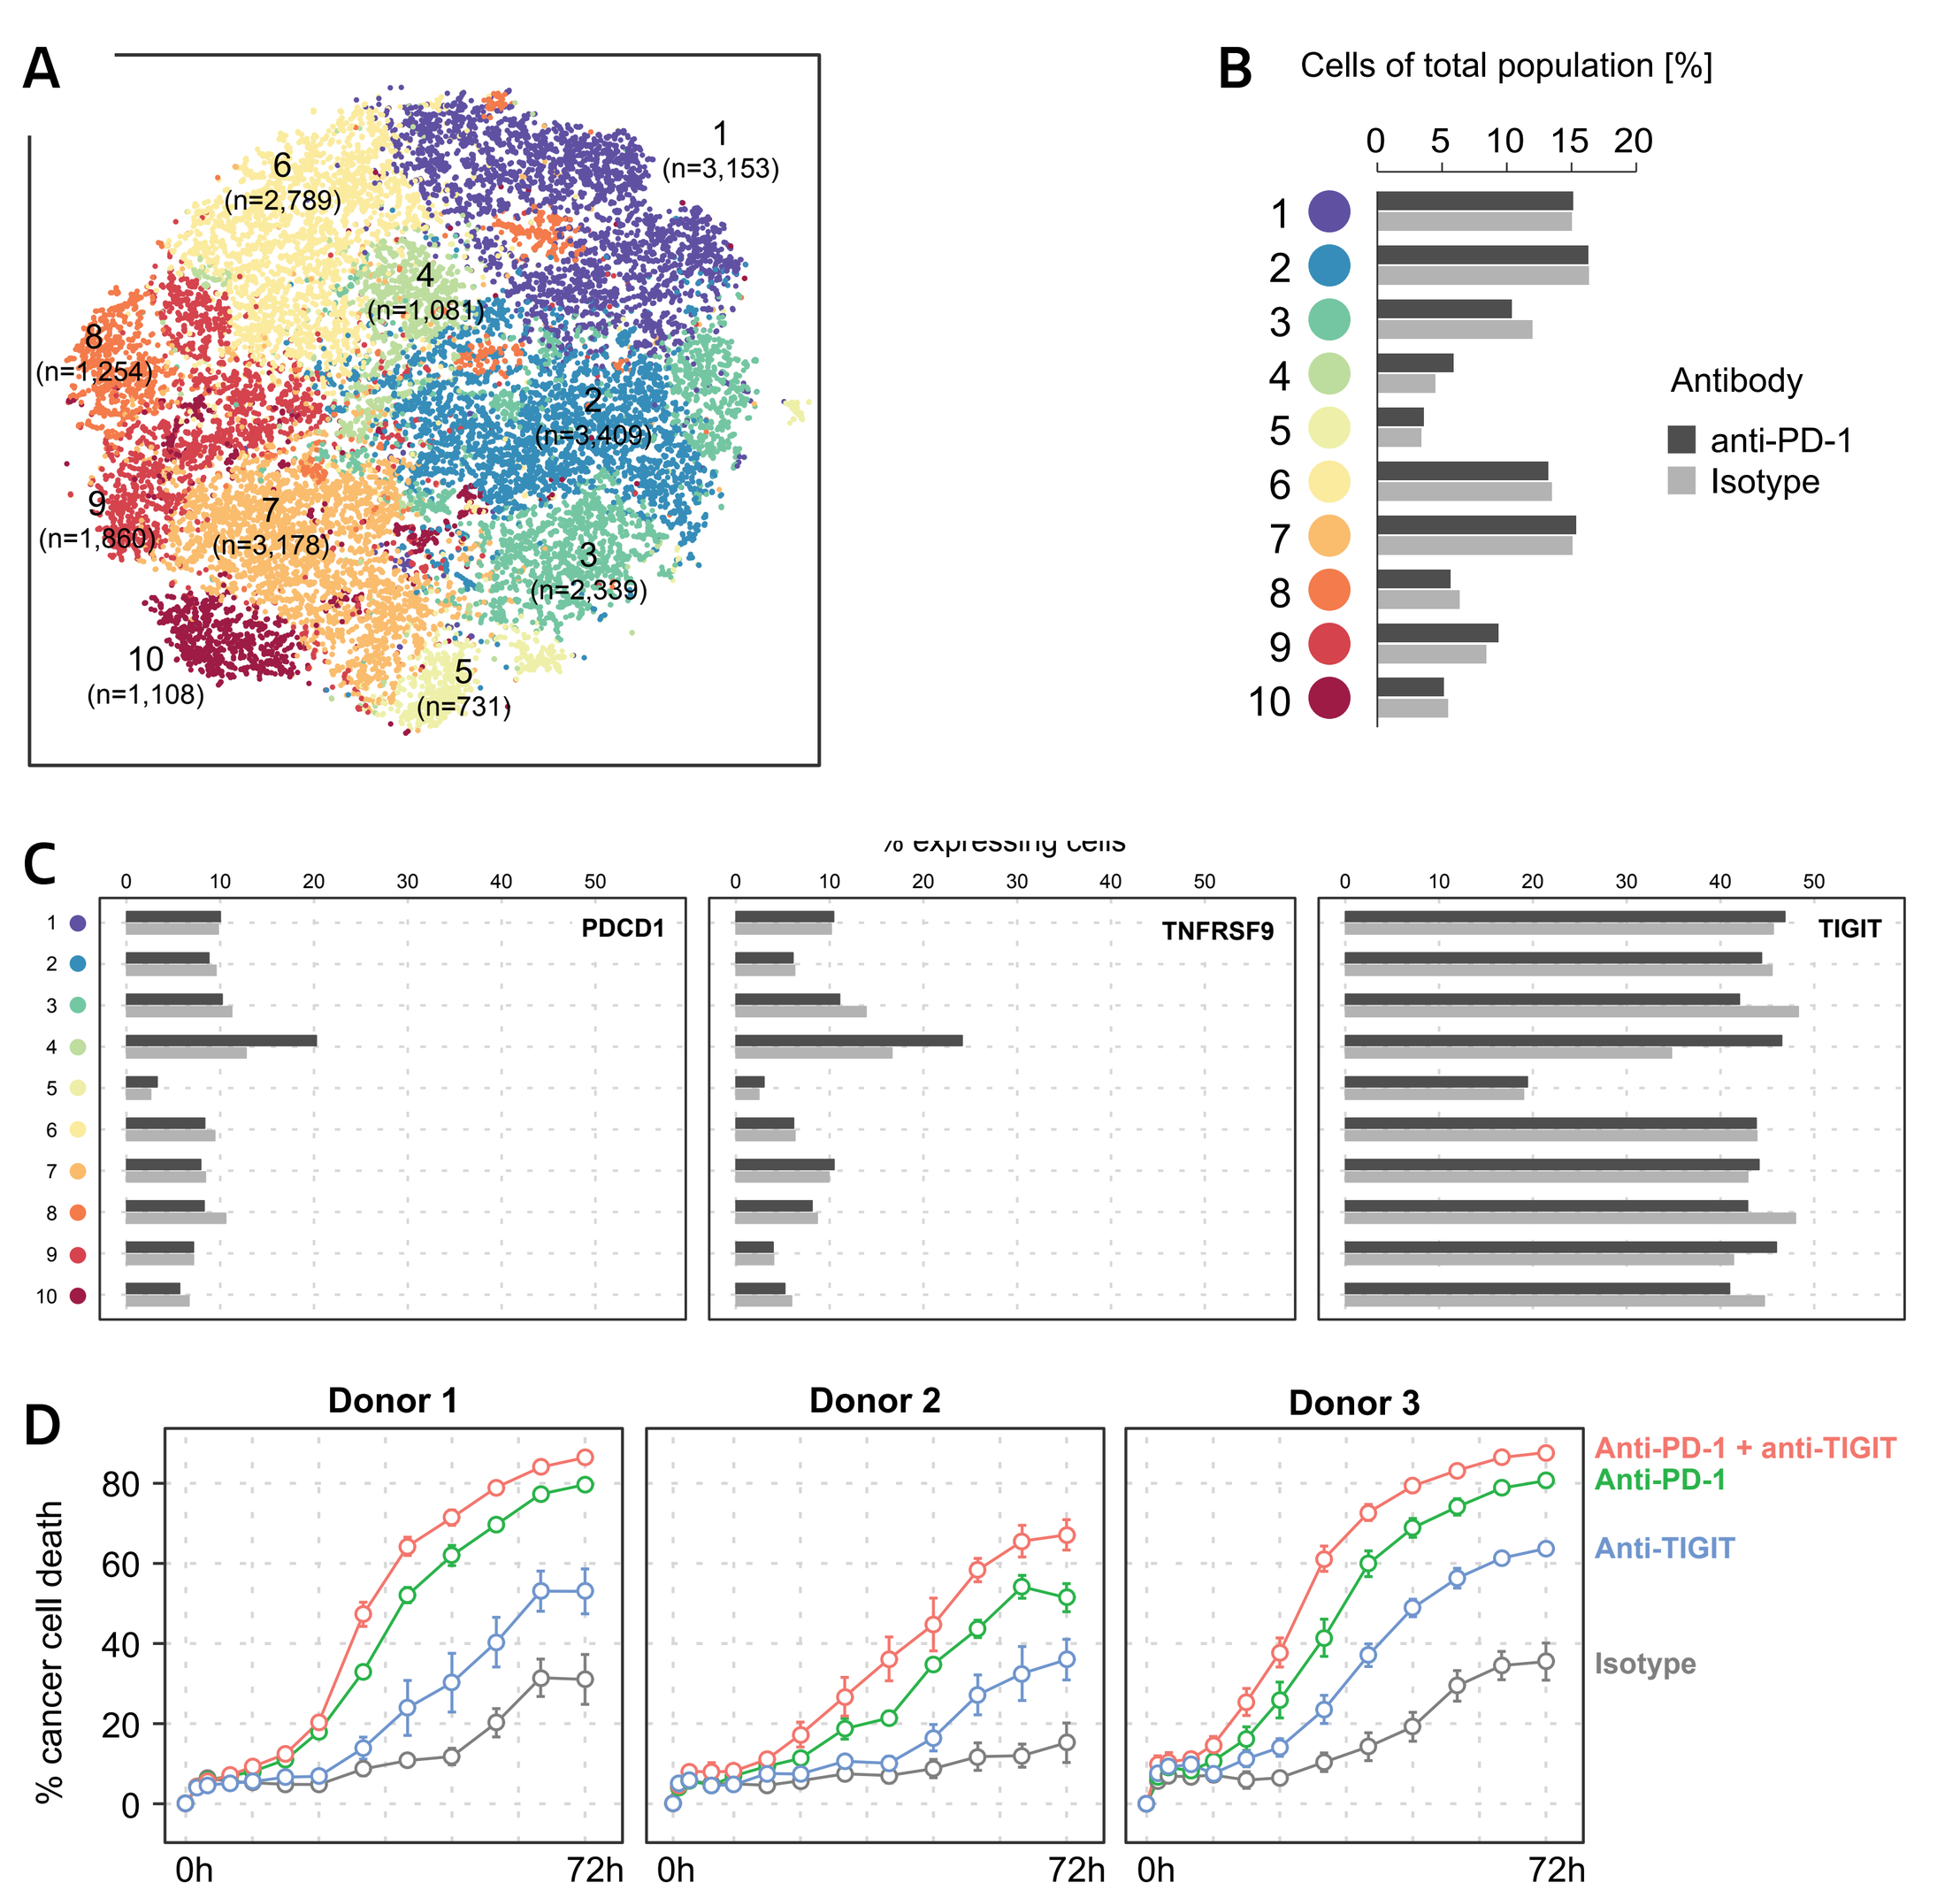
A
1 (n=3,153)
2 (n=3,409)
3 (n=2,339)
4 (n=1,081)
5 (n=731)
6 (n=2,789)
7 (n=3,178)
8 (n=1,254)
9 (n=1,860)
10 (n=1,108)
B
Cells of total population [%]
Antibody
anti-PD-1
Isotype
C
% expressing cells
PDCD1
TNFRSF9
TIGIT
D
Donor 1
Donor 2
Donor 3
% cancer cell death
0h
72h
Anti-PD-1 + anti-TIGIT
Anti-PD-1
Anti-TIGIT
Isotype

CD8+ T cells from three donors were co-cultured through five antigen exposures and harvested. Single cell cDNA libraries were constructed using the 10X Genomics single cell 3' reagent kit v2 and sequenced on a NextSeq 550 (26 bp/98 bp paired end run) generating >50,000 paired reads per cell. Reads were aligned to the NCBI Human Reference Genome Build GRCh38 (hg38) and processed with counts of unique molecular identifiers (UMIs) using Cell Ranger software and downstream analysis was performed using the R package Seurat. **A) tSNE visualization of ~20k cells** *from three donors, based on variably expressed genes. Cells are colored based on k-means cluster assignment.* **B) Frequency of cells** *in each cluster in the presence or absence of 5ug/ml Nivolumab.* **C) Frequency of PDCD1, TNFRSF9 & TIGIT expression** *across K-means clusters. Assay details in Figure 2 legend.* **D) Cytotoxicity kinetics** *as monitored by live cell fluorescence microscopy in the presence of 5ug/ml Nivolumab and/or anti-TIGIT antibodies. Assay details in Figure 1B legend.*

Figure 7. I-O target discovery and translational insight from deep, integrative profiling of the tumor:immune interface in vitro

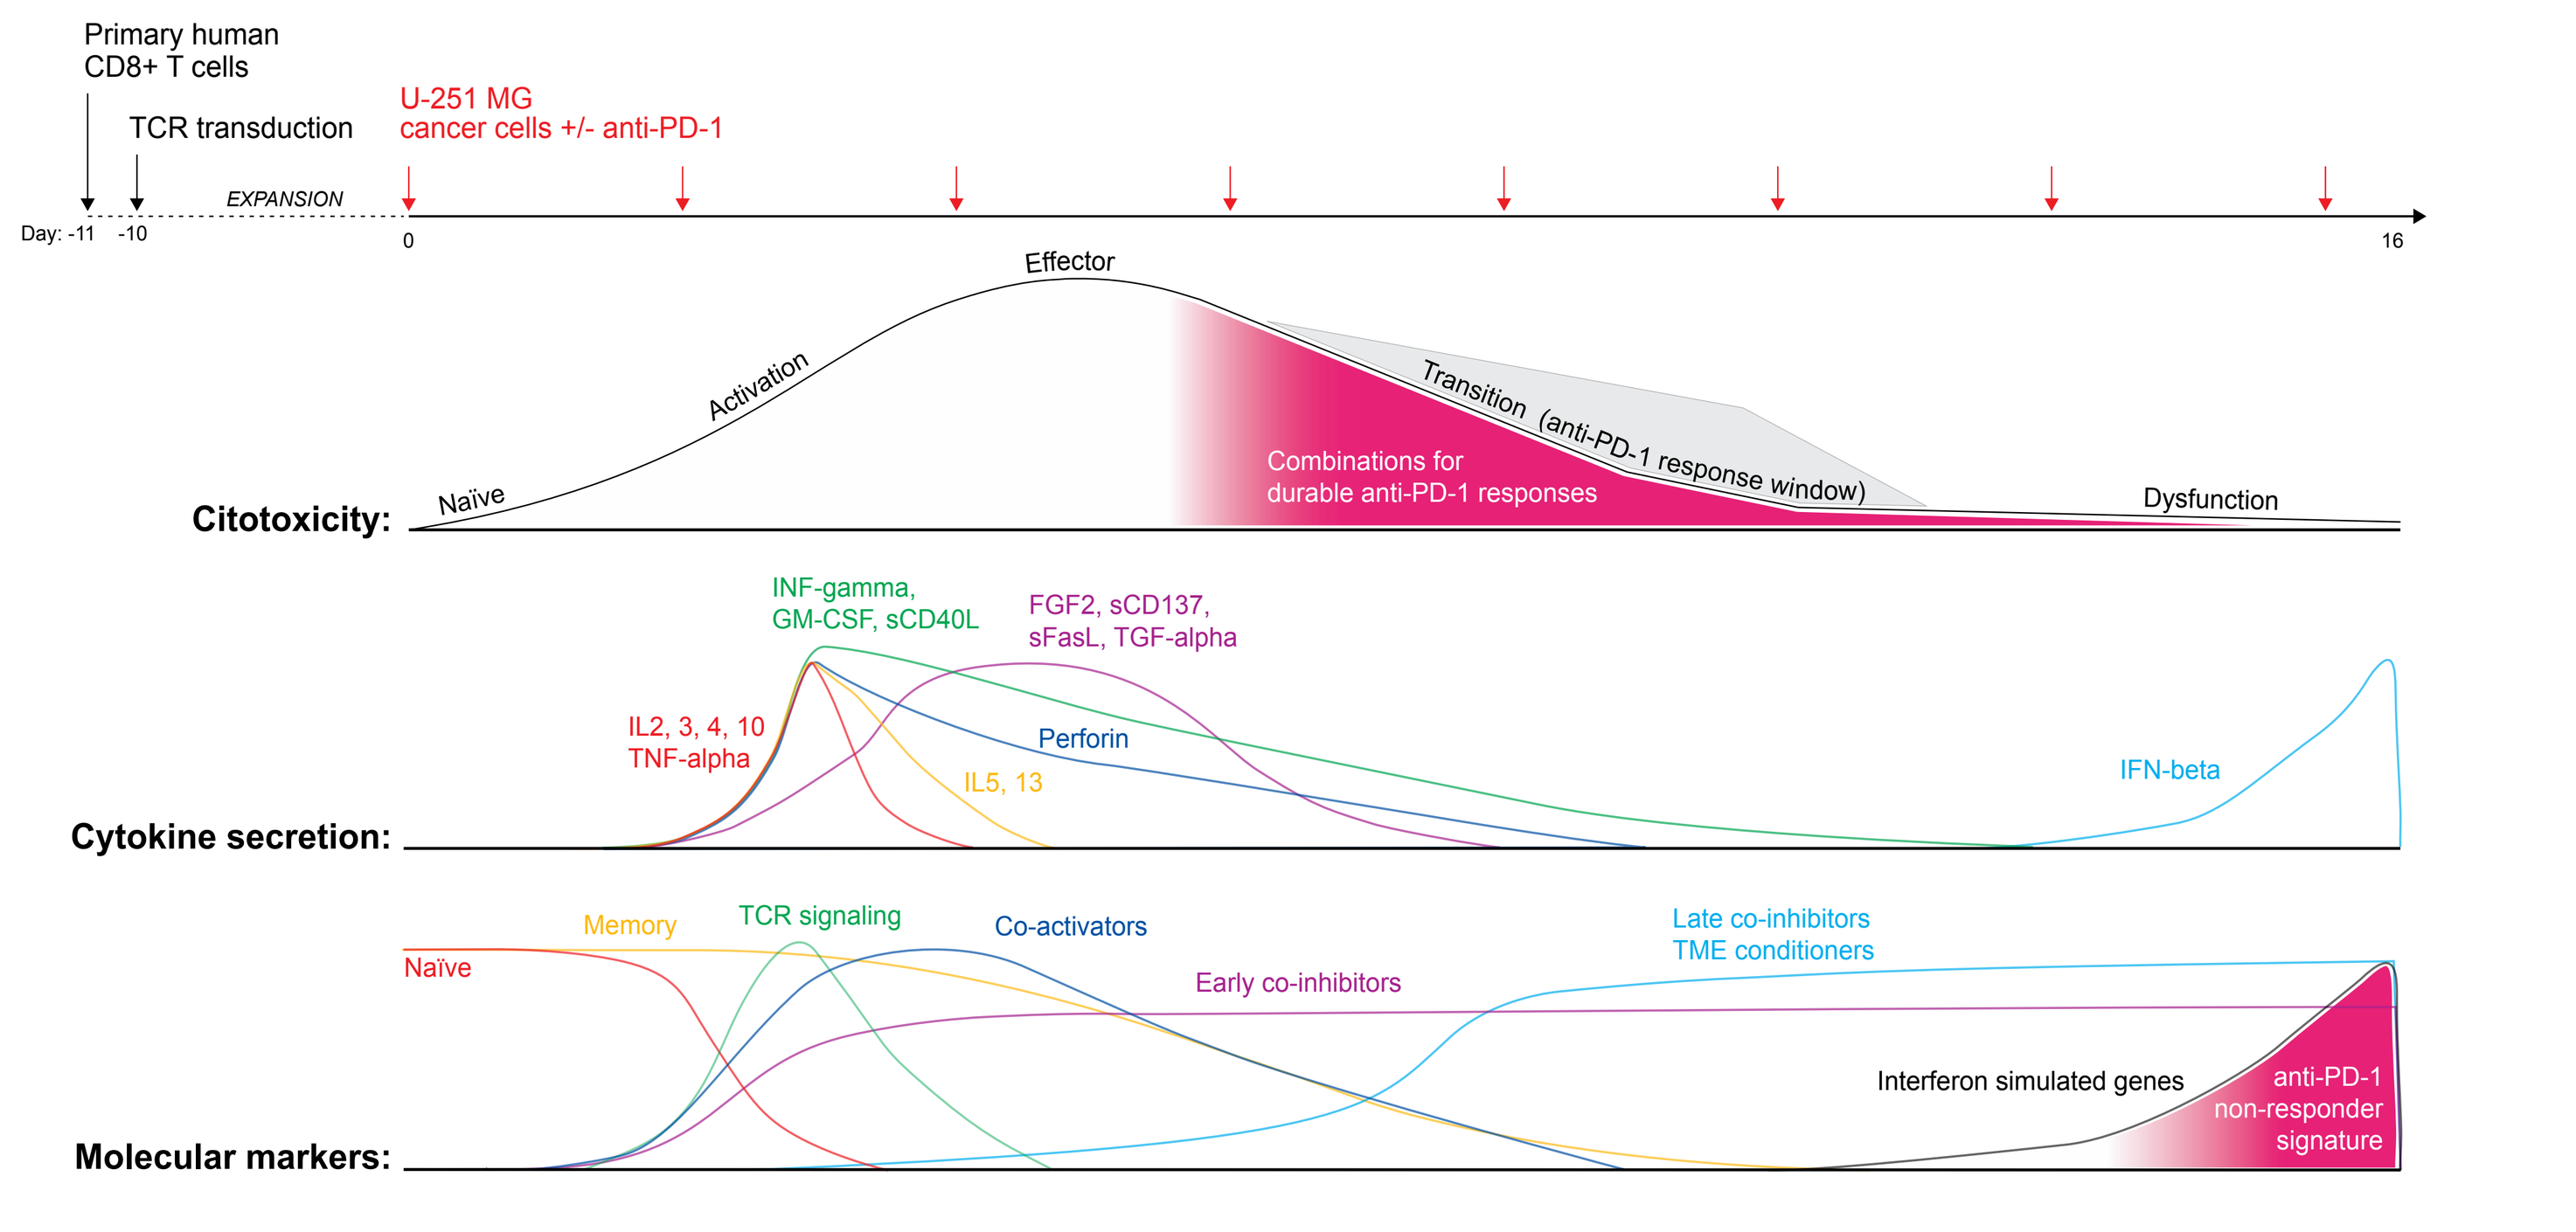
Primary human CD8+ T cells
TCR transduction
U-251 MG cancer cells +/- anti-PD-1
EXPANSION
Day: -11 -10
0
16
Citotoxicity:
Naïve
Activation
Effector
Transition (anti-PD-1 response window)
Combinations for durable anti-PD-1 responses
Dysfunction
Cytokine secretion:
IL2, 3, 4, 10
TNF-alpha
INF-gamma,
GM-CSF, sCD40L
IL5, 13
FGF2, sCD137,
sFasL, TGF-alpha
Perforin
IFN-beta
Molecular markers:
Naïve
Memory
TCR signaling
Co-activators
Early co-inhibitors
Late co-inhibitors
TME conditioners
Interferon simulated genes
anti-PD-1 non-responder signature

References

1. Hugo W, Zaretsky JM, Sun L, Song C, Moreno BH, Hu-Lieskovan S, et al. Genomic and transcriptomic features of response to anti-PD-1 therapy in metastatic melanoma. Cell. 2016;165(1):35-44
2. Sade-Feldman M, Yizhak K, Bjorgaard SL, Ray JP, de Boer CG, Jenkins RW, et al. Defining T cell states associated with response to checkpoint immunotherapy in melanoma. Cell. 2018;175(4):998-1013 e20.
3. Tirosh I, Izar B, Prakadan SM, Wadsworth MH, 2nd, Treacy D, Trombetta JJ, et al. Dissecting the multicellular ecosystem of metastatic melanoma by single-cell RNA-seq. Science. 2016;352(6282):189-96.

Acknowledgments

We thank M. Robinson, R. Stein, A. Hurwitz and R. Choudhary for critical discussion of the work. Design: Carlos Barrientos