AGEN2034, a novel anti-PD-1 antibody that combines effectively with CTLA-4 pathway blockade to enhance T cell activity

Dhan Chand[1], David Savitsky[1], Ana Gonzalez[1], Mariana Manrique[1], Christopher Clarke[1], Andrea Schuster[1], Elise E. Drouin[1], Jeremy D. Waight[1], Cornelia Mundt[1], Gerd Ritter[2], Taha Merghoub[3], David Schaer[3], Rikke B. Holmgaard[3], Roberta Zappasodi[3], Jedd Wolchok[3], Marc van Dijk[1], Jennifer S. Buell[1], Jean-Marie Cuillerot[1], Robert Stein[1] and Nicholas S. Wilson[1]

[1] Current or former employee of Agenus Inc., Lexington, MA, USA, or subsidiary thereof; [2]The Ludwig Institute for Cancer Research; [3]Memorial Sloan Kettering Cancer Center, New York, NY

agenus

Poster: #P312

SITC Annual Meeting

Washington, DC, USA • November 9-12, 2017

Abstract

PD-1 (or CD279) is a co-inhibitory receptor that suppresses T cell function upon binding to its ligands, PD-L1 or PD-L2. PD-1 signaling functions cooperatively with CTLA-4 to limit T cell activation during priming by antigen presenting cells, leading to reduced proliferation, cytokine and chemokine production and cell survival. Anti-PD-1 antibody therapies that block the interaction between PD-1 and its ligands have shown durable clinical benefit both as single agents, but particularly in combination with antibodies that antagonize CTLA-4.

AGEN2034, a novel human IgG4 anti-PD-1 antagonist antibody, potently inhibits PD-1 binding to PD-L1 and PD-L2, resulting in enhanced T cell responsiveness *in vitro* as well as in a non-human primate model. AGEN2034 combined effectively with AGEN1884, a human IgG1 anti-CTLA-4 antibody, anti-TIGIT or anti-LAG-3 to further enhance T cell responsiveness. Furthermore, the combination of AGEN2034 and anti-CTLA-4 blockade promoted a pharmacodynamic response in cynomolgus monkeys, including a transient increase in proliferation and ICOS (inducible co-stimulator molecule) expression in a subset of central memory and effector memory T cells.

AGEN2034 was well tolerated, and a no-observed-adverse-effect level (NOAEL) could be established up to 40 mg/kg in non-human primates. AGEN2034 is currently under evaluation in a Phase 1/2 study in subjects with advanced tumors and cervical cancer (NCT03104699) and clinical studies to evaluate AGEN2034 in combination with AGEN1884 are planned.

PD-1: A Key Pathway Of Immune Evasion

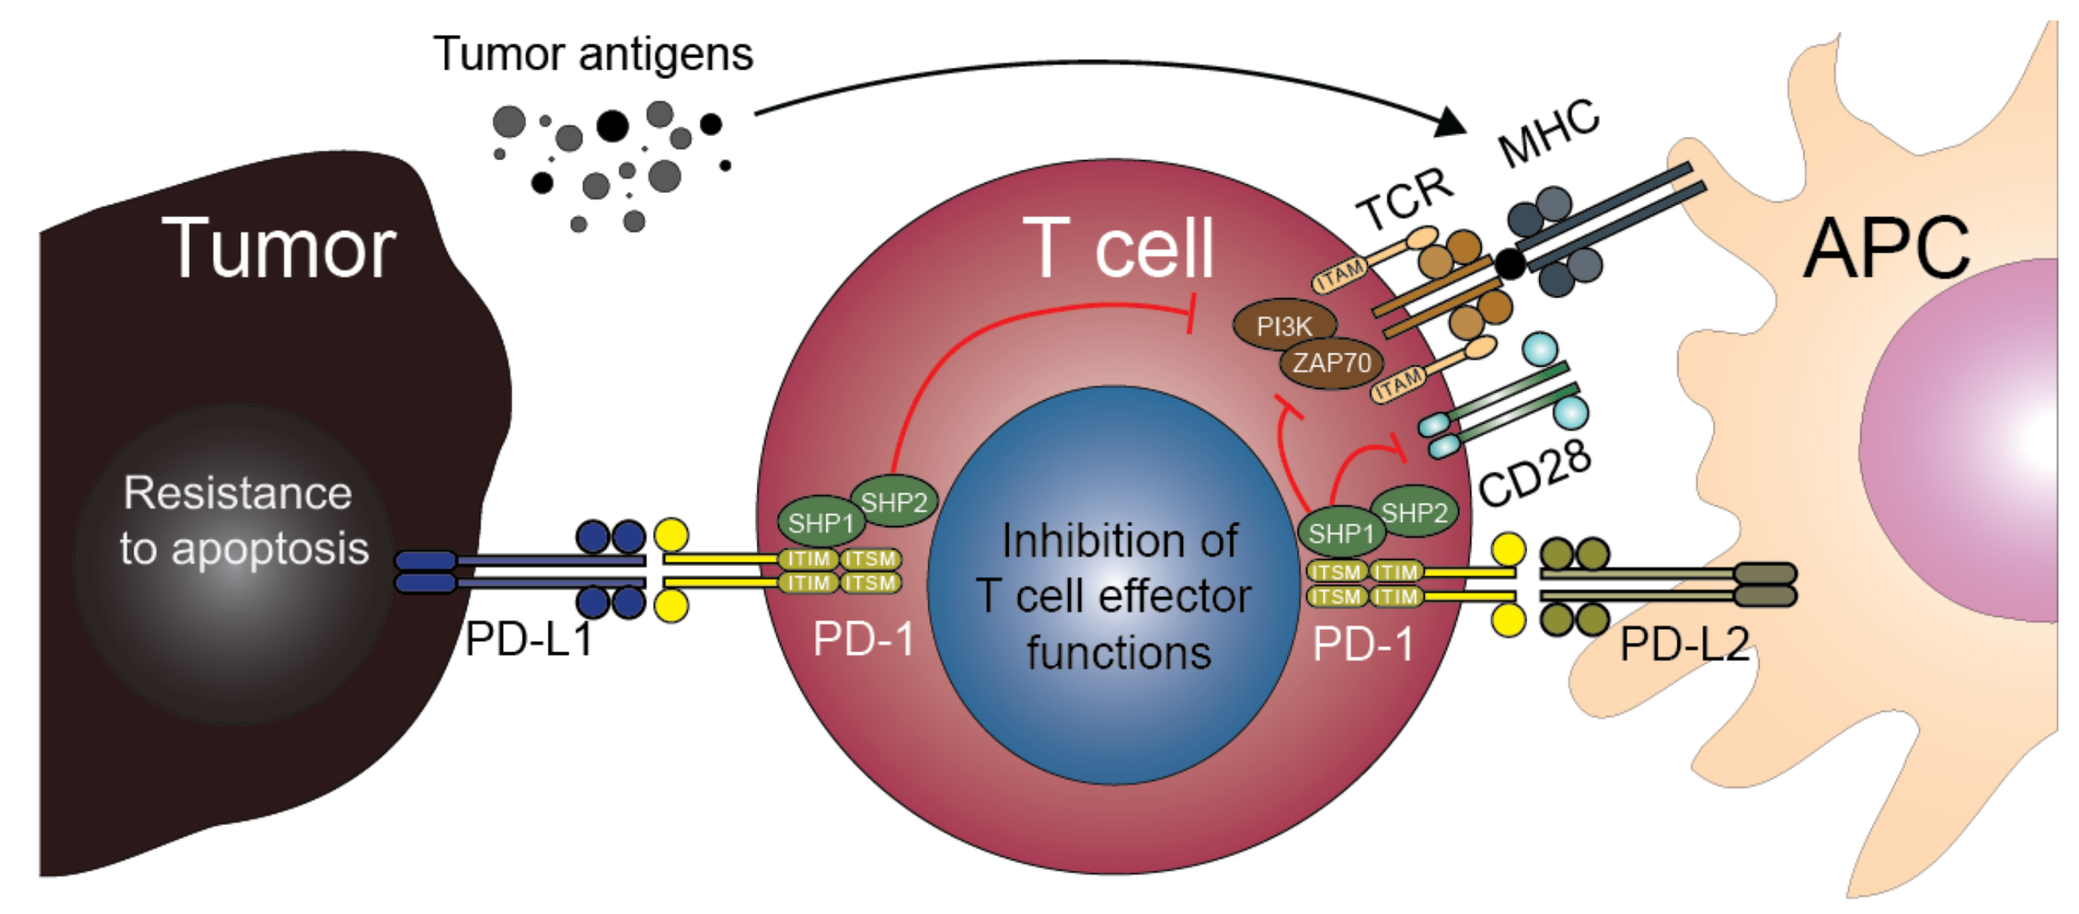

Tumor antigens
Tumor
Resistance to apoptosis
T cell
APC
TCR
MHC
PI3K
ZAP70
CD28
SHP1
SHP2
Inhibition of T cell effector functions
PD-L1
PD-1
PD-1
PD-L2

Kinetics of Expression

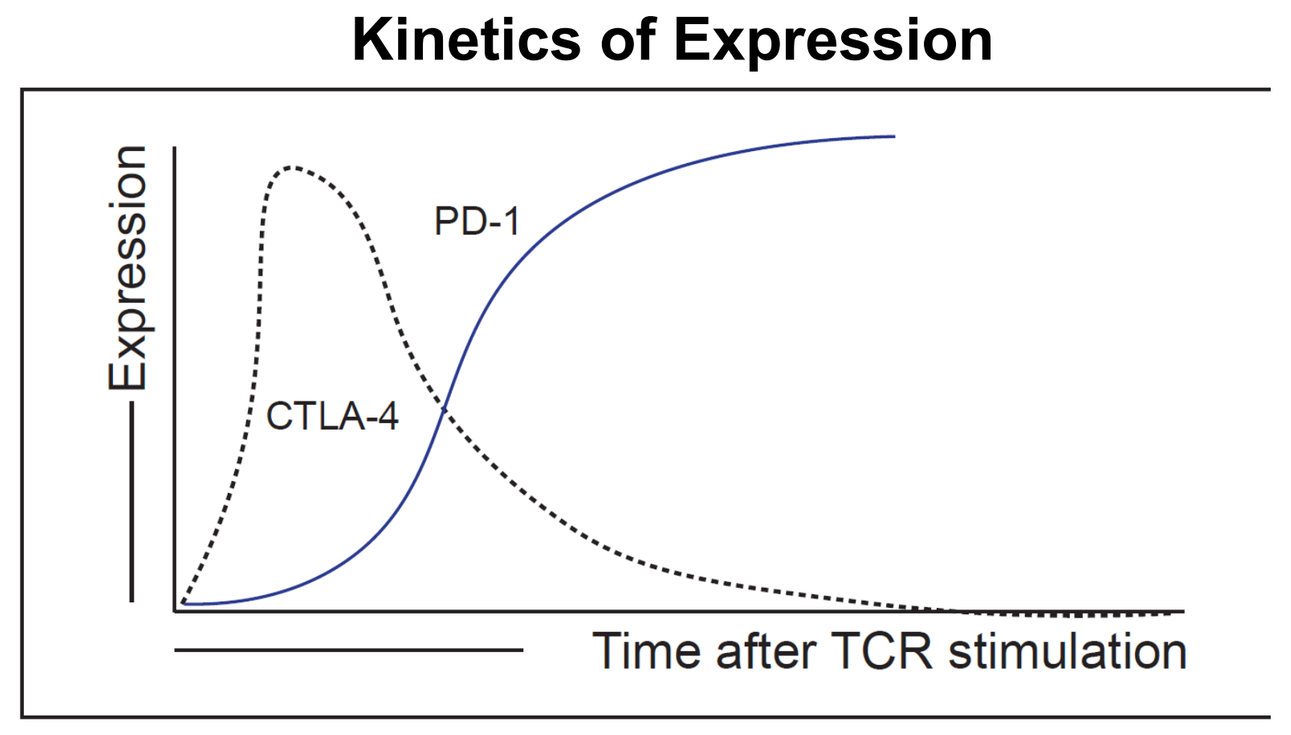

Expression
PD-1
CTLA-4
Time after TCR stimulation

Rationale Behind Anti-PD-1 And Anti-CTLA-4 Combination Therapy

Anti-PD-1 Blockade

Ligands: PD-L1 is expressed on immune cells, most normal tissues and tumor cells; PD-L2 is expressed on dendritic cells and macrophages

T cell expression: TILs commonly express elevated levels of PD-1 due to chronic tumor antigen stimulation

Anti-PD-1 blockade functions to restore antigen-specific T cell effector function mainly in the tumor microenvironment.

Anti-CTLA-4 Blockade

Ligands: CD80 (B7-1) and CD86 (B7-2) are expressed on professional APCs, but not on non-hematologic tumor cells

T cell expression: CTLA-4 is predominately expressed on CD4 "helper," not CD8 "killer" cells

Anti-CTLA-4 blockade is believed to enhance the priming phase of T cell activation mainly in lymphoid organs regardless of TCR clonality

Anti-PD-1 and anti-CTLA-4 combination therapy have demonstrated robust antitumor efficacy in preclinical mouse tumor models[a] and improved response rates in the clinic, such as in patients with metastatic melanoma[b], advanced small cell lung cancer (SCLC)[c] and metastatic renal cell carcinoma (RCC)[d]

Agenus Therapeutic Antibodies In Phase I / II Clinical Trials

	AGEN2034	AGEN1884
Target	PD-1	CTLA-4
Characterization	Fully human IgG4-S228P	Fully human IgG1
Discovery Platform	Retrocyte Display™	Retrocyte Display™
Mechanism of Action	Antagonist	Antagonist
Clinical Trial #	NCT03104699*	NCT02694822

**Clinical activity of AGEN2034 in subjects with metastatic or locally advanced solid tumors, with a consecutive Phase 2 expansion to evaluate efficacy in subjects with recurrent, unresectable, or metastatic (advanced) cervical cancer that has progressed after a platinum doublet.*

References

a. Curan MA. et al. PNAS. 2010; 107(9):4275-80.

b. Wolchok JD. et al. N Engl J Med 2017; 377:1345-1356.

c. Hellmann MD. *et al.* J Clin Oncol. 2017; 35:15 suppl. 8503.

d. Escudier B. *et al.* Checkmate 214: ESMO 2017 congress. Abstract LBA5

AGEN2034 Binds With High Affinity to Human and Cynomolgus Monkey PD-1

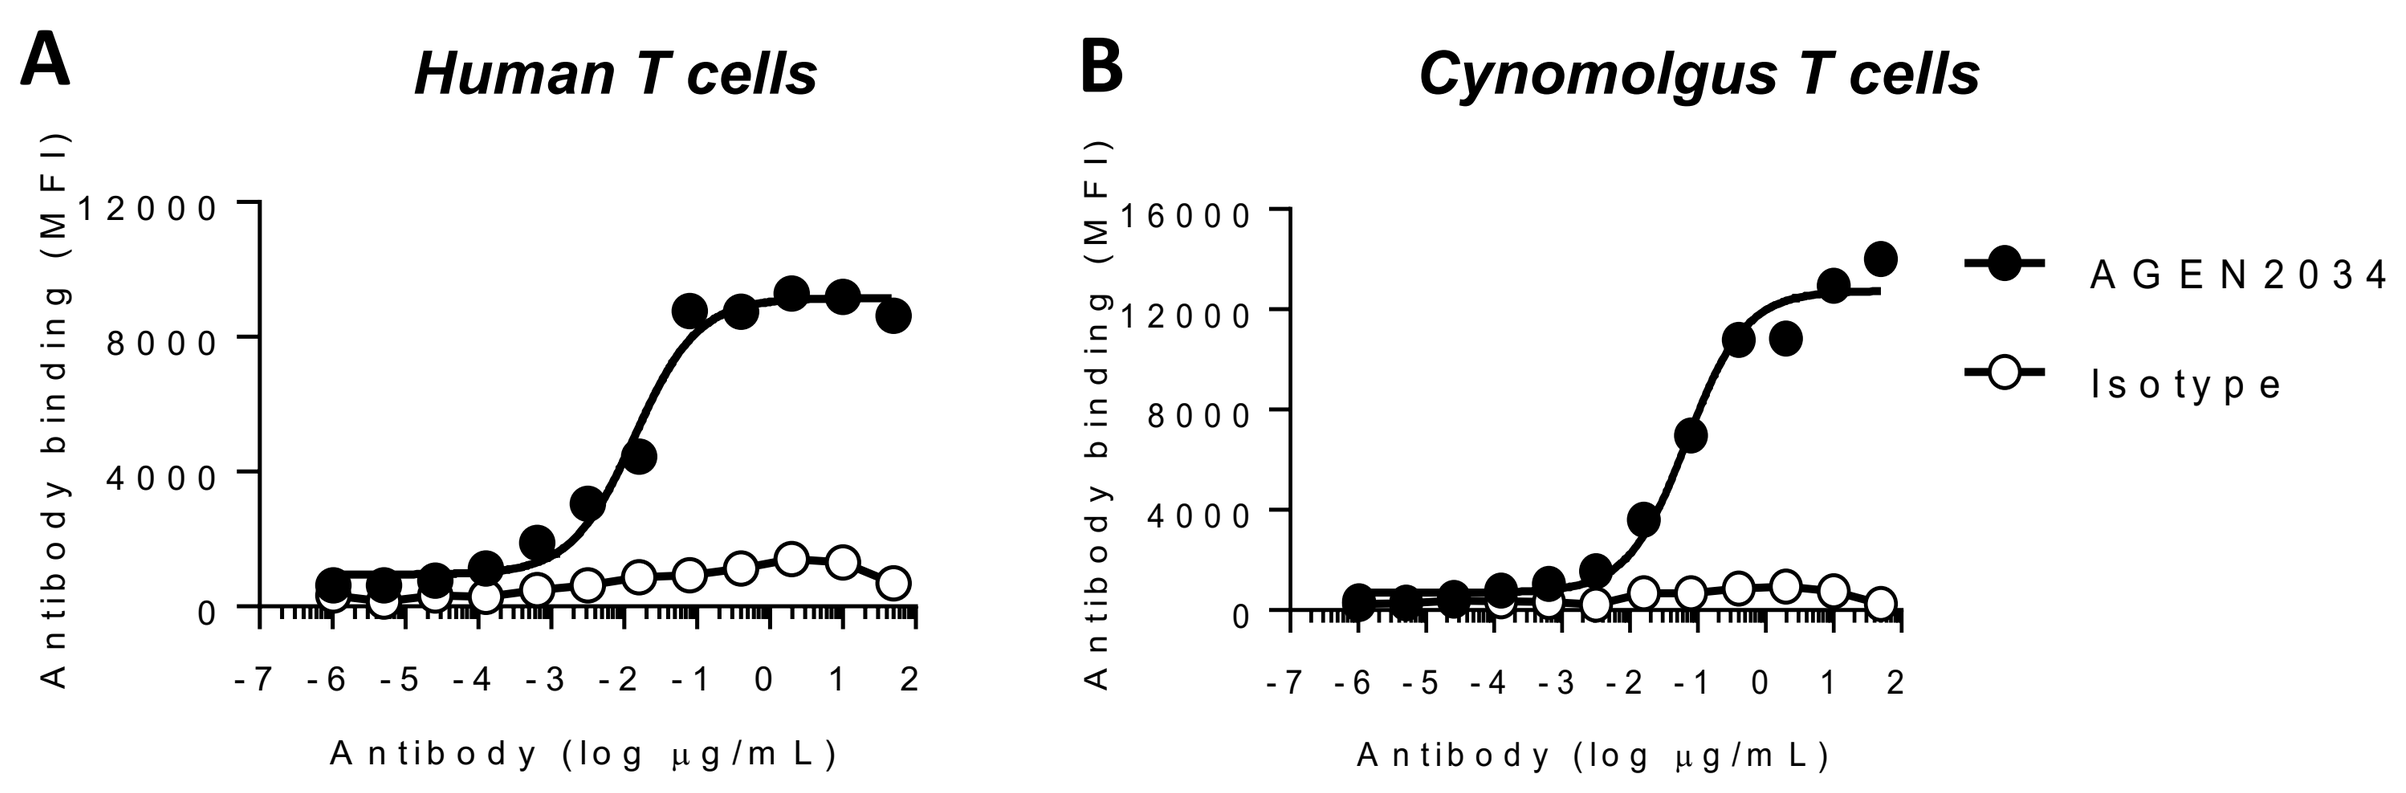

A
Human T cells
Antibody binding (MFI)
12000
8000
4000
0
-7 -6 -5 -4 -3 -2 -1 0 1 2
Antibody (log µg/mL)
B
Cynomolgus T cells
Antibody binding (MFI)
16000
12000
8000
4000
0
-7 -6 -5 -4 -3 -2 -1 0 1 2
Antibody (log µg/mL)
AGEN2034
Isotype

C

Binding Affinity of AGEN2034			
Ligand	**k_d (s^{-1})**	**k_a (Ms^{-1})**	**K_D (nM)**
Human PD-1	5.7×10^{-5}	4.1×10^{5}	0.14
Cyno PD-1	4.4×10^{-5}	3.8×10^{5}	0.12

***Legend**:* AGEN2034 binding to **(A)** activated human T cells and (**B**) activated cynomolgus T cells from human or cynomolgus peripheral blood mononuclear cells (PBMCs) stimulated with Staphylococcal enterotoxin A bacterial peptide for 5 days. Binding of increasing doses of AGEN2034 or isotype control antibody was assessed by flow cytometry. **(C)** Representative surface plasmon resonance (SPR) experiment for the binding of AGEN2034 to human PD-1-Fc or cynomolgus monkey PD-1-Fc recombinant proteins.

AGEN2034 Antagonizes Ligand Binding to PD-1 to Restore T cell Activation

A

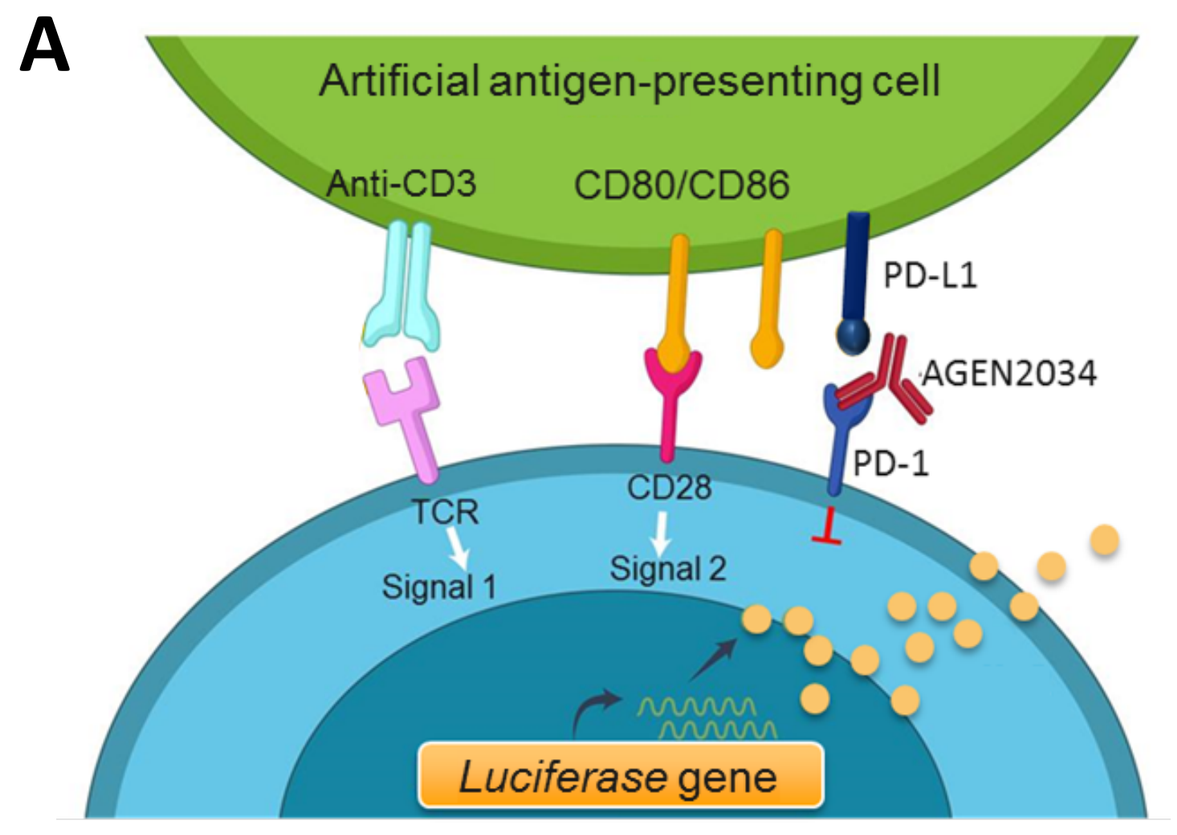

Artificial antigen-presenting cell
Anti-CD3
CD80/CD86
PD-L1
AGEN2034
PD-1
TCR
CD28
Signal 1
Signal 2
Luciferase gene

***Legend**:* (**A**) Illustration of reporter cell line assay. A human T cell line (Jurkat) were engineered to express PD-1 and a luciferase reporter gene driven by an NFAT promoter. Jurkat cells were incubated with Chinese hamster ovarian (CHO) cells engineered to express PD-L1 and a membrane-bound anti-CD3 antibody. (**B**) A dose titration of AGEN2034 or an IgG4 isotype control antibody were added to the T cell:CHO co-culture for 6 hours.

B

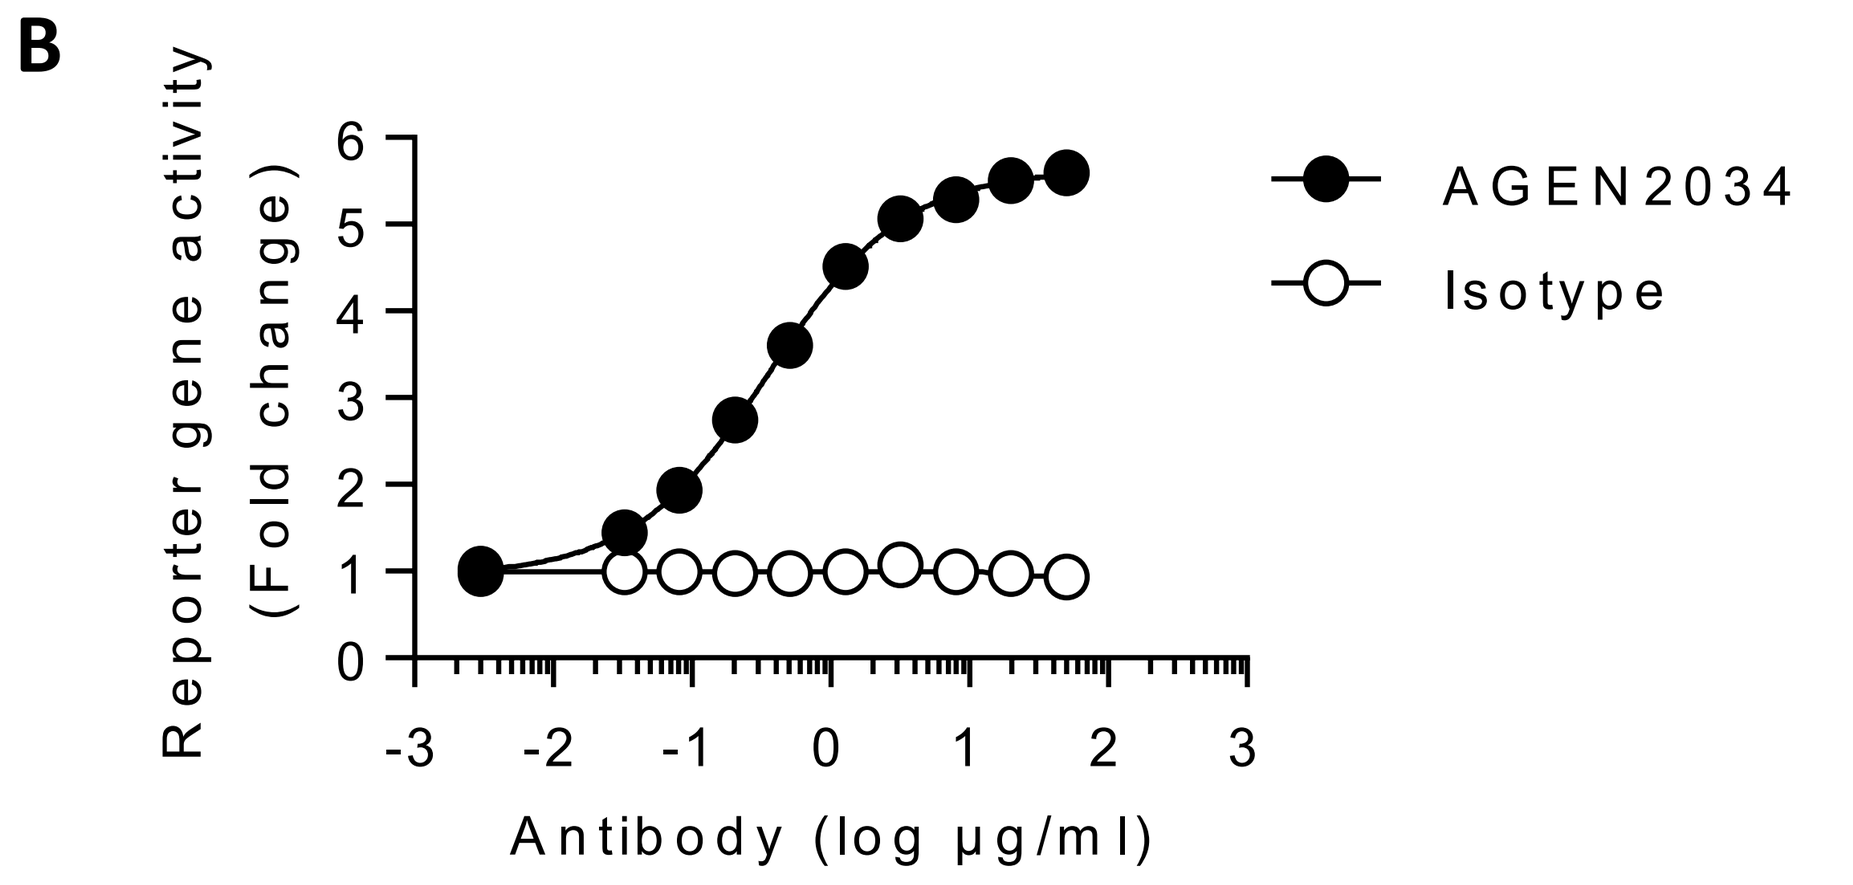

Reporter gene activity (Fold change)
6
5
4
3
2
1
0
-3 -2 -1 0 1 2 3
Antibody (log µg/ml)
AGEN2034
Isotype

AGEN2034 Does Not Activate FcγR Receptor IIIA Signaling, Consistent with an IgG4 Fc Region

A

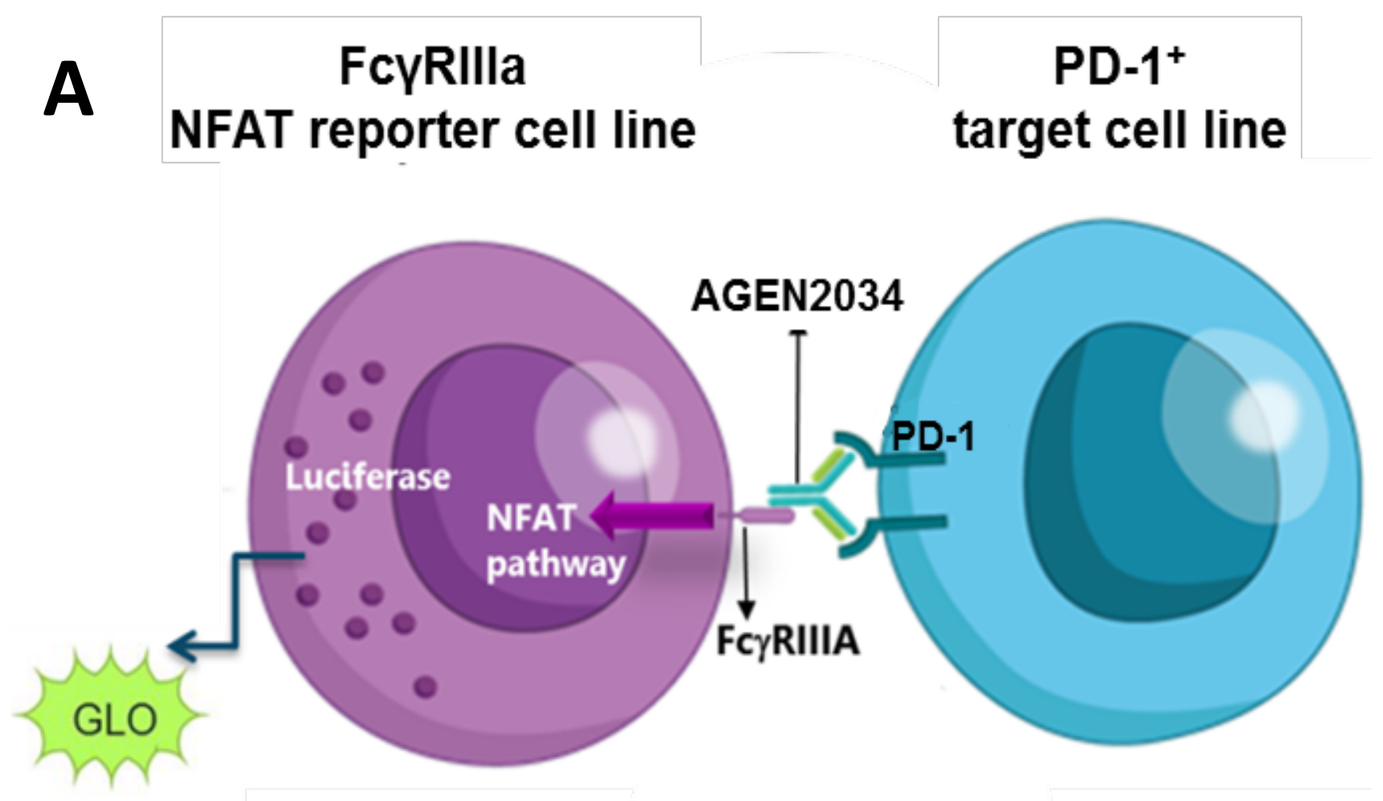

FcγRIIIa
NFAT reporter cell line
PD-1⁺
target cell line
AGEN2034
PD-1
Luciferase
NFAT pathway
FcγRIIIA
GLO

***Legend**:* **(A)** Illustration of antibody-mediated Fc receptor signaling assay. **(B)** FcγRIIIA signaling was evaluated by luciferase expression from a Jurkat cell line engineered to over express FcγRIIIA and an NFAT- luciferase reporter gene incubated in the presence of a dose titration of AGEN2034 IgG4 (black circles), AGEN2034 IgG1 (black squares), isotype IgG4 (white circles), and isotype IgG4 (white squares) antibodies and Jurkat cells expressing PD-1 target.

B

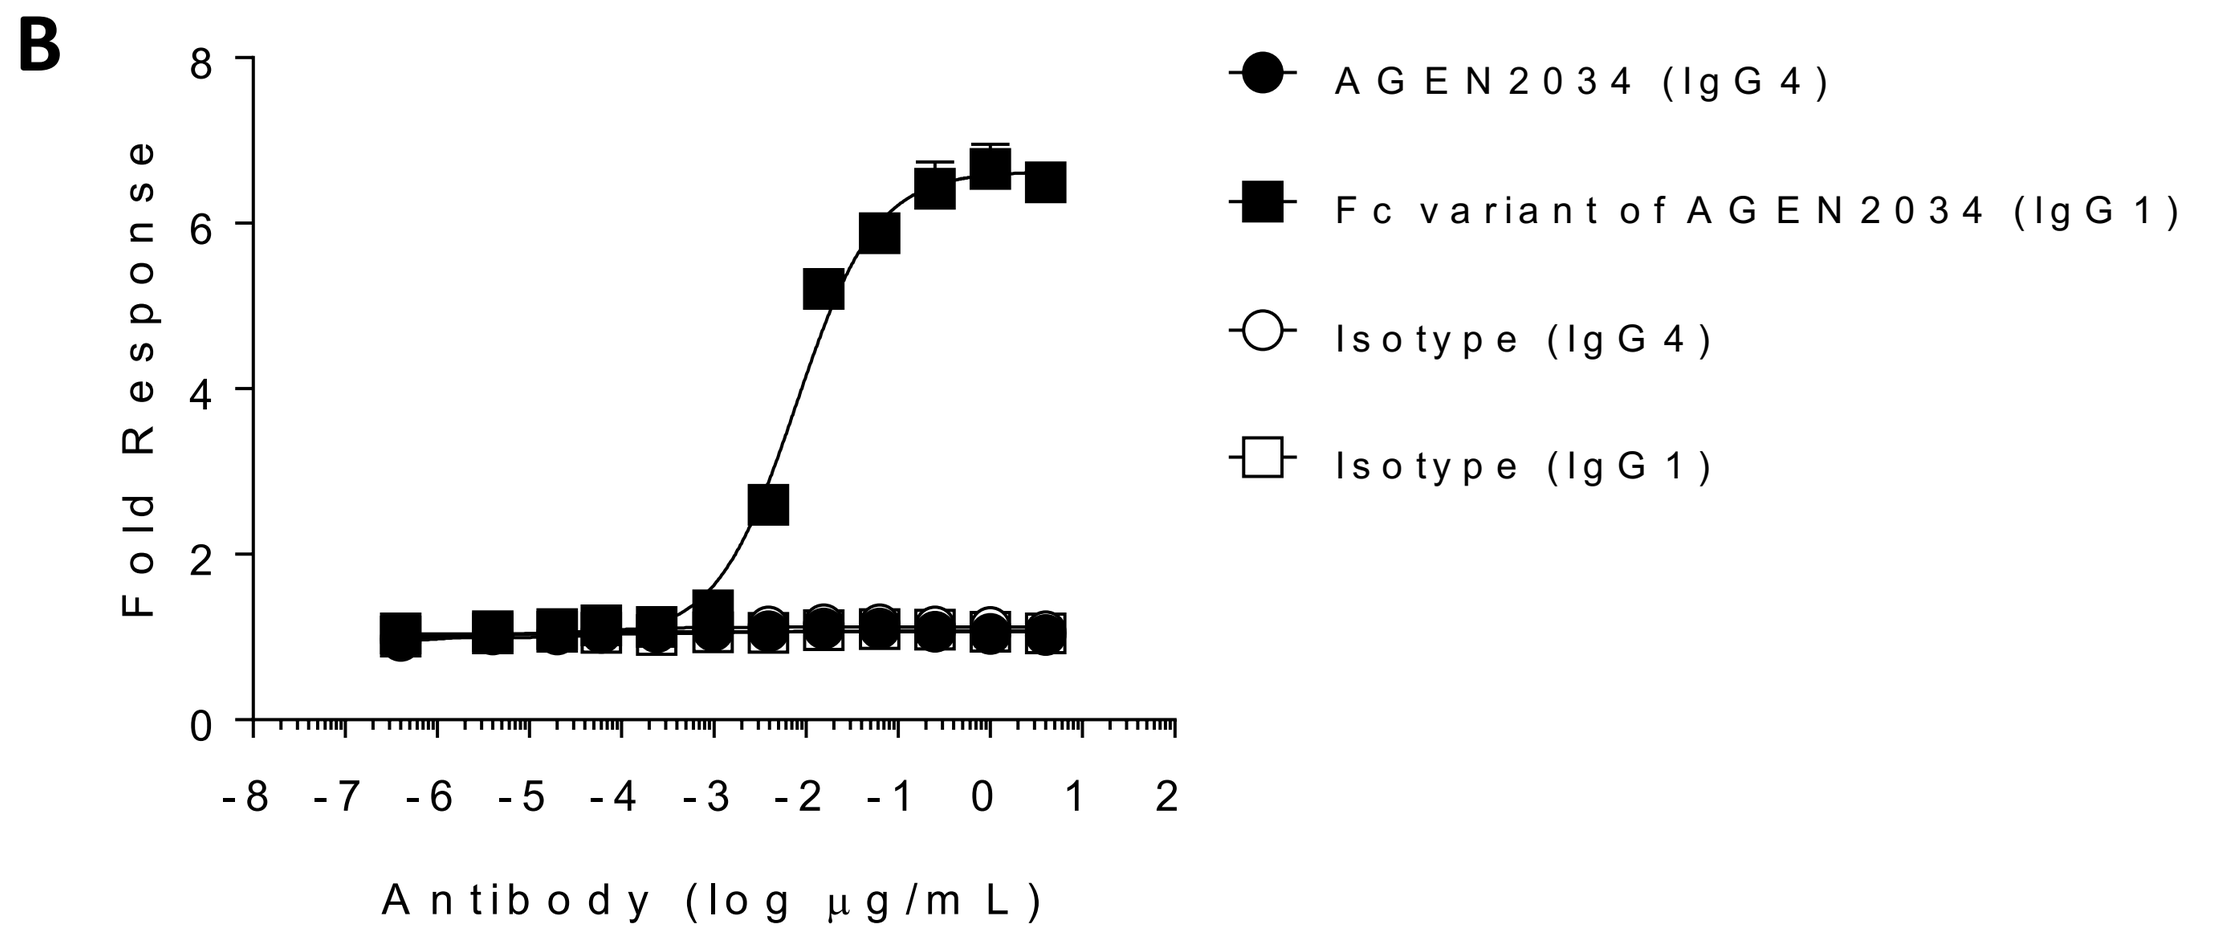

Fold Response
8
6
4
2
0
-8 -7 -6 -5 -4 -3 -2 -1 0 1 2
Antibody (log µg/mL)
AGEN2034 (IgG4)
Fc variant of AGEN2034 (IgG1)
Isotype (IgG4)
Isotype (IgG1)

AGEN2034 Enhances Primary Human T Cell Responsiveness

A

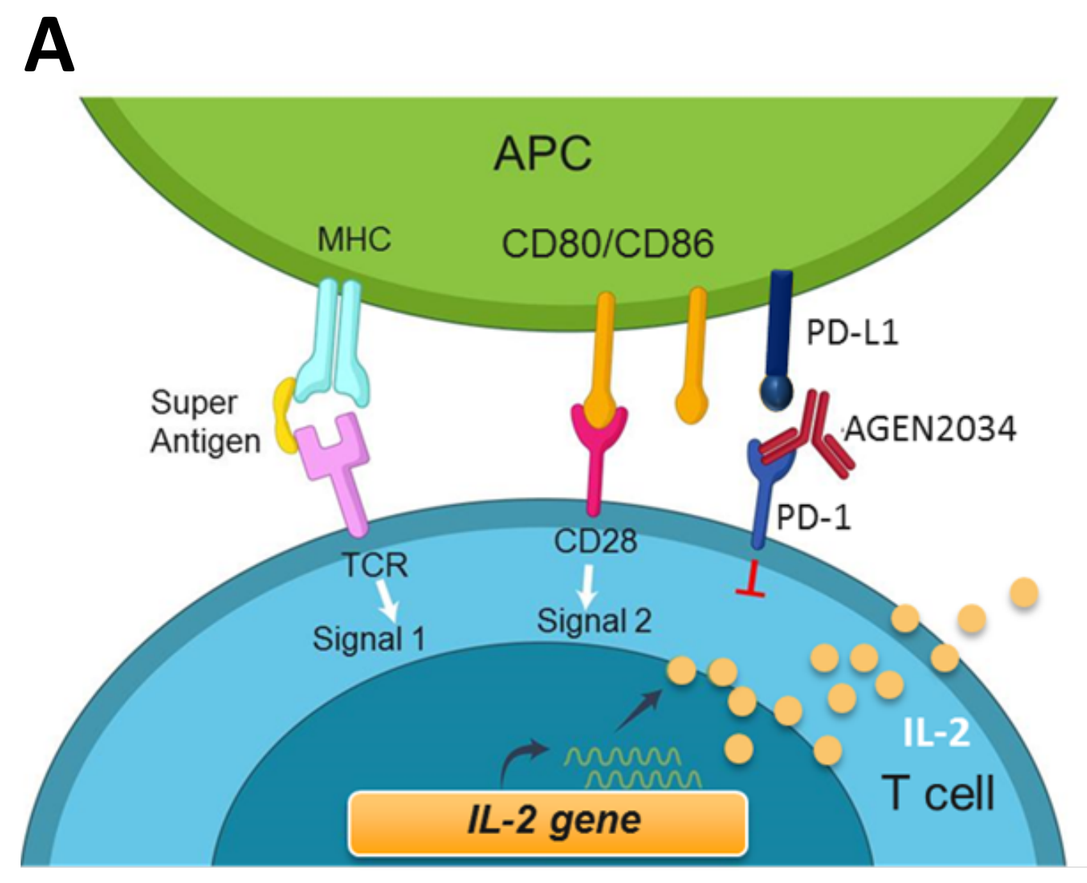

APC
MHC
CD80/CD86
PD-L1
Super Antigen
AGEN2034
TCR
CD28
PD-1
Signal 1
Signal 2
IL-2
T cell
IL-2 gene

B

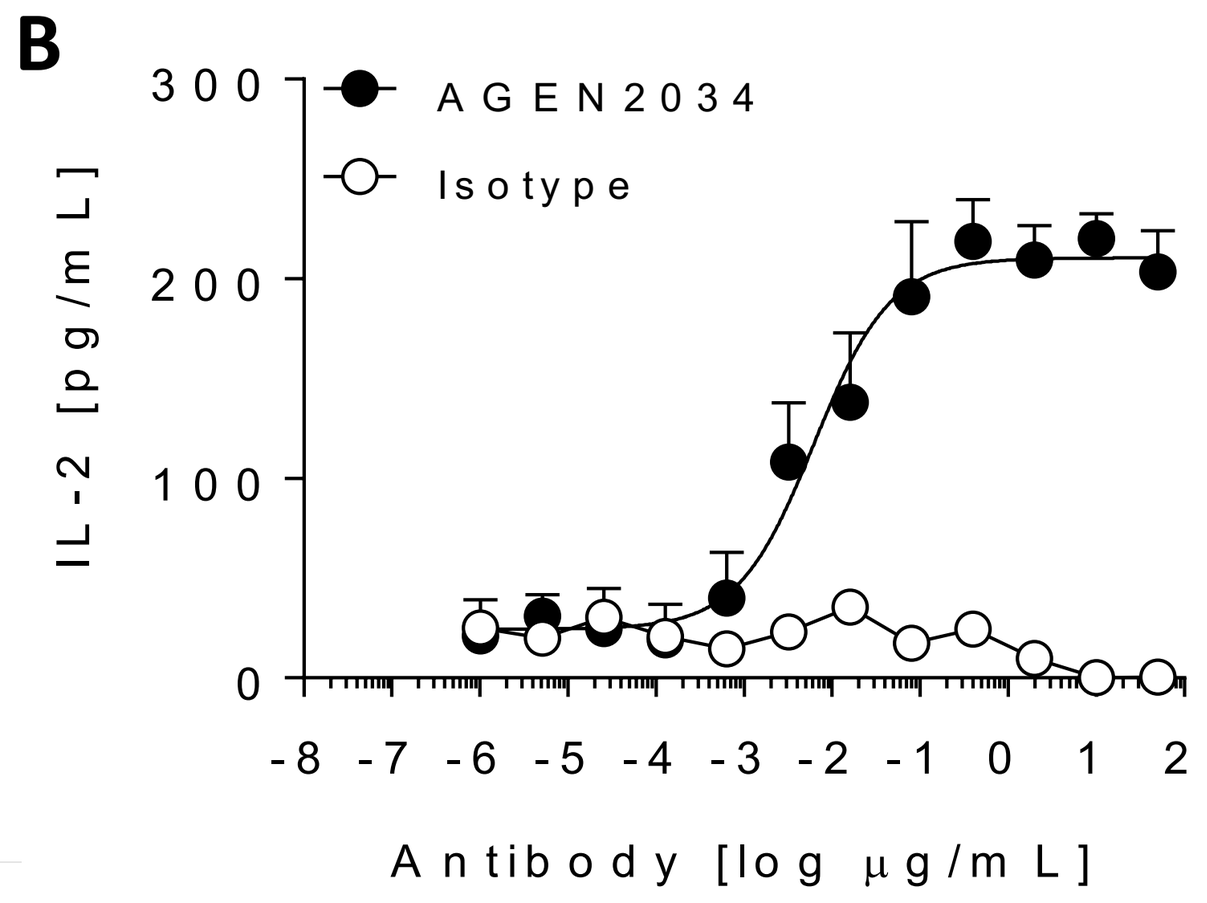

IL-2 [pg/mL]
300
200
100
0
-8 -7 -6 -5 -4 -3 -2 -1 0 1 2
Antibody [log µg/mL]
AGEN2034
Isotype

***Legend**:* **(A)** Illustration of primary cell stimulation assay. **(B)** Healthy human PBMCs were stimulated with Staphylococcal enterotoxin A peptide in the presence of dose titration of AGEN2034 or IgG4 isotype antibody for 5 days. Cytokine production (example: IL-2) was measured in the culture supernatant at day 5.

AGEN2034 Promotes T cell Proliferation and Reverses Ovarian Ascites-Mediated T cell Suppression

A

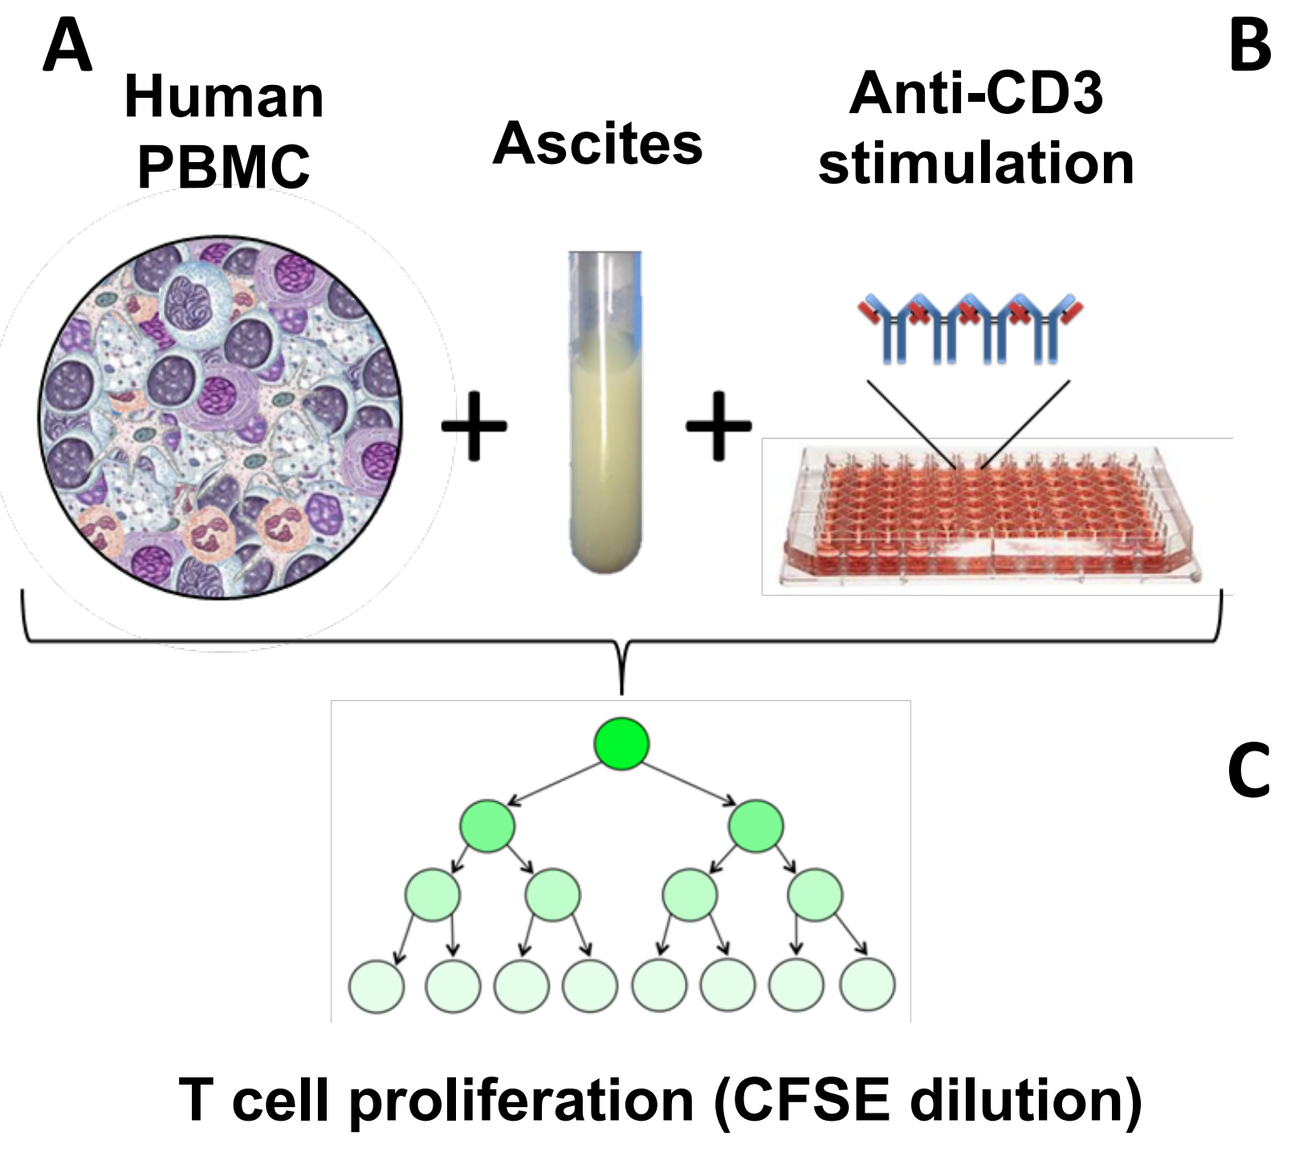

Human PBMC
Ascites
Anti-CD3 stimulation
T cell proliferation (CFSE dilution)

B

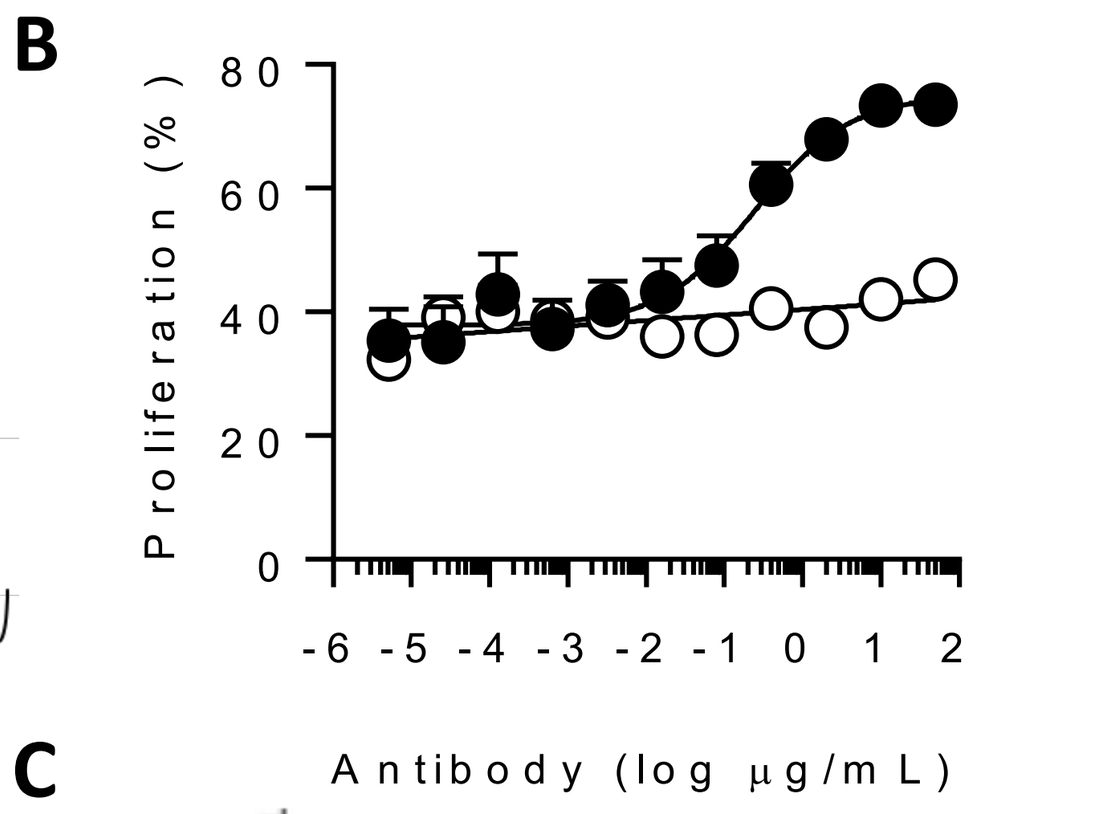

Proliferation (%)
80
60
40
20
0
-6 -5 -4 -3 -2 -1 0 1 2
Antibody (log µg/mL)

C

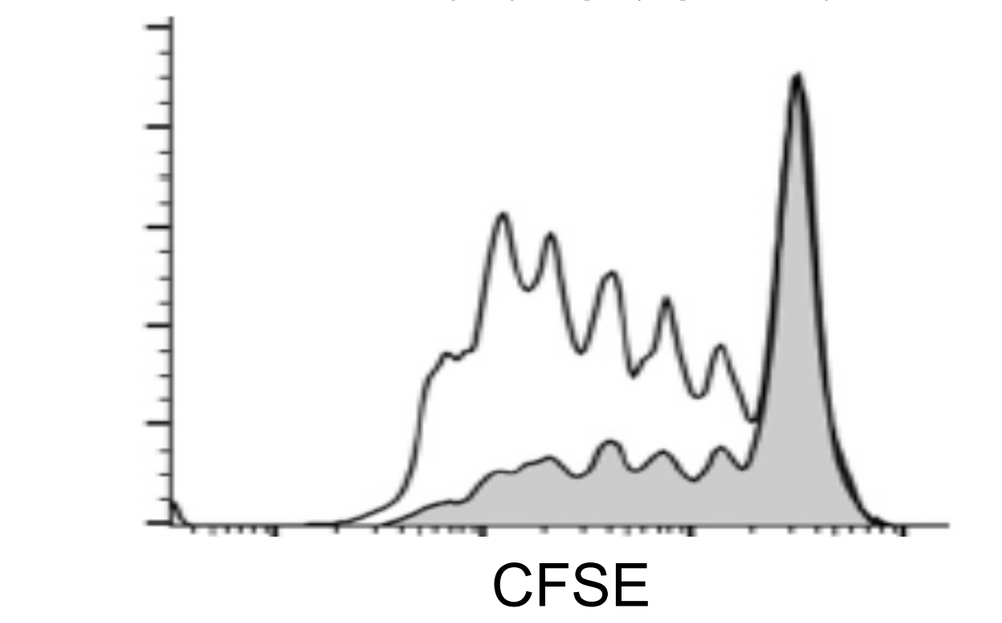

CFSE

Legend: (**A**) Illustration of primary cell suppression assay. (**B**) The percentage of CD4+ CFSE low (*i.e.* proliferated T cells) after 4 days in PBMC cultures stimulated with plate-bound anti-CD3 antibody, a fixed concentration of human ovarian cancer ascites, and a dose titration of AGEN2034 or an IgG4 isotype antibody. **(C)** Representative histogram for CFSE dilution at 10 µg/mL of AGEN2034 (gray) or isotype antibody (white) treated cultures is shown.

AGEN2034 Effectively combines with AGEN1884 To Further Enhance Cytokine Secretion

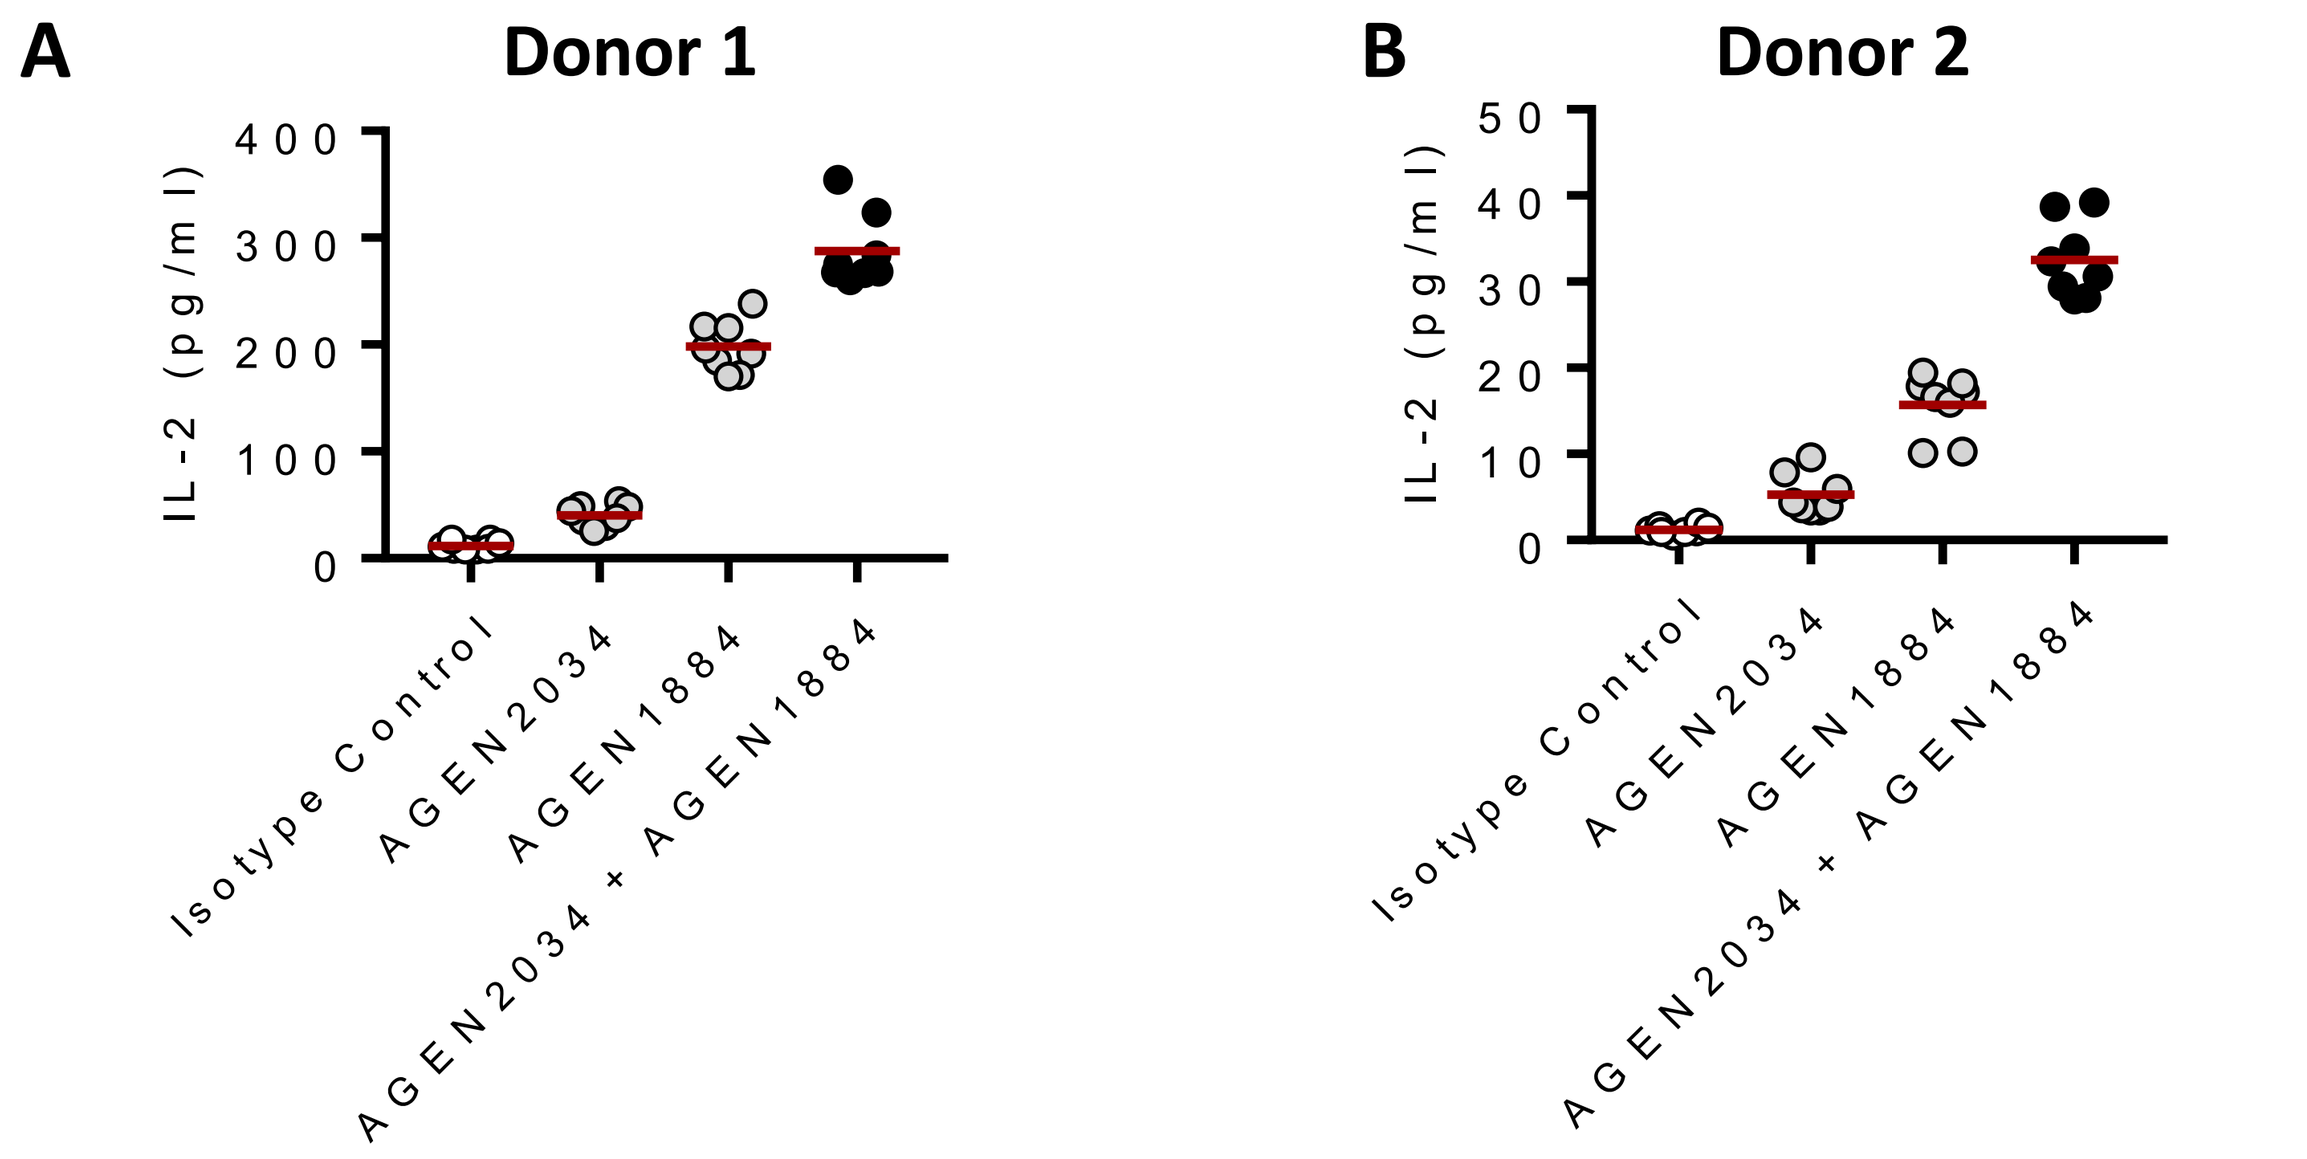

A
Donor 1
IL-2 (pg/ml)
400
300
200
100
0
Isotype Control
AGEN2034
AGEN1884
AGEN2034 + AGEN1884
B
Donor 2
IL-2 (pg/ml)
50
40
30
20
10
0
Isotype Control
AGEN2034
AGEN1884
AGEN2034 + AGEN1884

Legend: Human PBMCs from two healthy donors were stimulated with Staphylococcal enterotoxin A peptide in the presence of AGEN2034 or IgG4 isotype antibody (10µg/ml) alone or in combination with anti-CTLA-4, AGEN1884, antibody (10µg/mL) for 5 days. Cytokine production (example: IL-2) was measured in the culture supernatant at day 5.

AGEN2034 Cooperates with other immunomodulatory antibodies to Enhance T cell Responsiveness

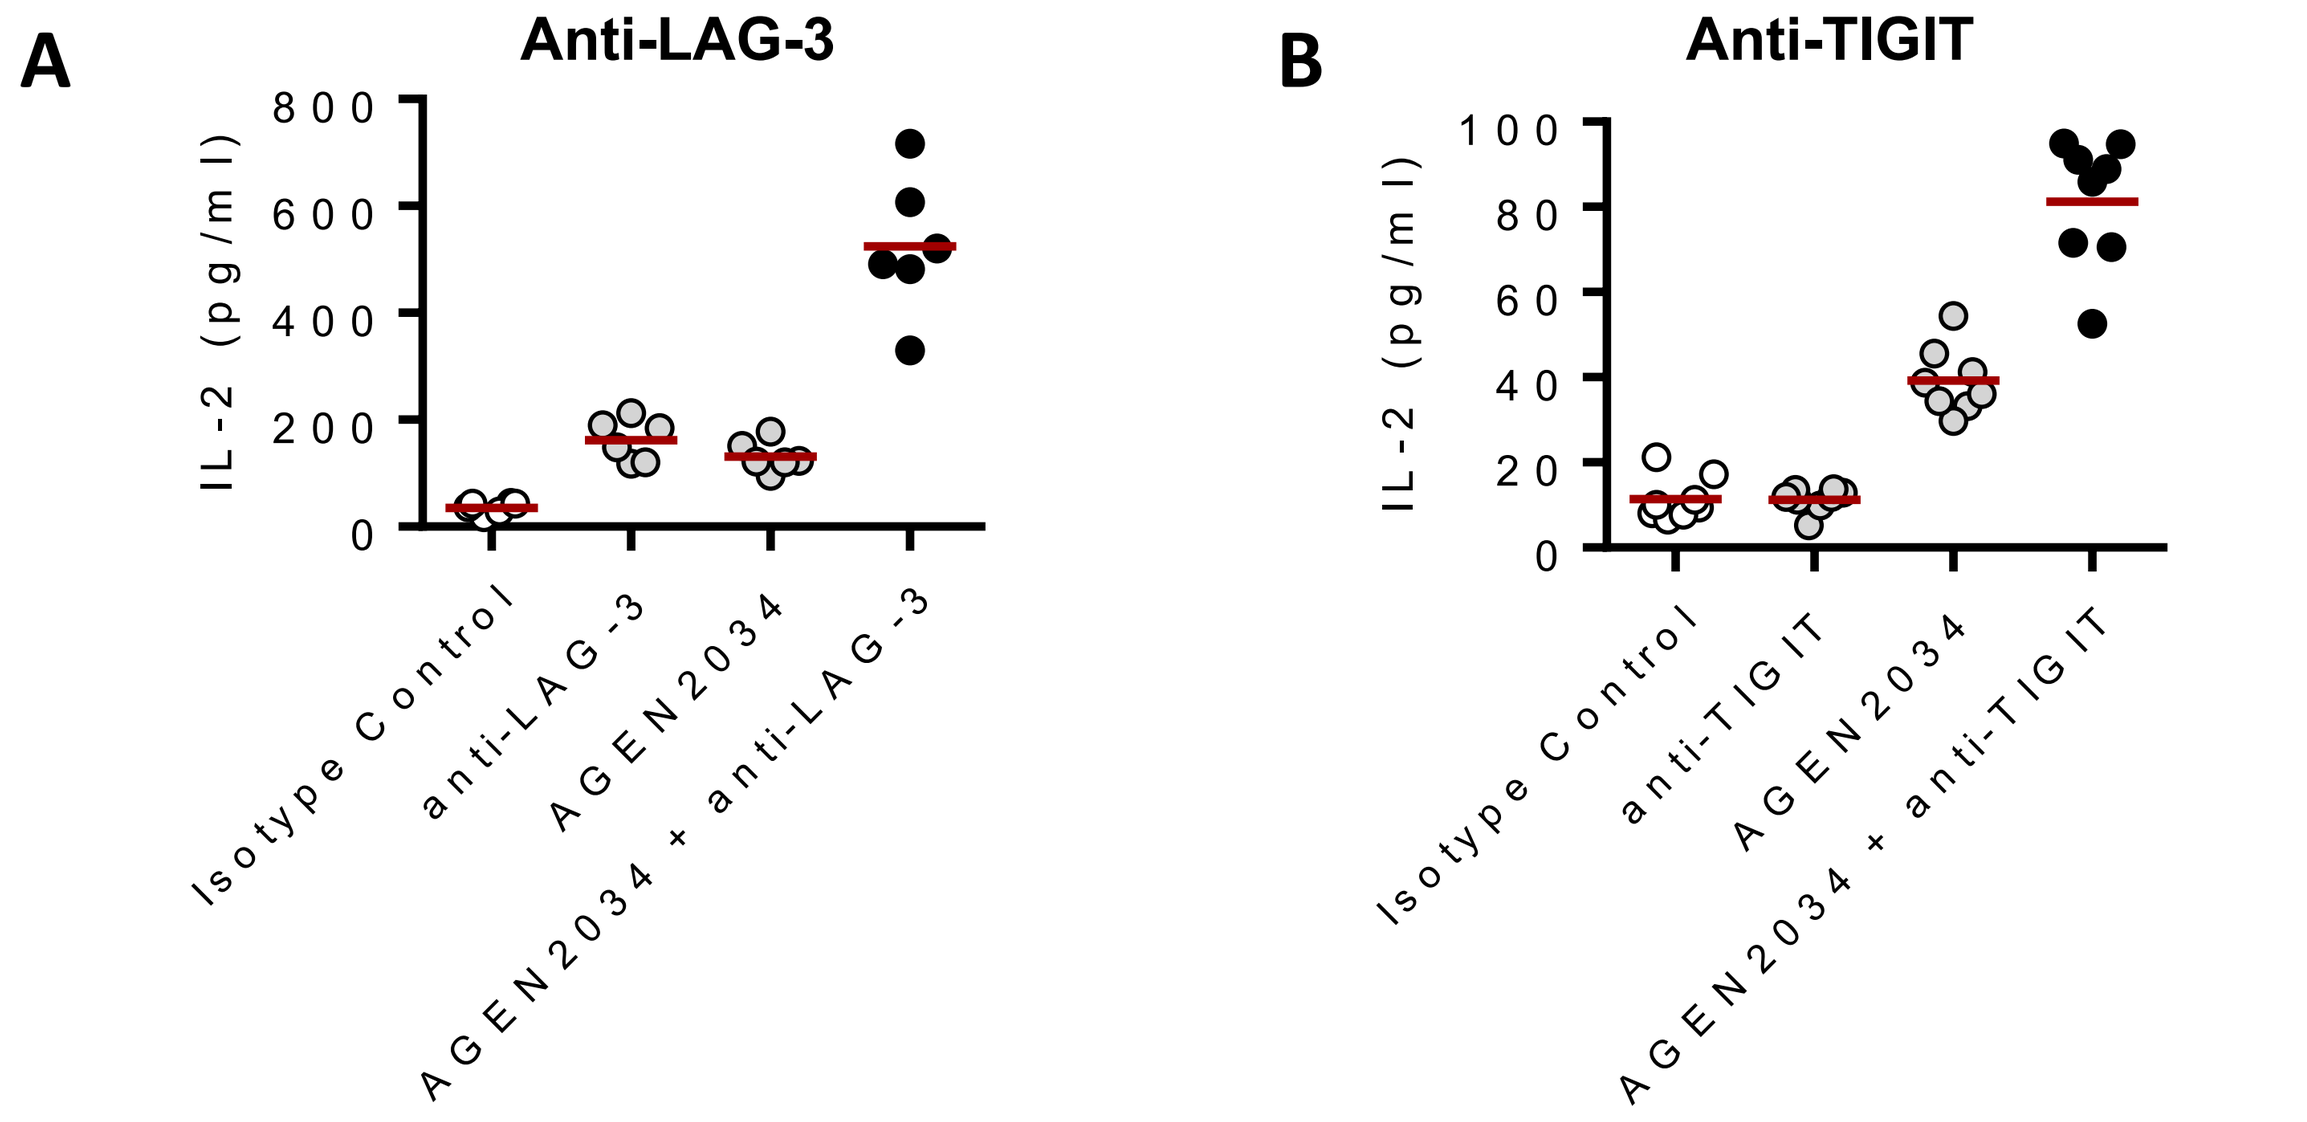

A
Anti-LAG-3
IL-2 (pg/ml)
800
600
400
200
0
Isotype Control
anti-LAG-3
AGEN2034
AGEN2034 + anti-LAG-3
B
Anti-TIGIT
IL-2 (pg/ml)
100
80
60
40
20
0
Isotype Control
anti-TIGIT
AGEN2034
AGEN2034 + anti-TIGIT

Legend: **(A)** Human PBMCs were stimulated with Staphylococcal enterotoxin A peptide in the presence of AGEN2034 or IgG4 isotype antibody (10µg/ml) alone or in combination with anti-LAG-3 **(A)** or anti-TIGIT **(B)** antibodies (10µg/mL) for 5 days. Cytokine production (example: IL-2) was measured in the culture supernatant at day 5.

AGEN2034 Combines With Anti-CTLA-4 To Enhance Activation and Proliferation of Central Memory T cells In Non-human Primates

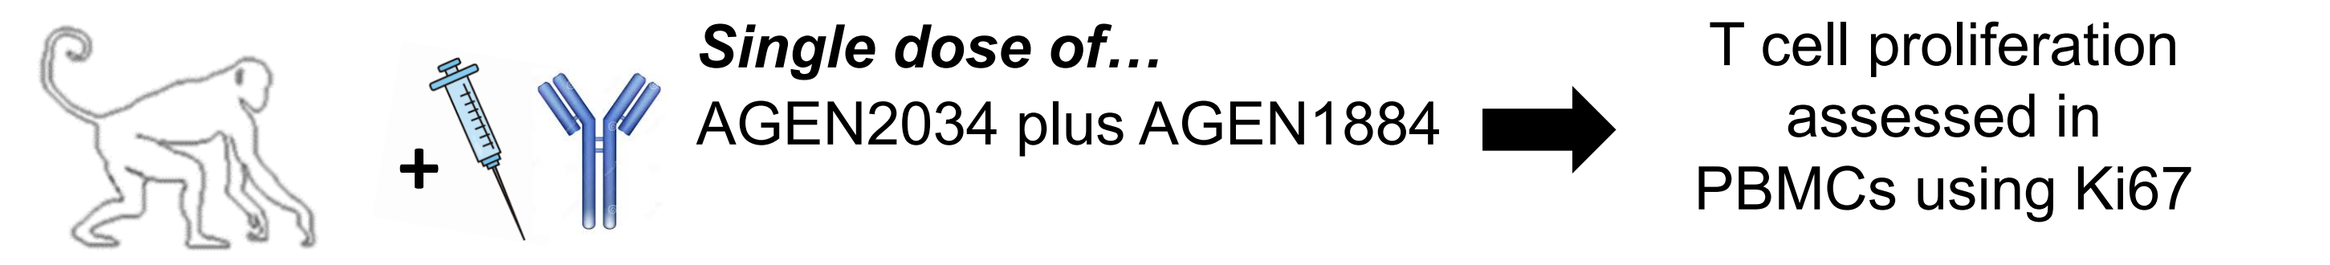

Single dose of...
AGEN2034 plus AGEN1884
T cell proliferation assessed in PBMCs using Ki67

A

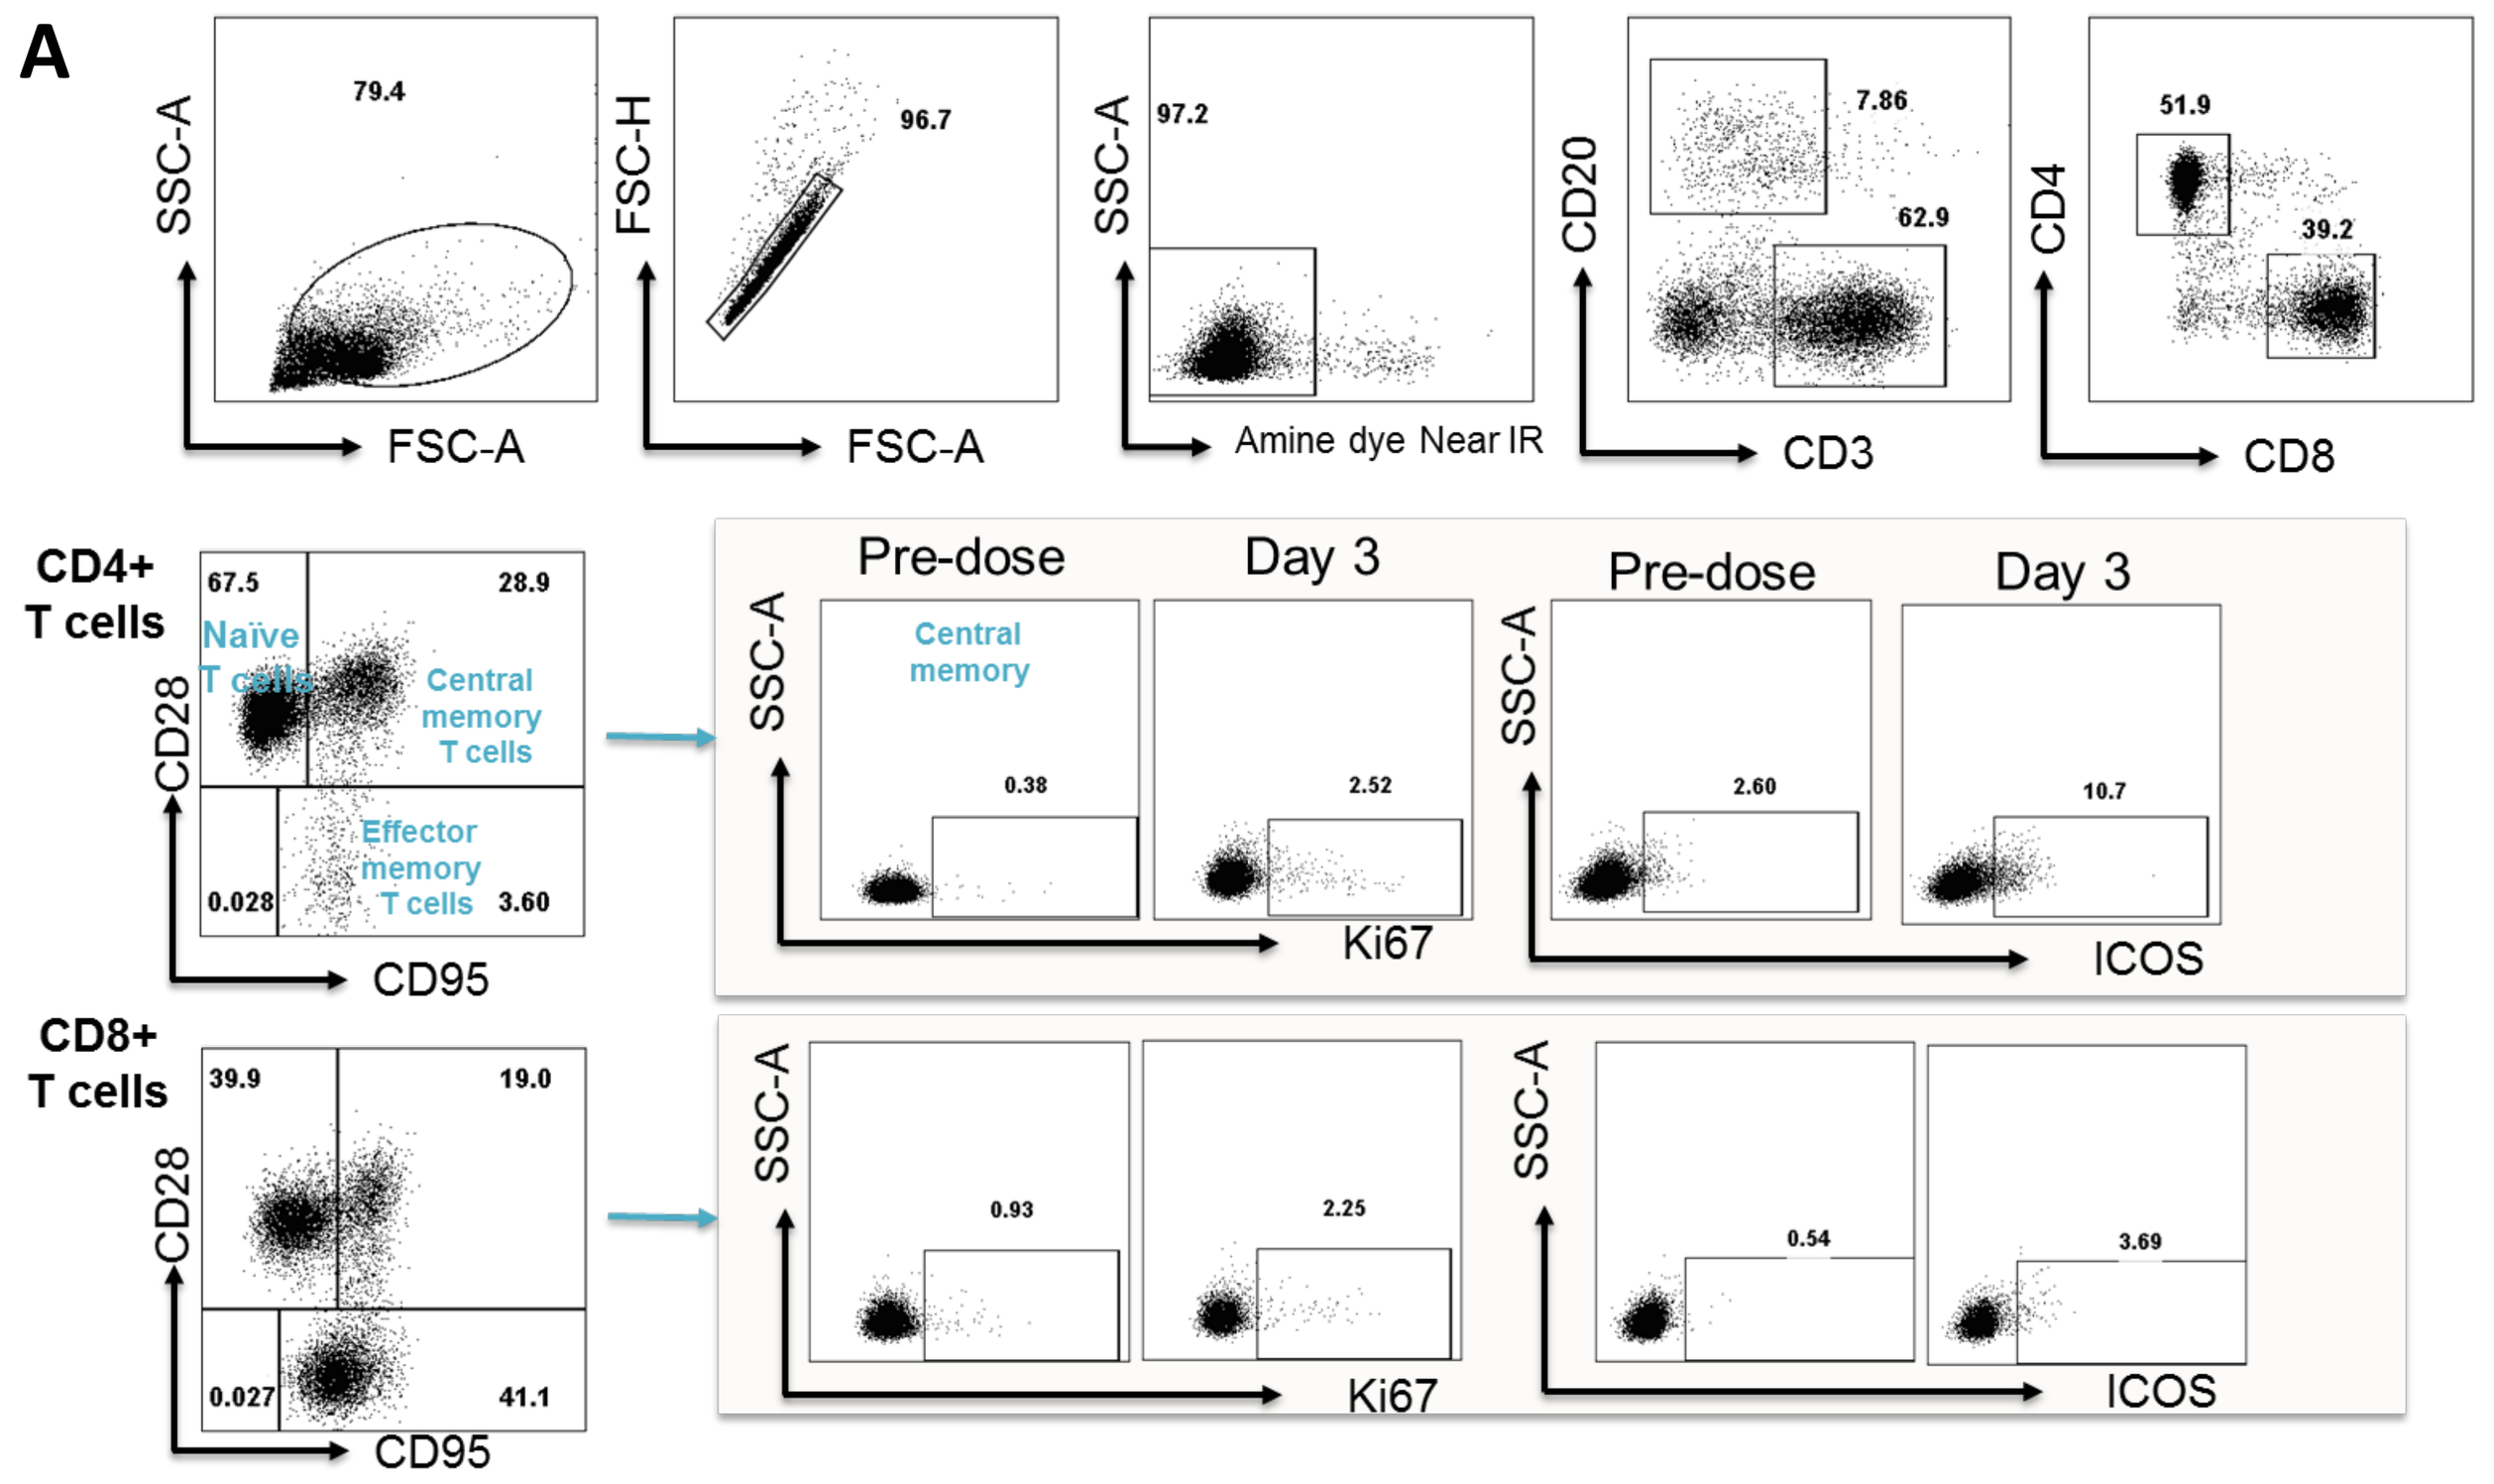

SSC-A
FSC-A
FSC-H
Amine dye Near IR
CD20
CD3
CD4
CD8
CD4+ T cells
CD8+ T cells
CD28
CD95
Naïve T cells
Central memory T cells
Effector memory T cells
Pre-dose
Day 3
Central memory
Ki67
ICOS

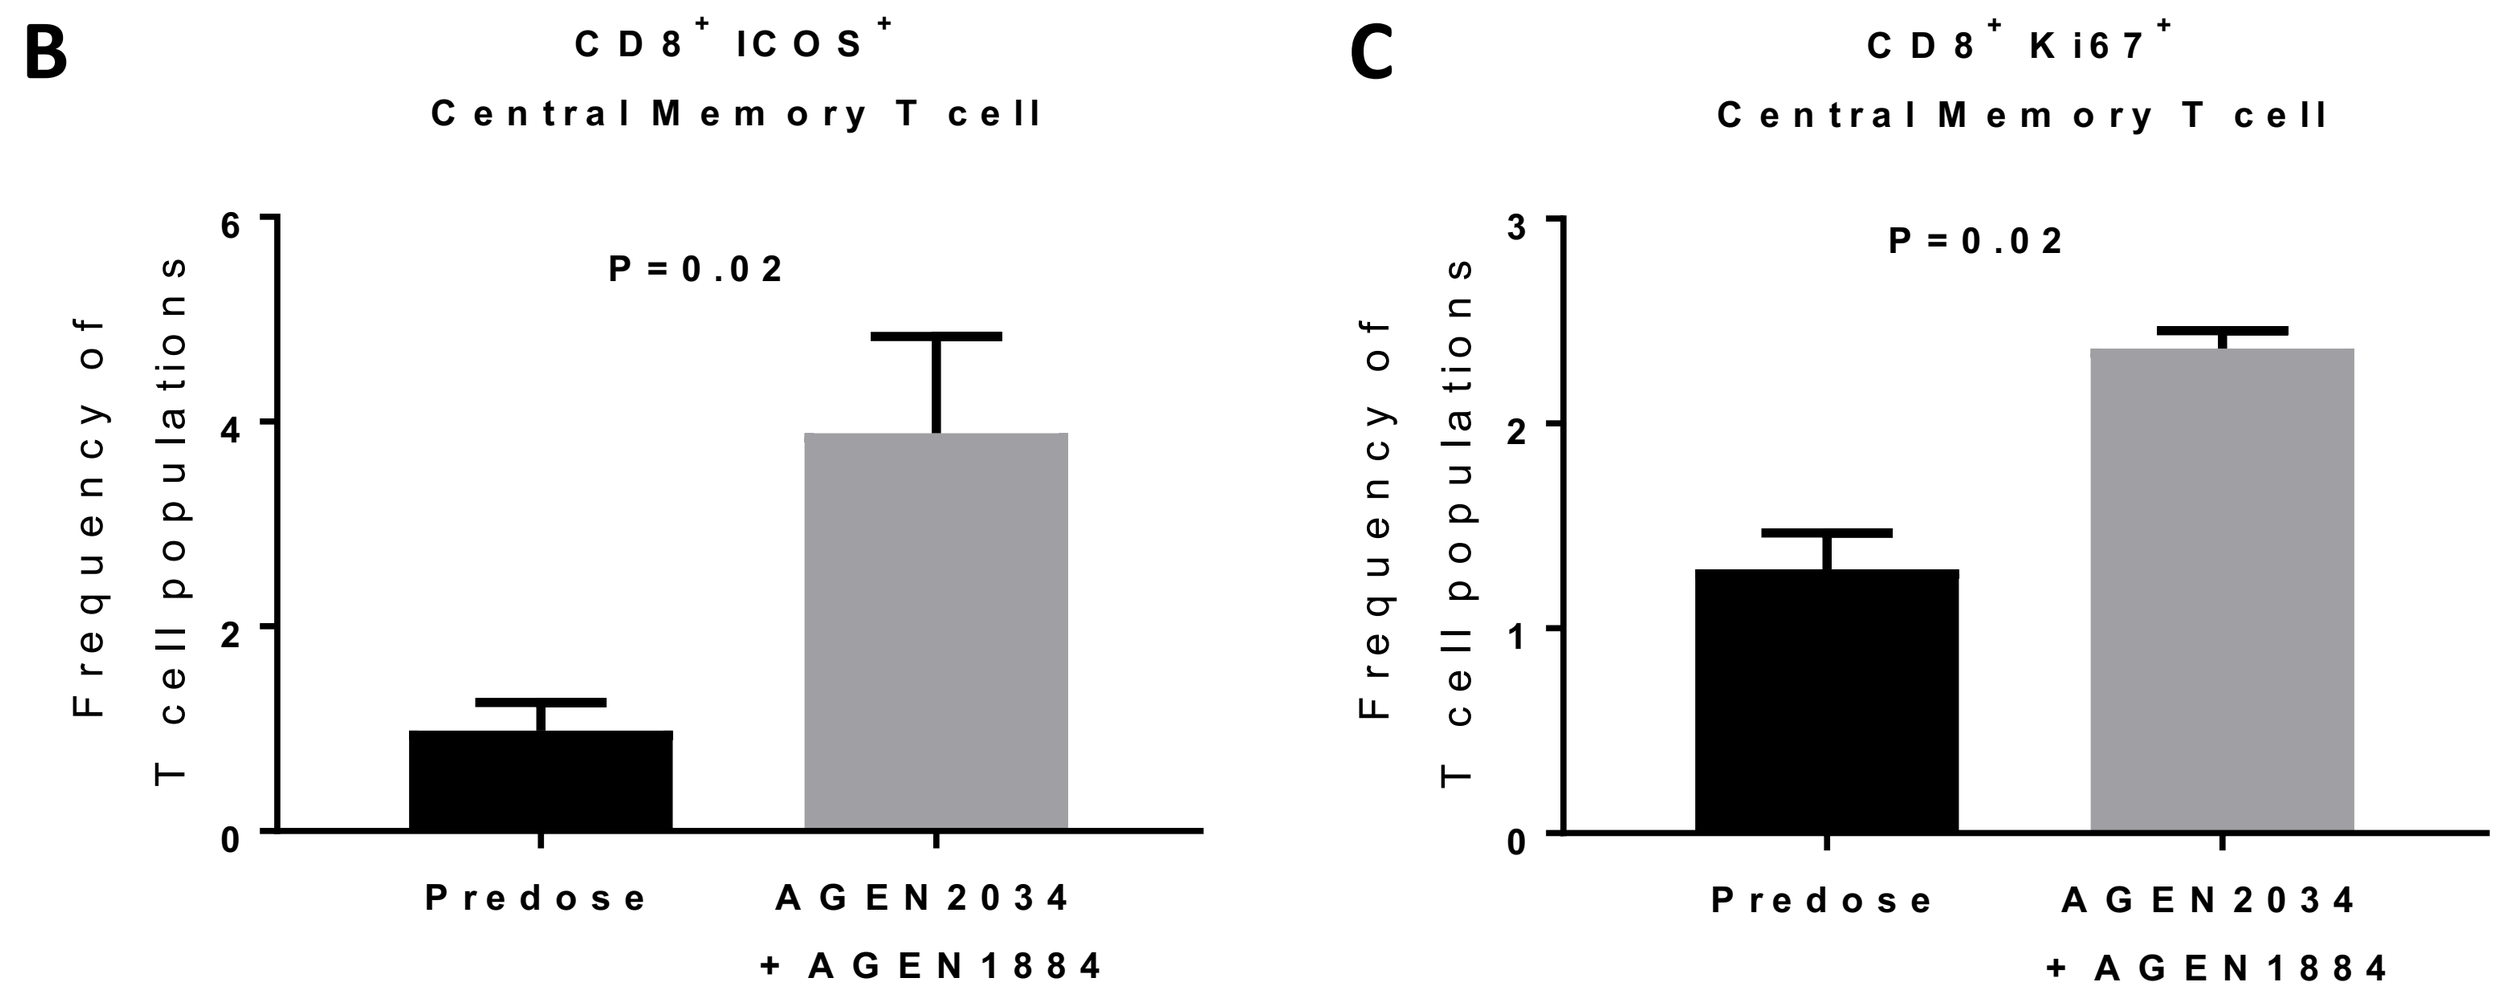

B
CD8⁺ ICOS⁺
Central Memory T cell
Frequency of T cell populations
P = 0.02
Predose
AGEN2034 + AGEN1884
C
CD8⁺ Ki67⁺
Central Memory T cell
Frequency of T cell populations
P = 0.02
Predose
AGEN2034 + AGEN1884

Legend: Cynomolgus monkeys were injected once with 10 mg/kg of AGEN1884 in combination with 3 mg/kg of AGEN2034. Blood samples were collected bi-weekly for 30 days and analyzed by flow cytometry. **(A)** Representative data for the frequencies of CD4⁺ and CD8⁺ naïve, central memory, and effector memory T cells are shown. Frequency of CD8⁺ ICOS⁺ **(B)** and CD8⁺ Ki67⁺ **(C)** central memory T cells 3 days following antibody treatment (n=4 animals).

AGEN2034 Is Safe And Well-Tolerated

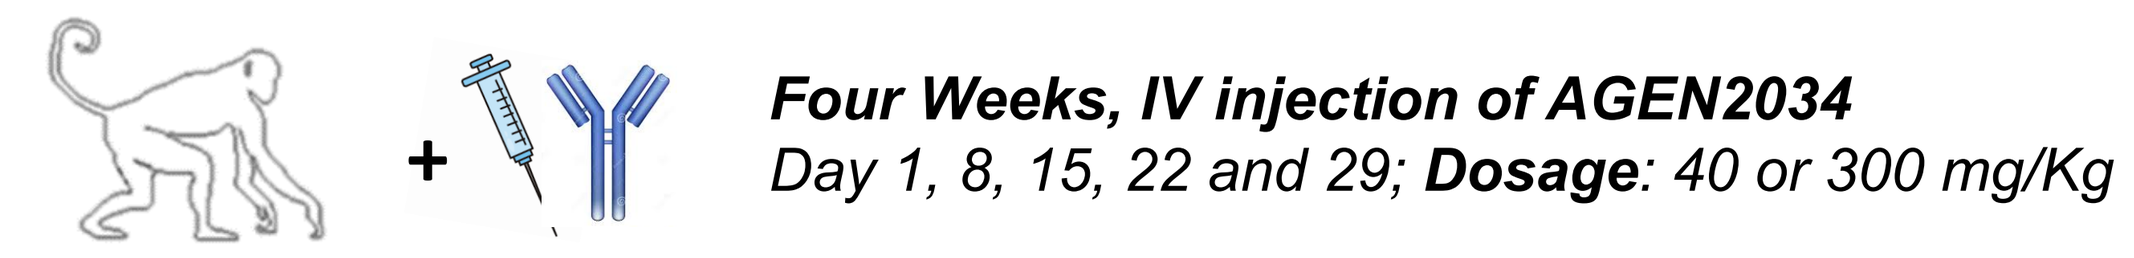

Four Weeks, IV injection of AGEN2034
Day 1, 8, 15, 22 and 29; Dosage: 40 or 300 mg/Kg

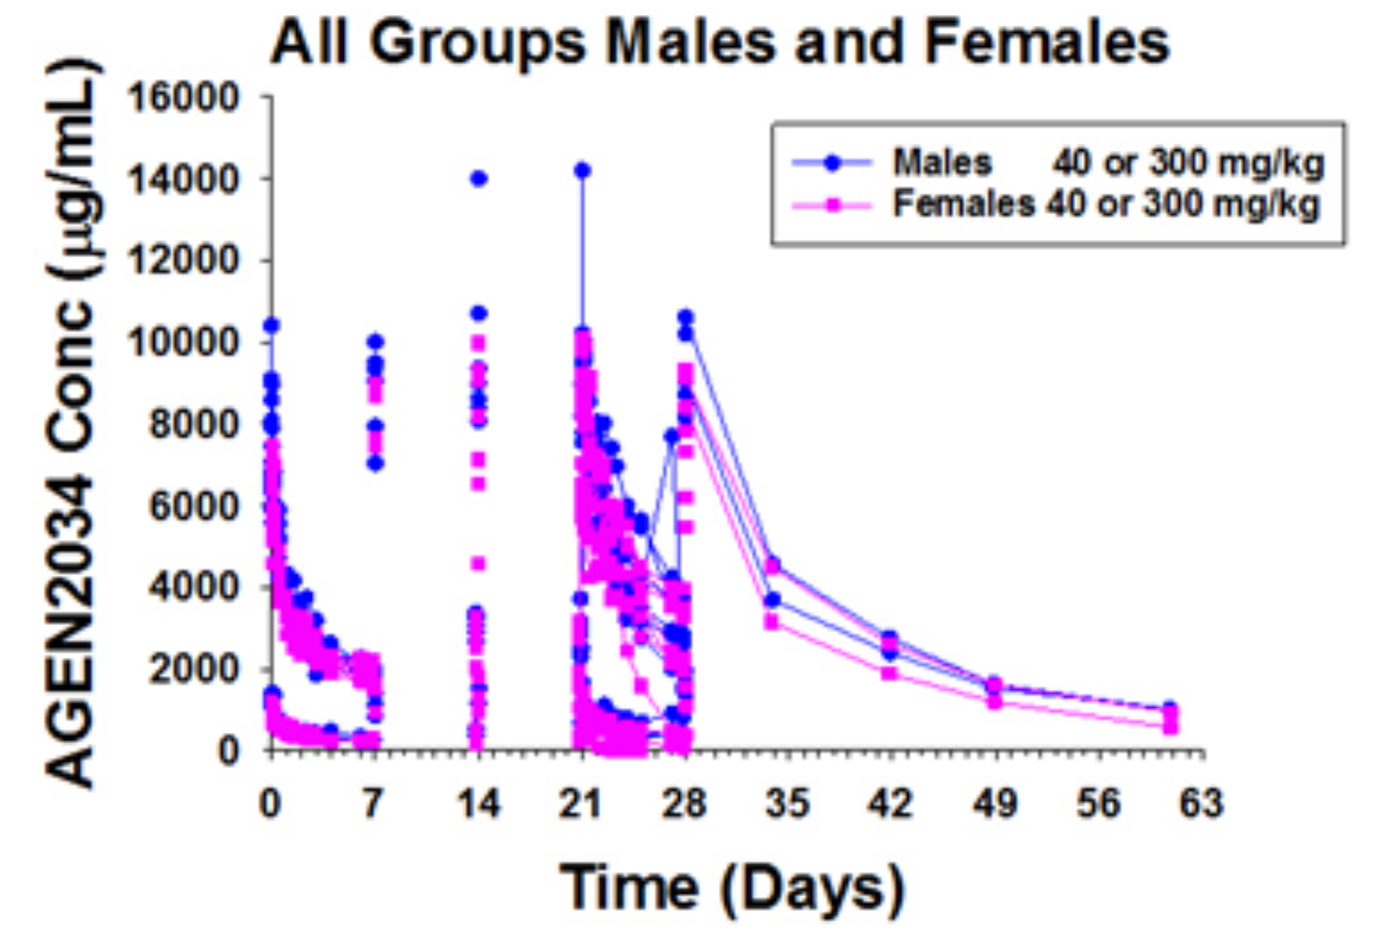

All Groups Males and Females
AGEN2034 Conc (µg/mL)
Males 40 or 300 mg/kg
Females 40 or 300 mg/kg
Time (Days)

- AGEN2034 was well-tolerated up to the NOAEL of 40 mg/kg
- TK analysis showed dose proportionality with some accumulation; no gender differences.
- Estimated AGEN2034 half-life of ~12 days

Summary

- AGEN2034 (anti-PD-1, IgG4) and AGEN1884 (anti-CTLA-4, IgG1) antibodies were discovered using Agenus' proprietary mammalian display antibody platform (Retrocyte Display™)
- AGEN2034 potently blocks ligand binding to enhance T cell activation and rescue T cell responsiveness in peptide stimulation and T cell suppression assays
- AGEN2034 cooperates with AGEN1884 and other immuno-modulatory pathways to further enhance T cell responsiveness
- In cynomolgus monkey, AGEN2034 in combination with AGEN1884, also promoted a pharmacodynamic proliferative and activation response in circulating T cells *in vivo*
- Clinical trials (NCT03104699) evaluating AGEN2034 in patients with advanced solid tumors (Phase 1) with expansion to second-line cervical cancer (Phase 2) is ongoing.

Author Disclosures

Dhan Chand, David Savitsky, Ana Gonzalez, Mariana Manrique, Christopher Clarke, Andrea Schuster, Elise E. Drouin, Jeremy D. Waight, Cornelia Mundt, Marc van Dijk, Jennifer S. Buell, Jean-Marie Cuillerot, Robert Stein and Nicholas S. Wilson: *Agenus Inc. and subsidiaries thereof:* Current or former employment and stock ownership.

Jedd Wolchok, Gerd Ritter, Taha Merghoub, David Schaer, Rikke B. Holmgaard, Roberta Zappasodi: No competing interests declared.

Acknowledgments

The licensed antibodies AGEN1884 and AGEN2034 were originally developed under a Collaborative Research and Development Agreement between Ludwig Cancer Research, 4-Antibody AG (now Agenus Switzerland Inc.) and Recepta Biopharma S.A. These antibodies are partnered with Recepta Biopharma S.A. for certain South American rights.