agenus
Agenus Inc. or subsidiary thereof (current or former employee), Lexington, MA 02421, USA.

Poster # 1878

Fc-enhanced anti-CTLA-4 Antibody, AGEN1181: New Mechanistic Insights for Potent Antitumor Immunity and Combination Potential in Treatment-resistant Solid Tumors

Antoine Tanne[1], Claire Galand[1], Marilyn Marques[1], Margaret Wilkens[1], Bishnu Joshi[1], Alvaro Sebastian Yague[1], Serina Ng[2], Haiyong Han[2], Sylvia Vincent[1], Mariana Manrique[1], Eleni Chantzoura[1], Daniel Levey[1], David Savitsky[1], Jennifer Buell[1], Dhan Chand[1]
[2] Molecular Medicine Division, Translational Genomics Research Institute, Phoenix, AZ; [1]Agenus Inc. or subsidiary thereof (current or former employee), Lexington, MA

Background

AGEN1181, an Fc-enhanced anti-CTLA-4 antibody, engages multiple mechanisms of action to promote effector and memory T cell-mediated anti-tumor immunity

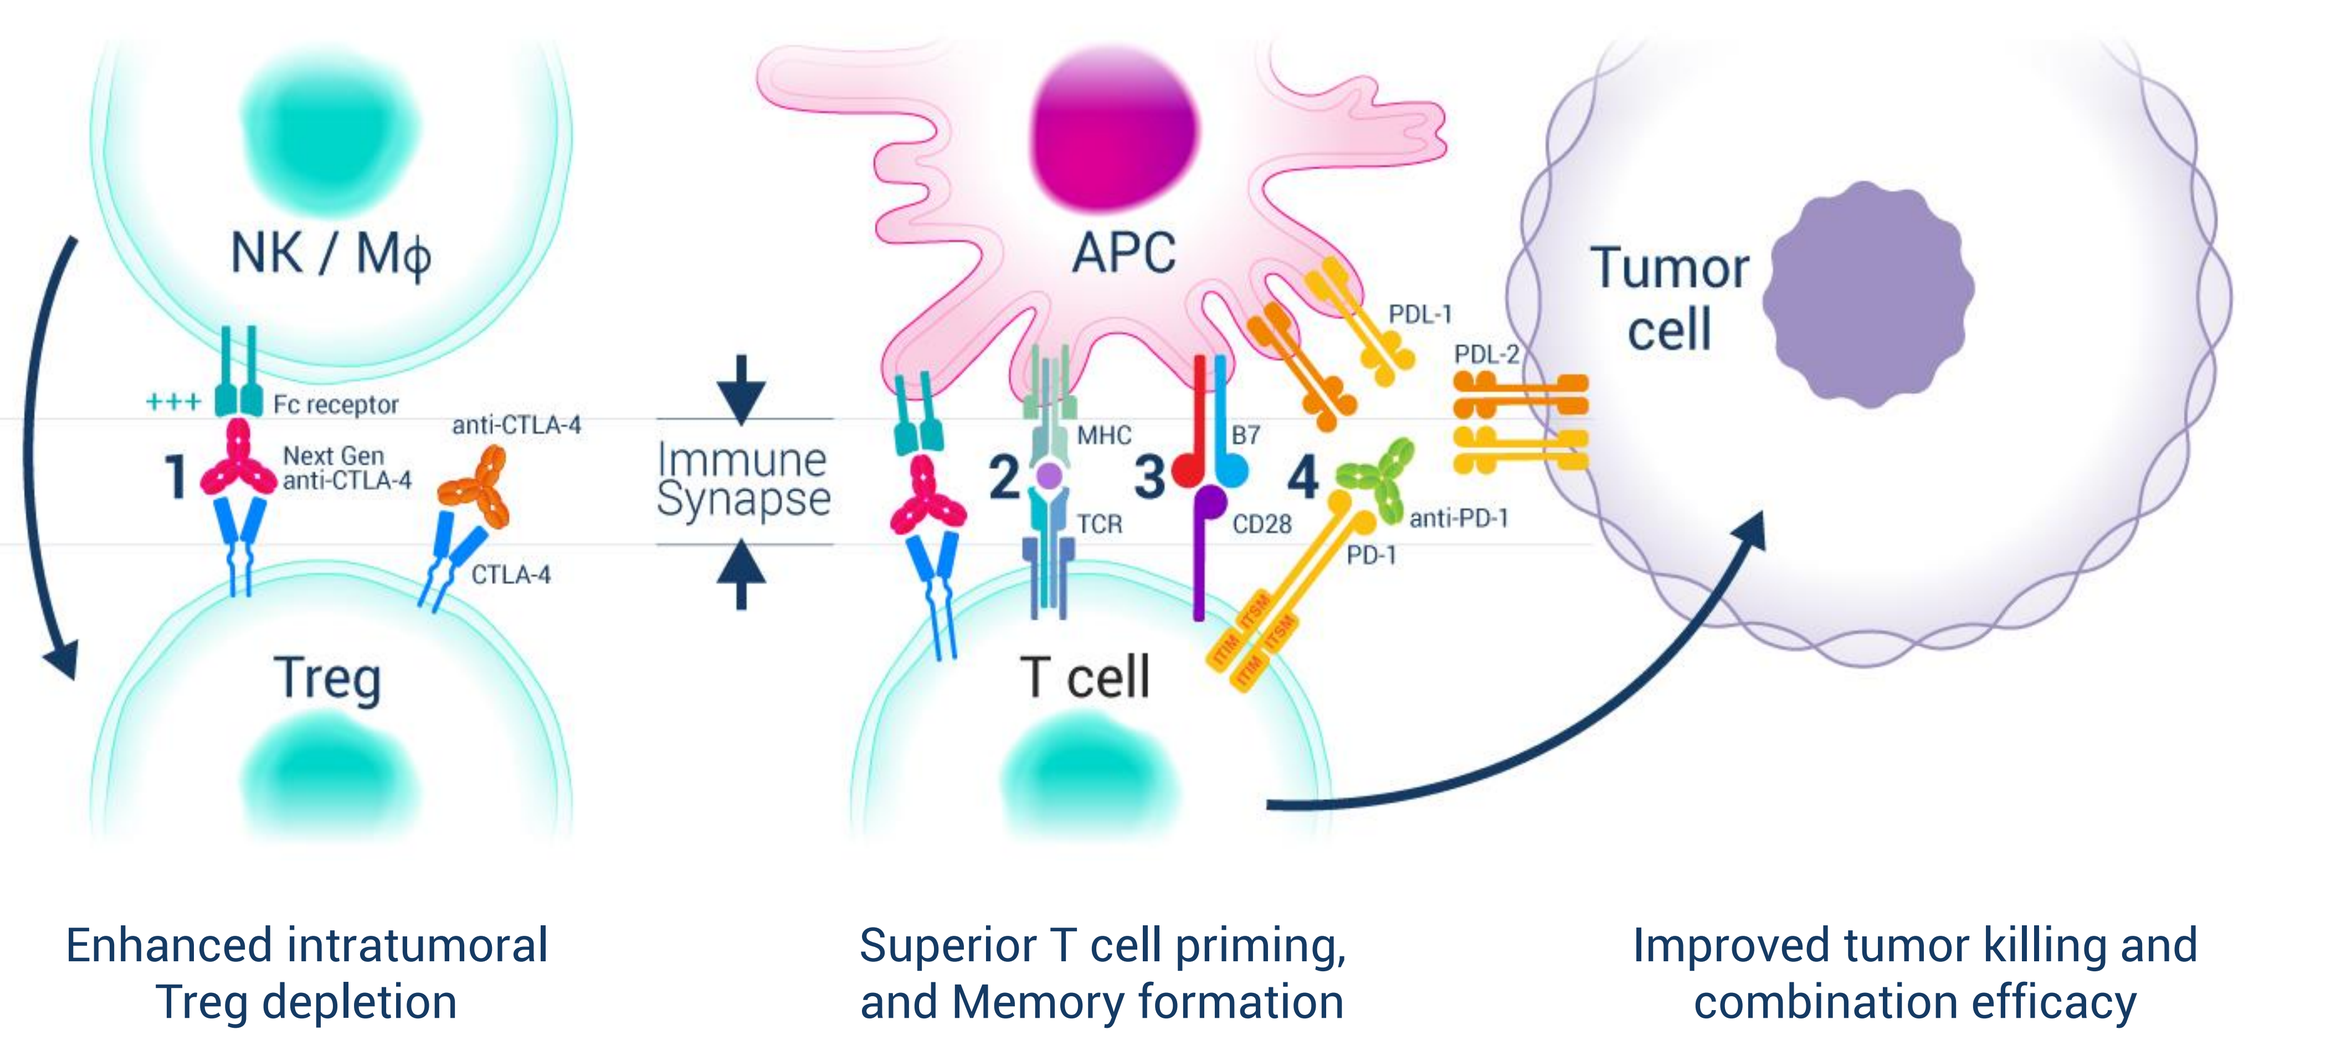
NK / Mϕ
APC
Tumor cell
Treg
T cell
Fc receptor
Next Gen anti-CTLA-4
anti-CTLA-4
CTLA-4
Immune Synapse
MHC
TCR
B7
CD28
PDL-1
PDL-2
anti-PD-1
PD-1
Enhanced intratumoral Treg depletion
Superior T cell priming, and Memory formation
Improved tumor killing and combination efficacy

Hypothesis:

Optimizing Fc - FcγR co-engagement enhances the activity of anti-CTLA-4 antagonist antibodies[1,2]. CTLA-4 antibodies with increased binding affinities to activating Fcγ receptors IV (CD16-2, mouse) or FcγRIIIA (CD16a, human) enhance NK mediated ADCC of CTLA-4 high expressing cells such as T regulatory cells (Treg) and strengthen the immune synapse between a T cell and an antigen presenting cell (APC) to amplify the breadth and depth of effector T cell priming.

Characteristics of studied antibodies

Table 1:

Anti-CTLA-4 Antibody		Species Reactivity	Fc Isotype	Fc Mutations	Blocking Properties	FcγR Binding Characteristics
Human	Parental	Human Cyno.	IgG1	-	+	Low
	AGEN1181 (Fc-enhanced)	Human Cyno.	IgG1.DLE	S239D.A330L.I332E	+	> FcγRIIIA binding ("Fc enhanced")
Murine Surrogates	9D9 (Parental)	Mouse	mIgG2b	-	+	Low
	AGEN1181ms (9D9 Fc-enhanced)	Mouse	mIgG2b.DLE	S241D.A332L.I334E	+	> FcγRIV binding

AGEN1181ms promotes single-agent activity against non-immunogenic and immunogenic tumors

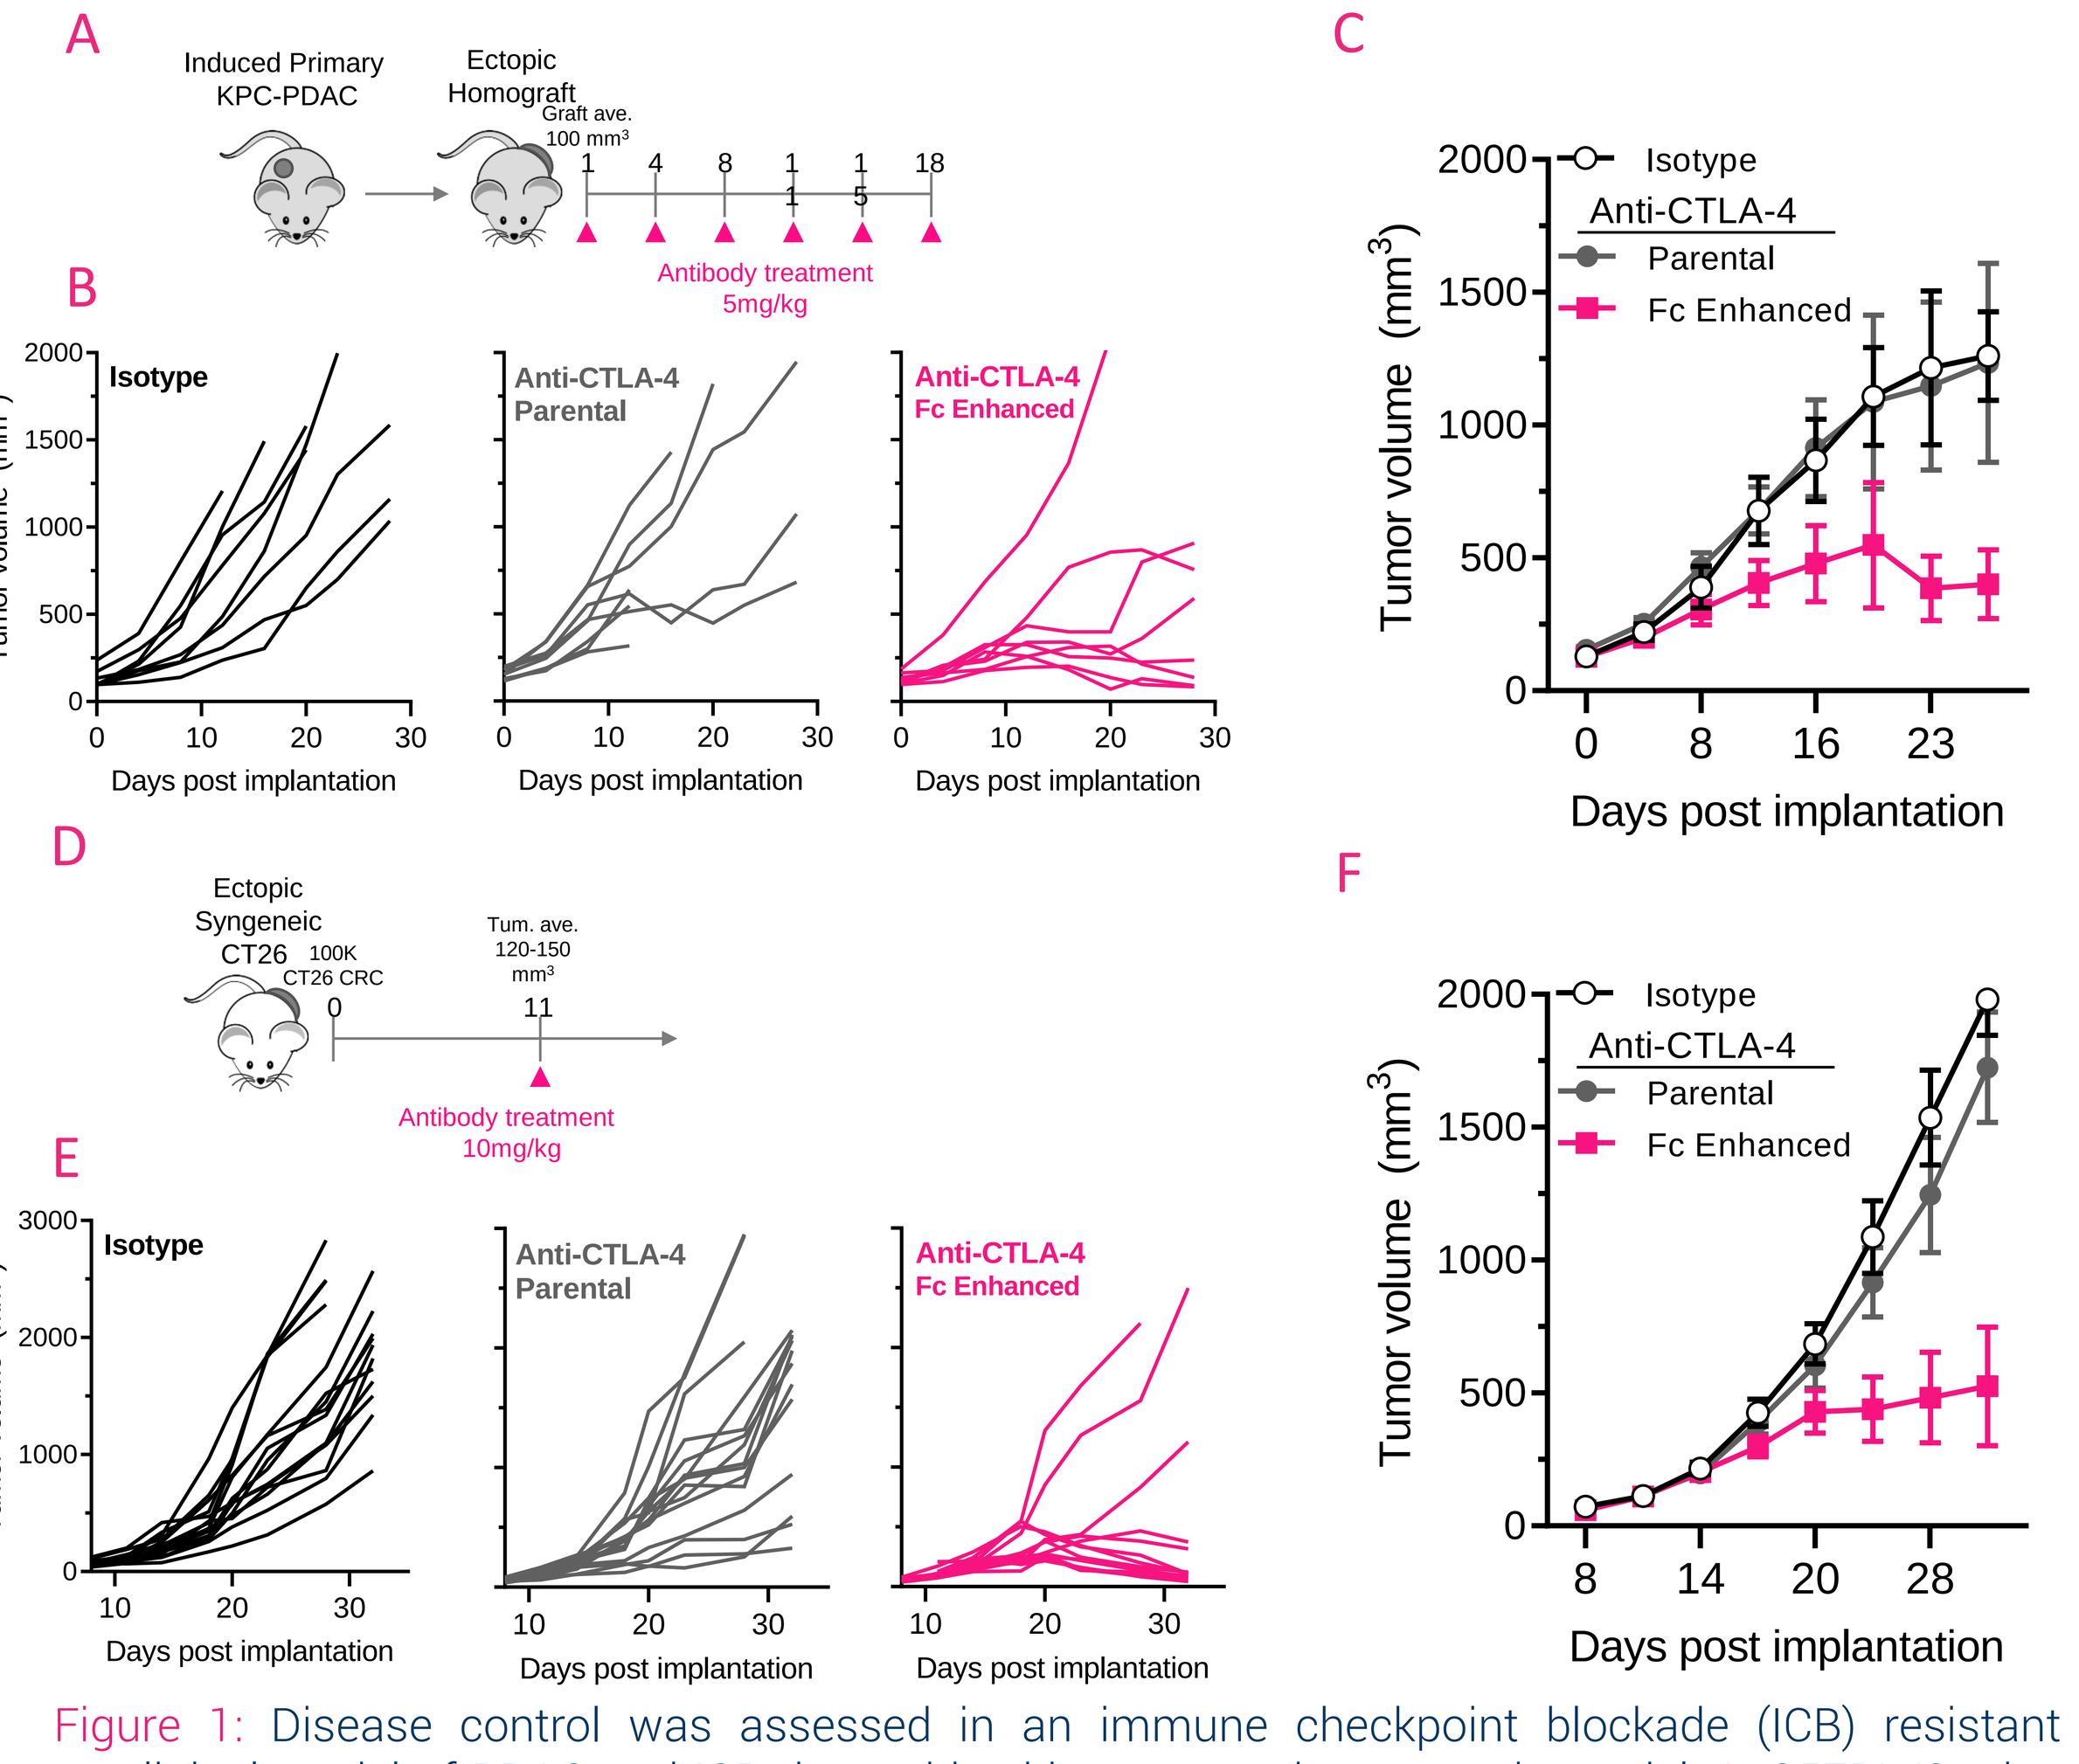
A
Induced Primary KPC-PDAC
Ectopic Homograft
Graft ave. 100 mm³
Antibody treatment 5mg/kg
B
Isotype
Anti-CTLA-4 Parental
Anti-CTLA-4 Fc Enhanced
Tumor volume (mm³)
Days post implantation
C
Isotype
Anti-CTLA-4
Parental
Fc Enhanced
D
Ectopic Syngeneic CT26
100K CT26 CRC
Tum. ave. 120-150 mm³
Antibody treatment 10mg/kg
E
F

Figure 1: Disease control was assessed in an immune checkpoint blockade (ICB) resistant preclinical model of PDAC and ICB desensitized immunogenic syngeneic model. A: C57BL/6 mice were implanted s.c with KPC (KrasG12D, P53-/- Pdx1-Cre) tumors with an approximate size of 100mm³ isolated from KPC-tumor bearing mice. Mice were treated with 6 biweekly intraperitoneal (i.p.) injections of parental, Fc-enhanced AGEN1181ms, or isotype control antibodies. D: BALB/c were challenged ectopically and subcutaneously with syngeneic CT26 colon carcinoma and received a late (120-150 mm³) single i.p. injection of Parental, Fc-enhanced AGEN1181ms, or isotype control antibodies. B-C;E-F: Tumor growth curves shown for individual mice (B,E) and average (C,F) emphasizing the increased potency of the murine AGEN1181 surrogate antibody compared with the parental.

AGEN1181ms depletes intratumoral Tregs

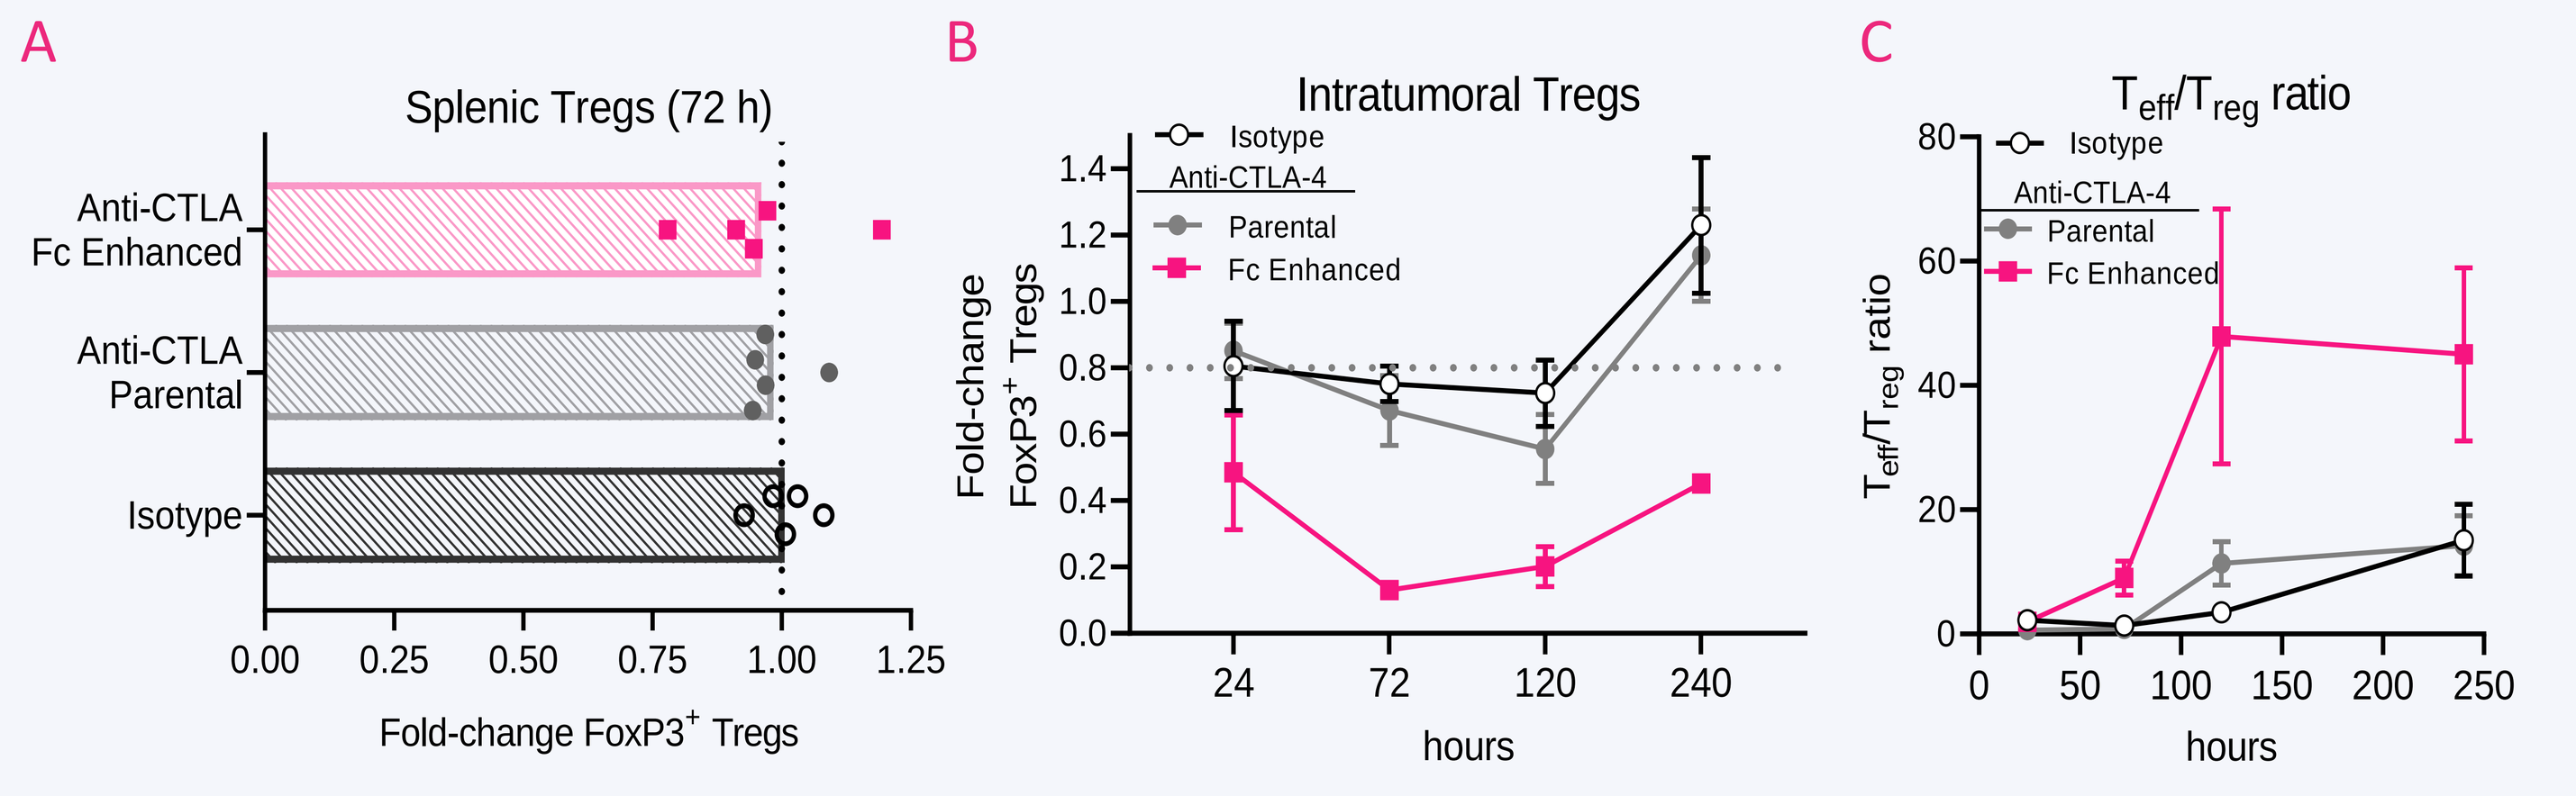
A
Splenic Tregs (72 h)
Anti-CTLA Fc Enhanced
Anti-CTLA Parental
Isotype
Fold-change FoxP3+ Tregs
B
Intratumoral Tregs
Isotype
Anti-CTLA-4
Parental
Fc Enhanced
Fold-change FoxP3+ Tregs
hours
C
Teff/Treg ratio
hours

Figure 2: Site-specific depletion of regulatory T cells (Treg) cells was measured in an ICB sensitive, immunogenic syngeneic ectopic CT26 model of colon carcinoma. Balb/cJ mice bearing CT26 tumors (50-75 mm³) were injected with a single 0.5 mg/kg i.p dose of parental, Fc-enhanced AGEN1181ms, or isotype control antibodies. To evaluate cell depletion, five mice per group were sacrificed at 24, 72, 120, and 240 hours following antibody treatment and tissues were collected. The Treg content was analyzed by focusing on hematopoietic lymphoid CD4 T cells expressing FOXP3 versus other cells subsets. A: Fold-change of Foxp3+ Tregs in the spleen did not demonstrate significant differences. B-C: In the tumor, the fold-change of FoxP3+ Tregs relative to vehicle control group (line) and the ratio of CD8 effector T cells to Tregs both demonstrate a higher Treg depletion potency for the Fc-enhanced molecule.

AGEN1181ms enhances tumor immune cell infiltration

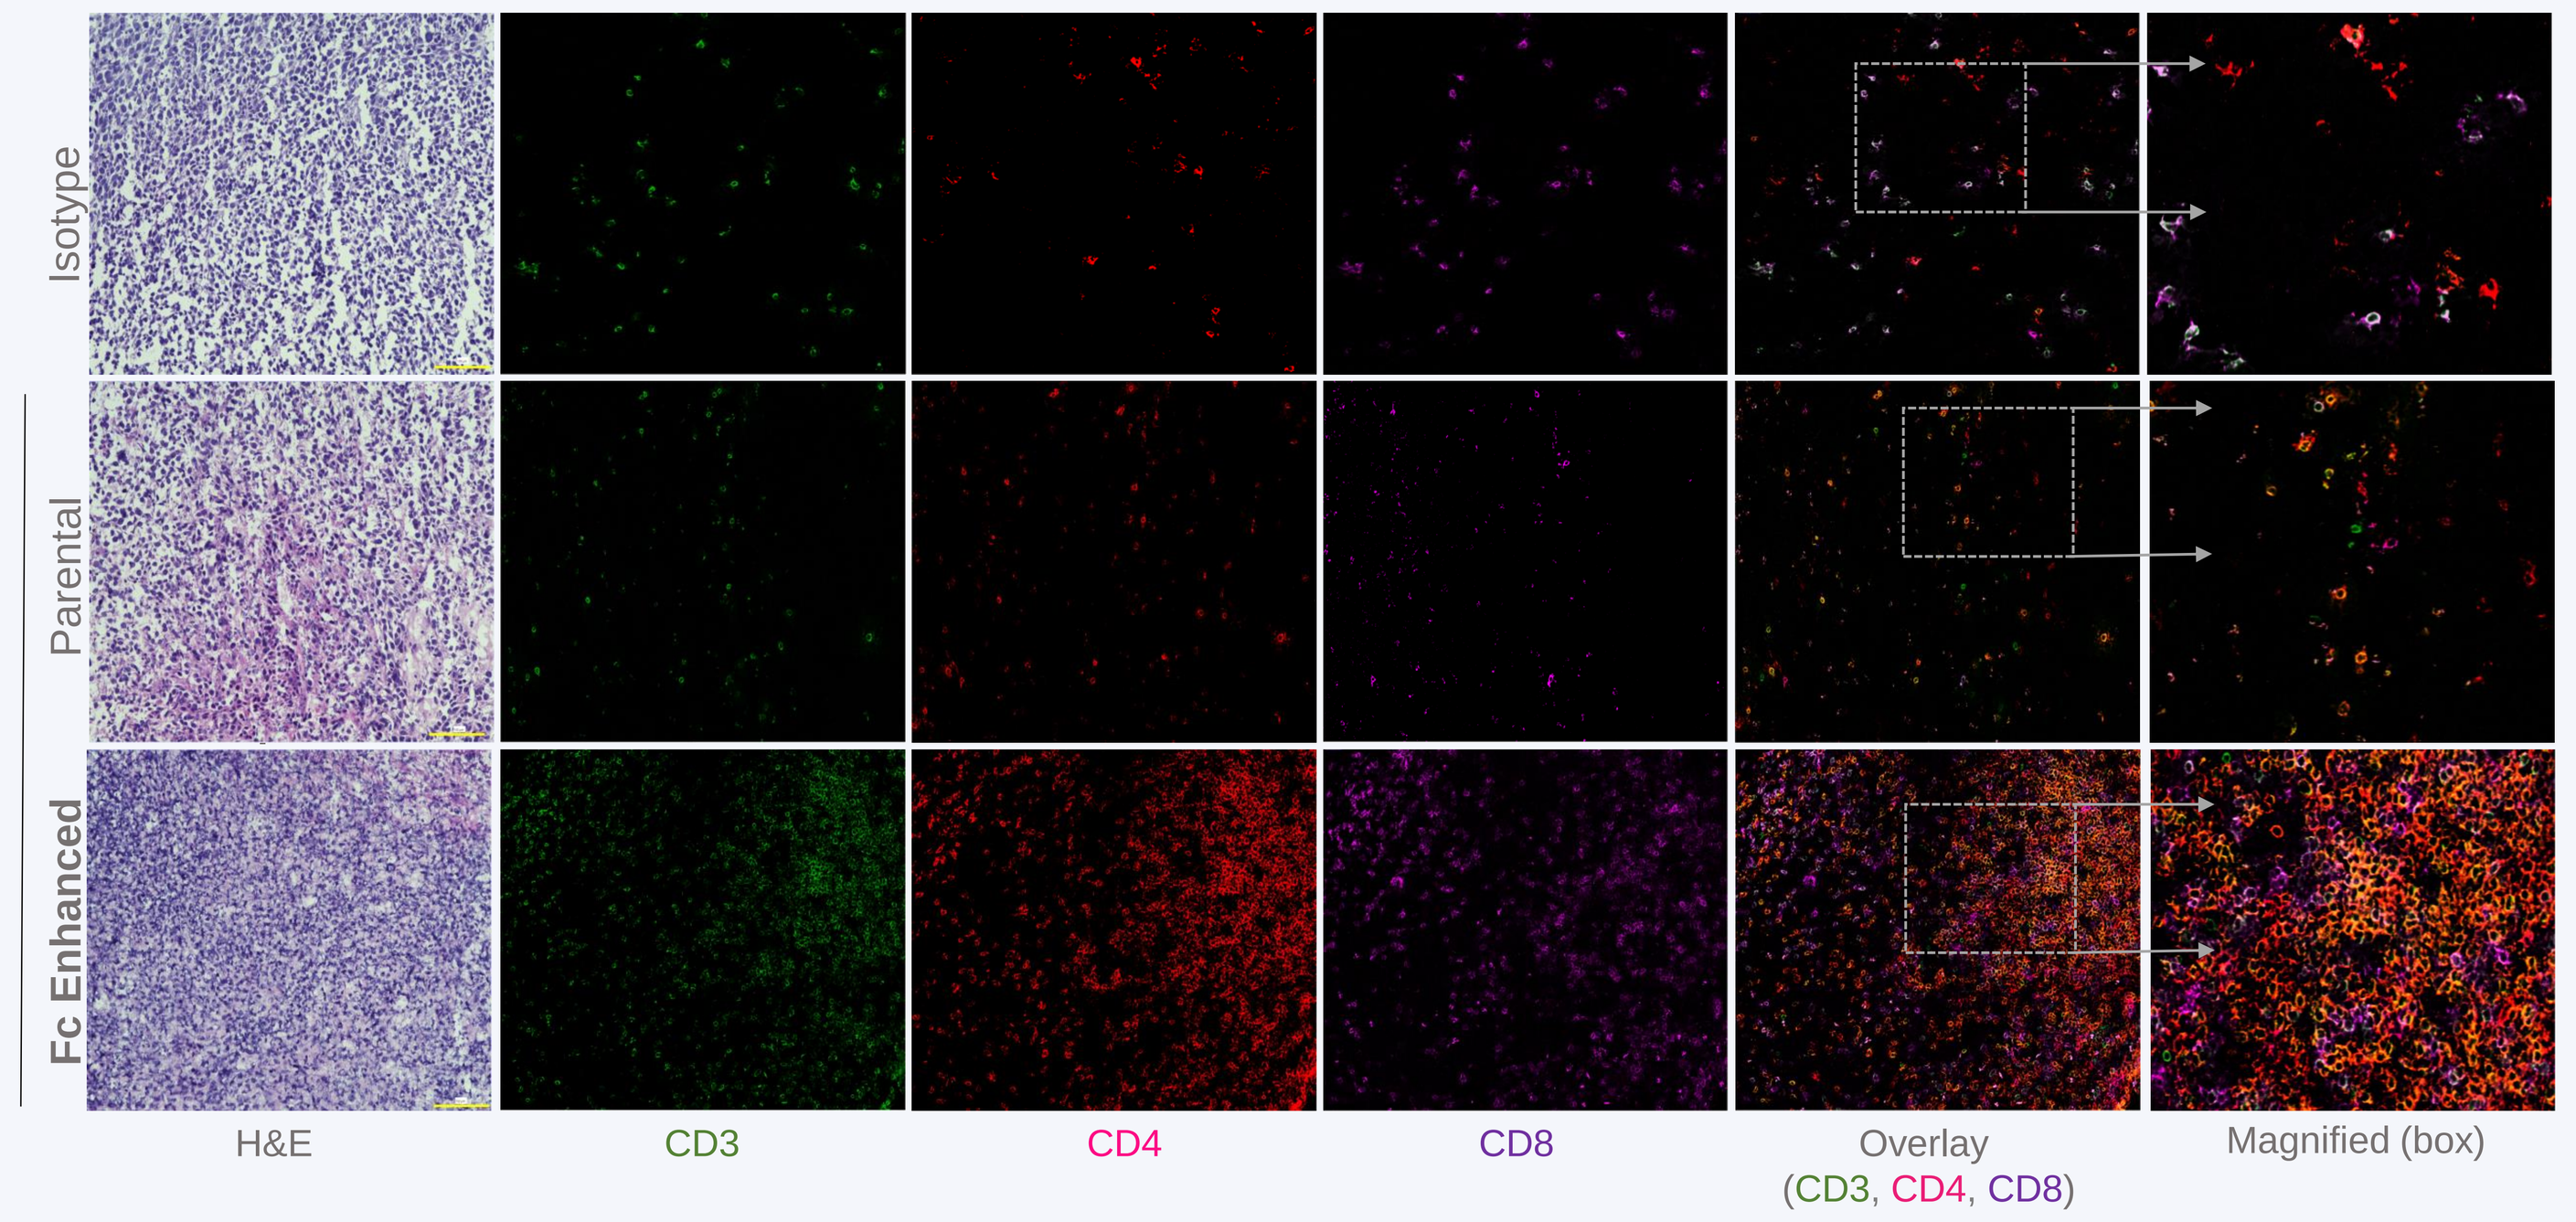
Isotype
Parental
Fc Enhanced
H&E
CD3
CD4
CD8
Overlay (CD3, CD4, CD8)
Magnified (box)

Figure 3: Immunogenicity in the tumor microenvironment phenotyping conducted in the syngeneic CT26 colon carcinoma model. Balb/cJ mice bearing CT26 tumors (100 mm³) were injected with a single 5 mg/kg i.p. dose of parental, Fc-enhanced AGEN1181ms, or isotype control antibodies. Representative hematoxylin & eosin (H&E) and multiplex immunofluorescence images showing isotype (top panel), parental (middle panel) and Fc-enhanced (bottom panel) anti-CTLA-4 staining in fresh frozen sections from CT26 subcutaneous tumor model collected 9 days post-treatment. Far left column represents H&E-stained tumor histology images demonstrate that the AGEN1181ms treatment increased immune cell infiltration. Second column for CD4, third column CD3, fourth column CD8, fifth and sixth column overlay of CD4, CD3 and CD8 demonstrate that AGEN1181ms induced robust T cell infiltration in this model.

AGEN1181ms enhances T cell clonality and expansion of tumor-associated T cells in tumor-bearing mice

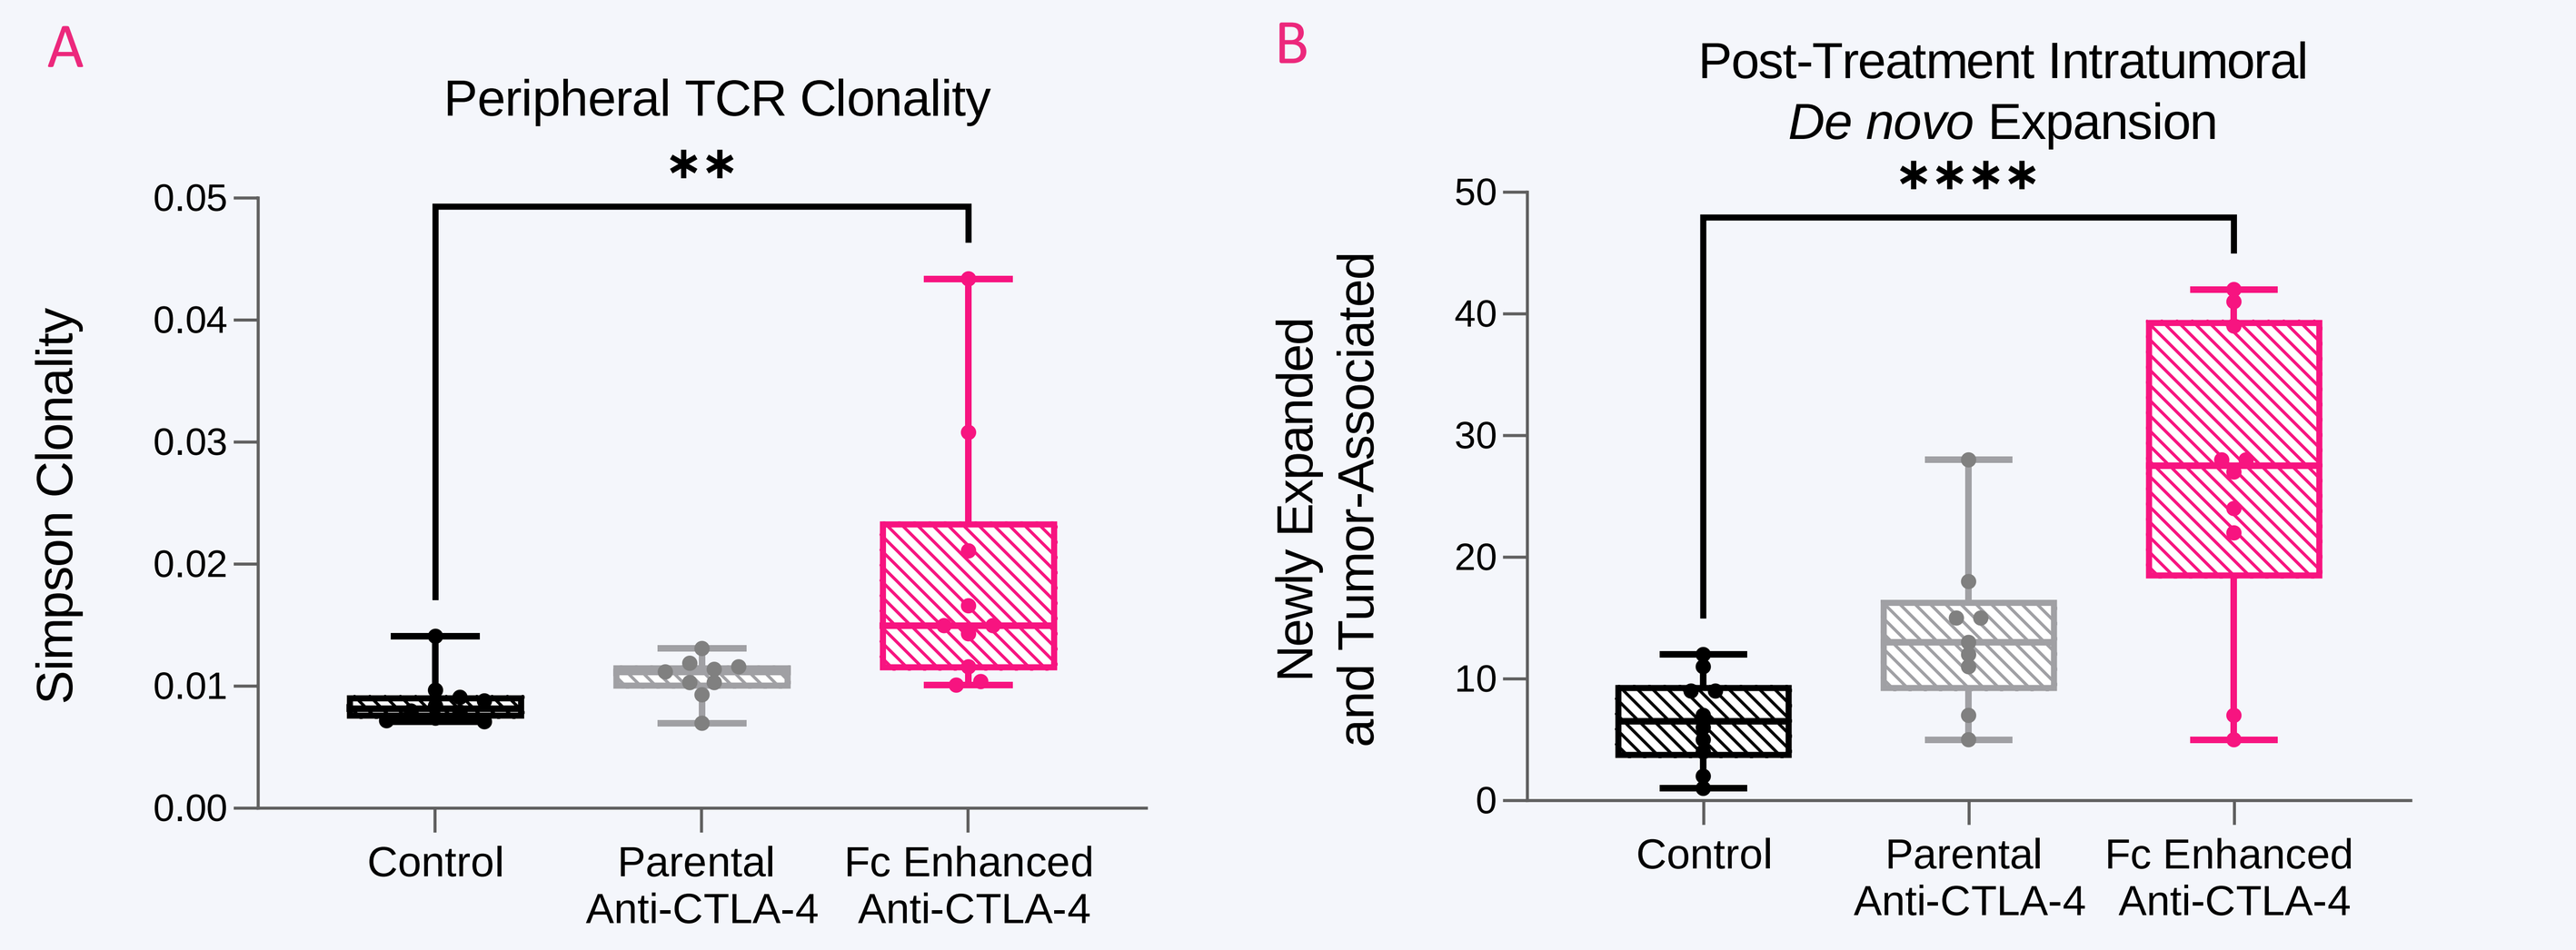
A
Peripheral TCR Clonality
Simpson Clonality
Control
Parental Anti-CTLA-4
Fc Enhanced Anti-CTLA-4
B
Post-Treatment Intratumoral De novo Expansion
Newly Expanded and Tumor-Associated

Figure 4: Qualitative and quantitative analysis of the TCR sequence spectrum conducted in the syngeneic CT26 colon carcinoma model. Balb/cJ mice bearing CT26 tumors (120 mm³) were injected with a single 2.5 mg/kg i.p. dose of parental, Fc-enhanced AGEN1181ms, or isotype control antibodies. Blood samples were taken pre-treatment and blood and tumor collected 5 days post-dosing for sequencing the TCR beta repertoire (immunoSEQ mouse TCRB assay; Adaptive Biotechnologies). A: Changes in peripheral TCR signatures pre- *vs.* post-treatment show an increased effect of AGEN1181ms on systemic T cell clonality. B: Evolution of the intratumoral T cell diversity demonstrates that AGEN1181ms induces ICB-driven priming and recruitment of newly expanded and tumor-associated clones as determined using Simpson clonality method.

AGEN1181ms enhances intratumoral T cell effector function

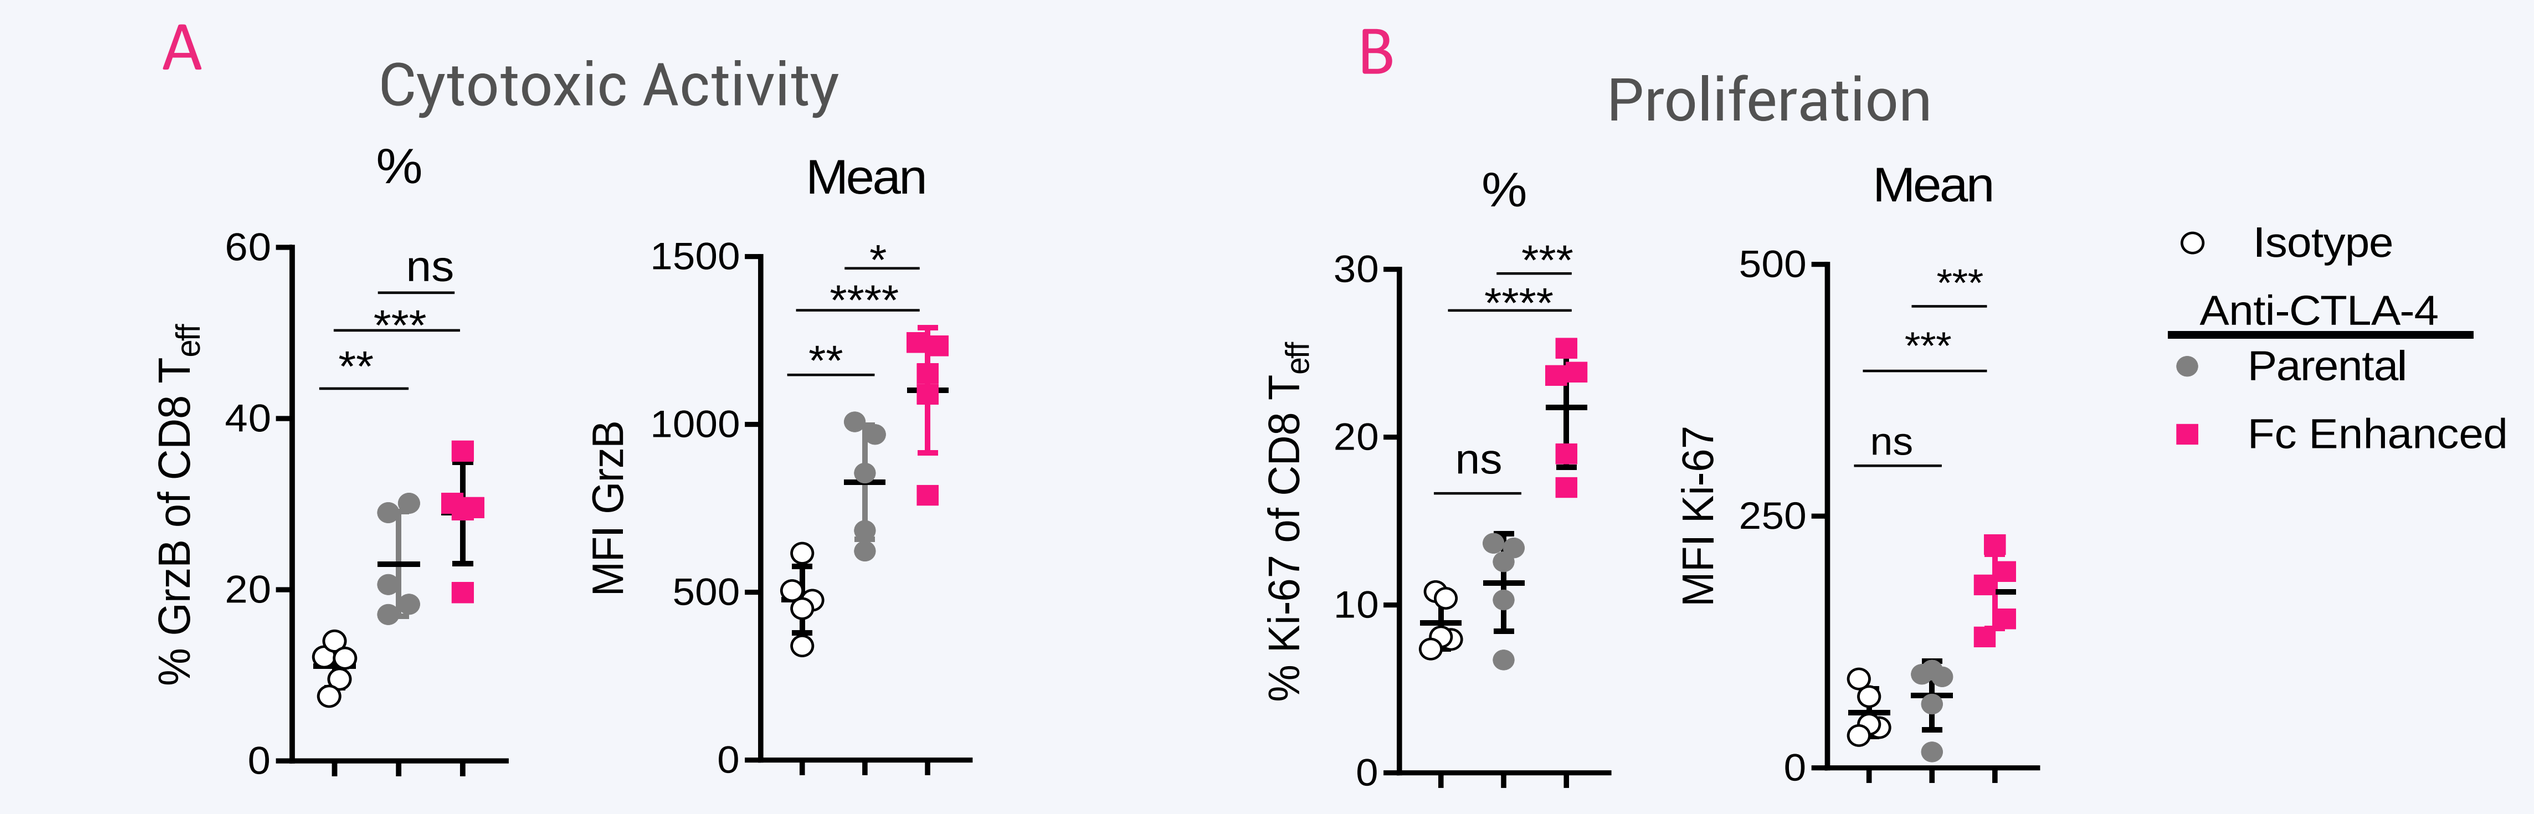
A
Cytotoxic Activity
%
Mean
% GrzB of CD8 Teff
MFI GrzB
B
Proliferation
% Ki-67 of CD8 Teff
MFI Ki-67
Isotype
Anti-CTLA-4
Parental
Fc Enhanced

Figure 4: Intratumoral effector T cell phenotyping conducted in ICB sensitive, immunogenic syngeneic ectopic MC38 model of colon carcinoma. C57BL/6J mice bearing MC38 tumors (50-75 mm³) received a single 5 mg/kg i.p. dose of parental, Fc-enhanced AGEN1181ms, or isotype control antibodies. Mice were sacrificed 120 hours post-treatment and tumors collected. The activation status of tumor infiltrating effector, CD4 and CD8 T cell phenotype was analyzed based on functional (GrzB) and mitosis (Ki-67) markers. A-B: The antitumoral CD8 T cell response was augmented by AGEN1181ms based on increased cytotoxicity (A) and proliferation (B).

AGEN1181ms promotes superior T cell memory responses

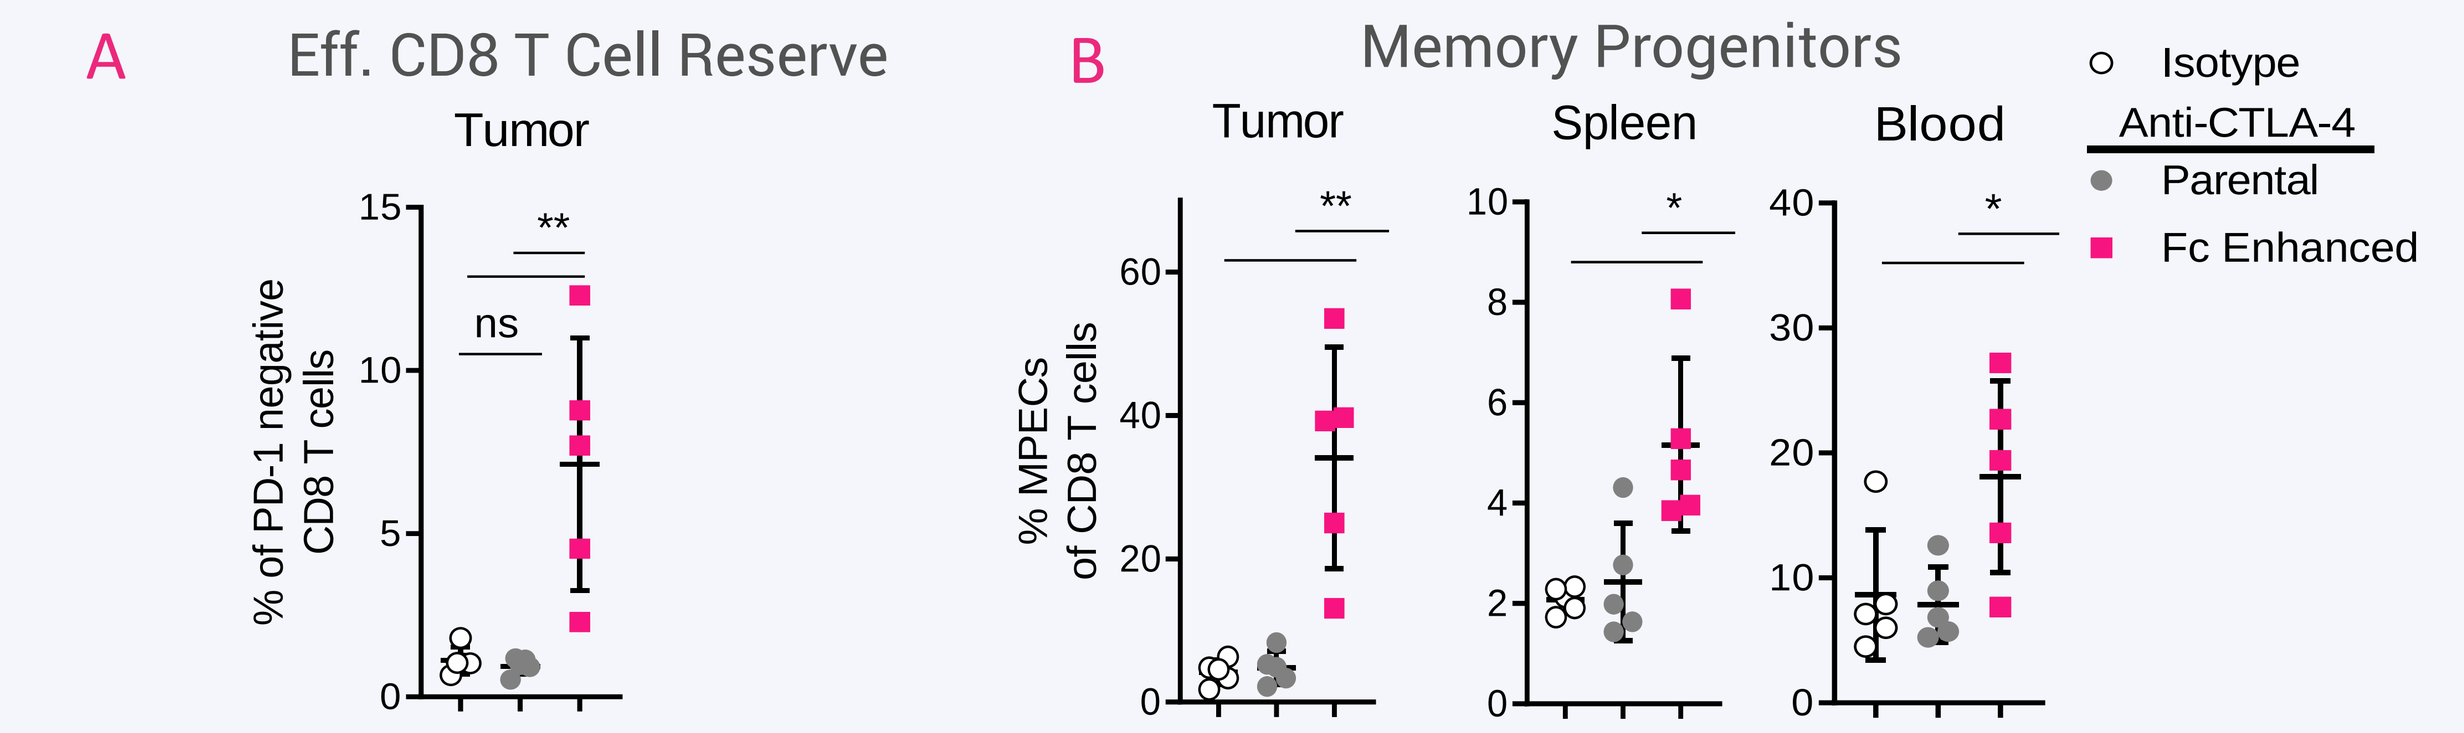
A
Eff. CD8 T Cell Reserve
Tumor
% of PD-1 negative CD8 T cells
B
Memory Progenitors
Tumor
Spleen
Blood
% MPECs of CD8 T cells
Isotype
Anti-CTLA-4
Parental
Fc Enhanced

Figure 5: Characterization of effect on antitumoral memory response conducted in the syngeneic CT26 colon carcinoma model. Balb/cJ mice bearing CT26 tumors (50 mm³) were treated with a single 5 mg/kg i.p. dose of parental, Fc-enhanced AGEN1181ms, or isotype control antibodies. Mice were sacrificed 10 days post-treatment. Effector CD8 T lymphocytes were phenotyped based on non-activated status (PD-1 neg) and expression of memory lineage markers (CX3CR1 and Slamf7). A: AGEN1181ms significantly augmented the pool of resting tumoral effector CD8 T cells. B: AGEN1181ms drives differentiation toward the memory lineage, resulting in tumoral and systemic accumulation of memory precursor effector cells (MPECs).

AGEN1181ms promotes durable antitumor responses

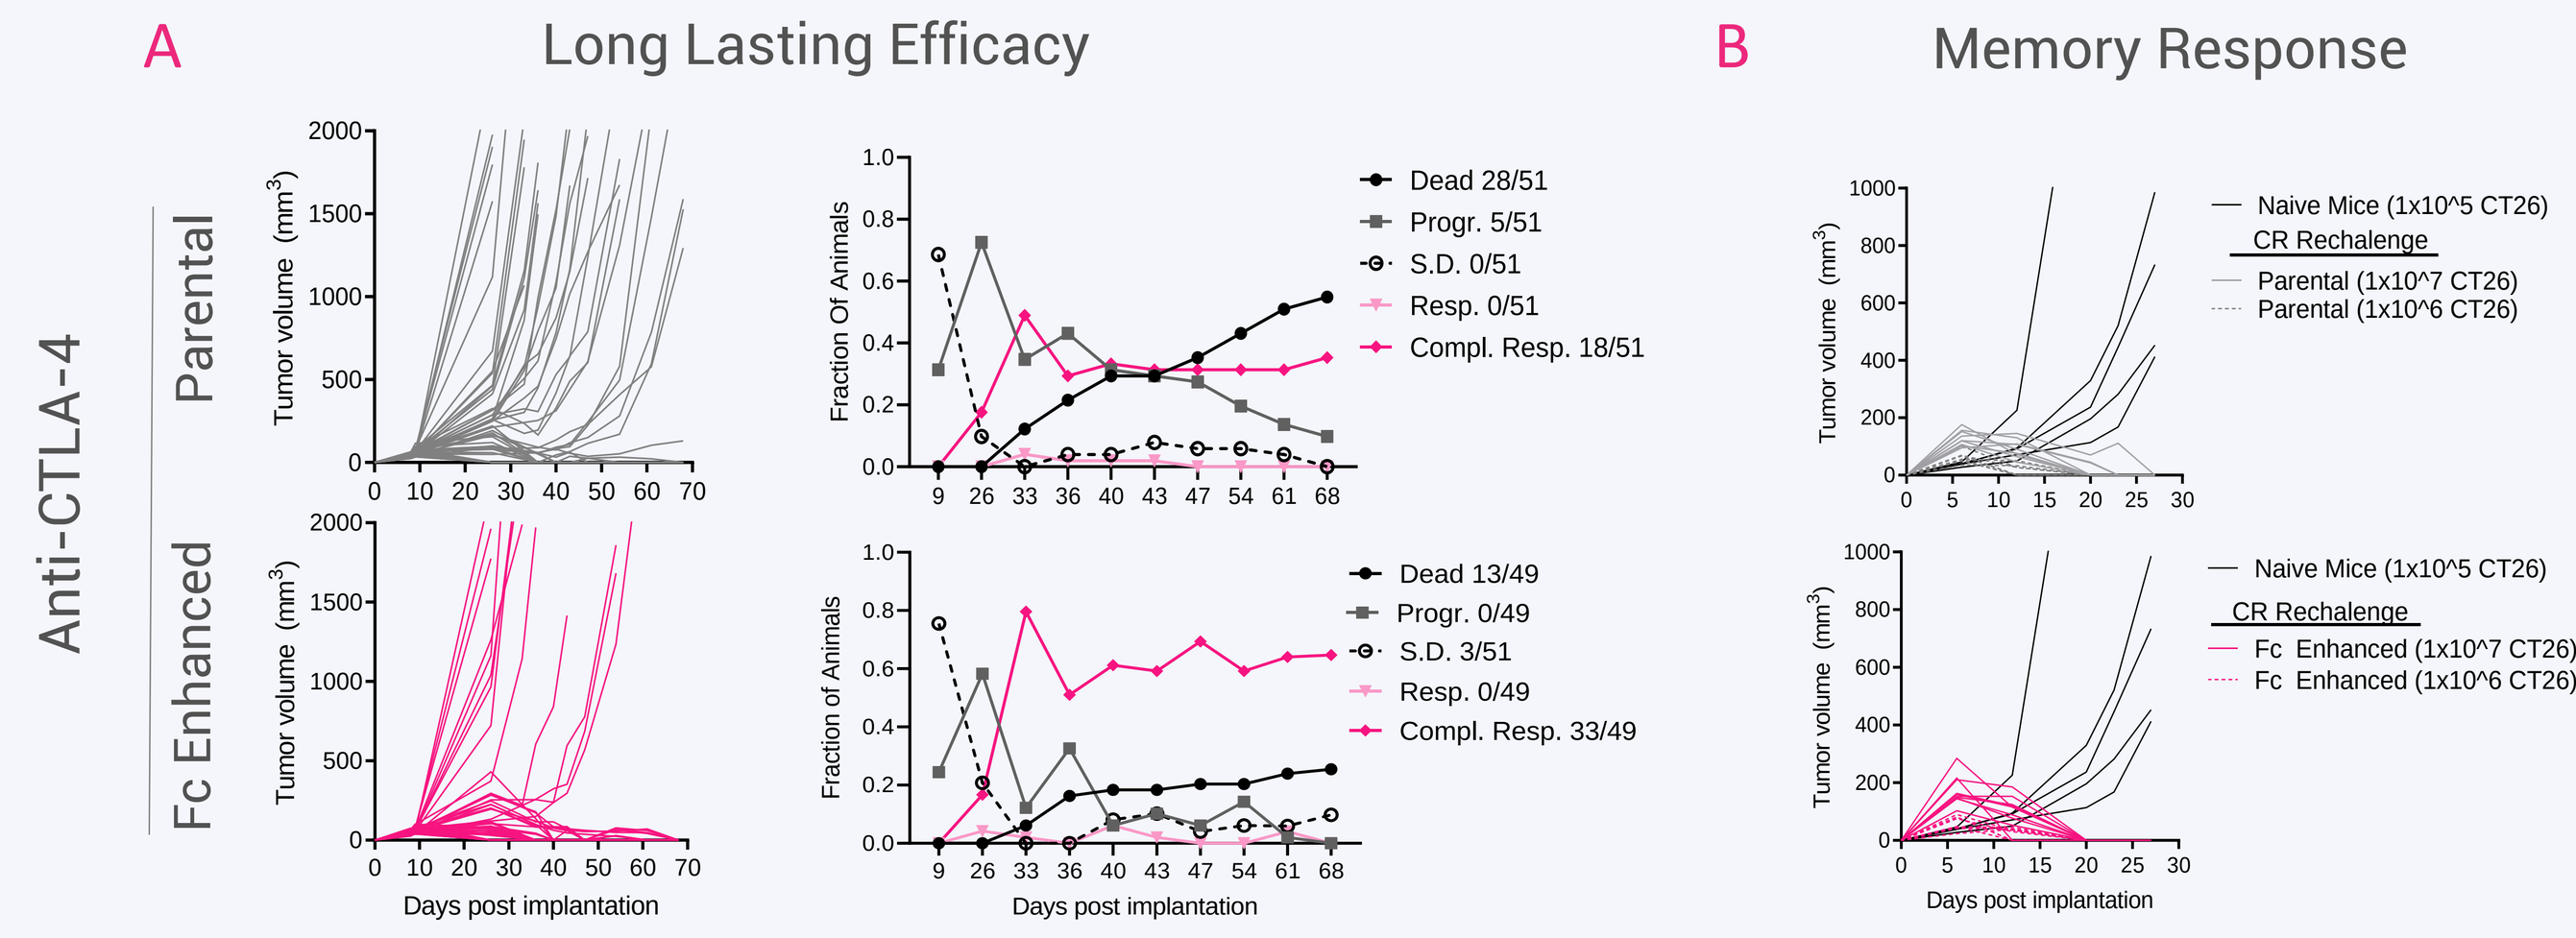
A
Long Lasting Efficacy
Anti-CTLA-4
Parental
Fc Enhanced
Tumor volume (mm³)
Fraction Of Animals
Dead 28/51
Progr. 5/51
S.D. 0/51
Resp. 0/51
Compl. Resp. 18/51
Dead 13/49
Progr. 0/49
S.D. 3/51
Resp. 0/49
Compl. Resp. 33/49
Days post implantation
B
Memory Response
Naive Mice (1x10^5 CT26)
CR Rechalenge
Parental (1x10^7 CT26)
Parental (1x10^6 CT26)
Fc Enhanced (1x10^7 CT26)
Fc Enhanced (1x10^6 CT26)

Figure 6: Characterization of effect on long lasting antitumoral response conducted in the syngeneic CT26 colon carcinoma model. Balb/cJ mice bearing CT26 tumors (100 mm³) were injected with a single 5 mg/kg i.p. dose of parental, Fc-enhanced AGEN1181ms, or isotype control antibodies. A: AGEN1181ms promoted long-lasting treatment benefits based on survival, complete responder (CR), and stable disease (SD) evolution over a >50-day period following treatment. B: Despite significant differences between the Parental (18/51 CR) vs AGEN1181ms (33/49 CR) in controlling primary tumor growth; upon rechallenge of the complete responders with the parental tumor cell line both treatments demonstrated an ability to induce efficient and comparable memory responses that led to protection.

AGEN1181ms in combination with adjuvant chemotherapy promotes curative responses in treatment-resistant PDAC

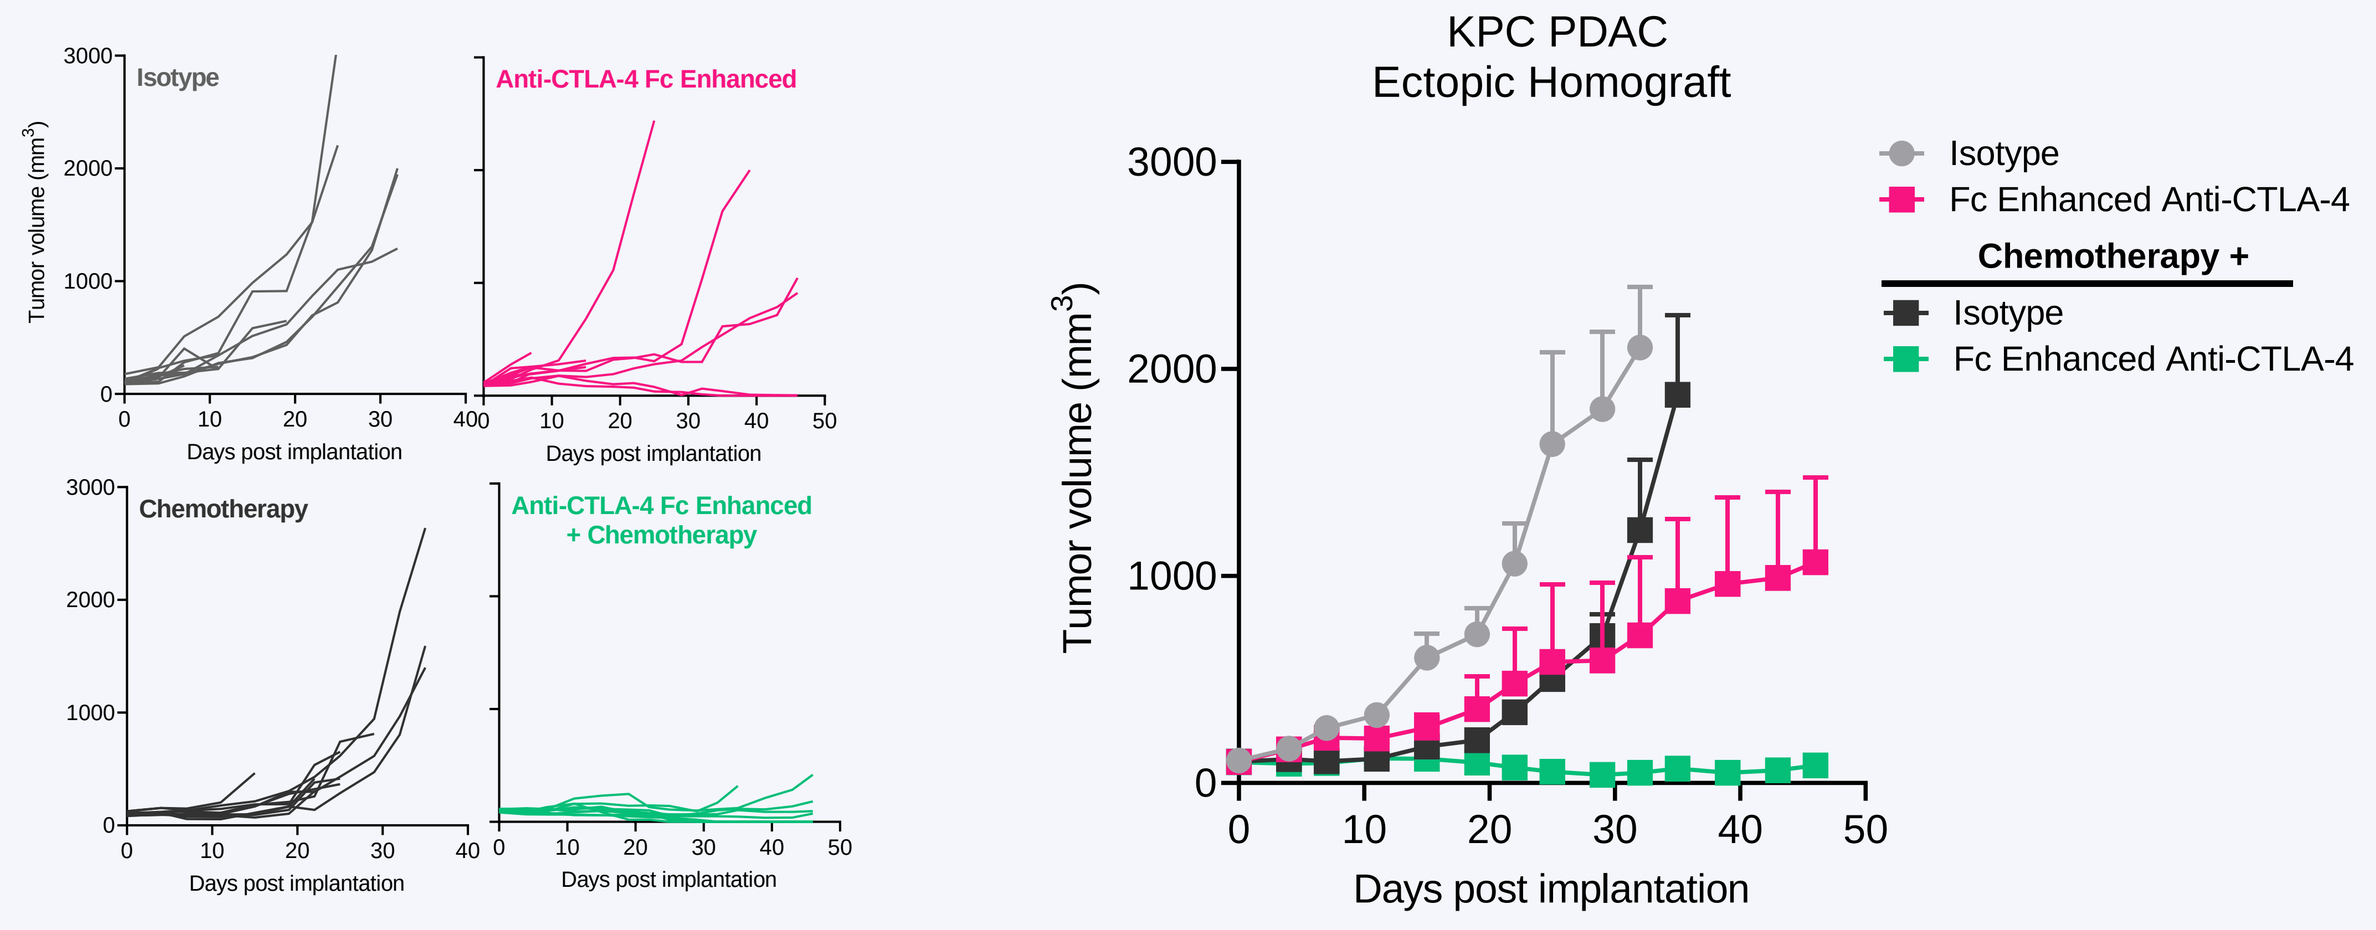
Isotype
Anti-CTLA-4 Fc Enhanced
Chemotherapy
Anti-CTLA-4 Fc Enhanced + Chemotherapy
Tumor volume (mm³)
Days post implantation
KPC PDAC
Ectopic Homograft
Isotype
Fc Enhanced Anti-CTLA-4
Chemotherapy +
Isotype
Fc Enhanced Anti-CTLA-4

Figure 7: Therapeutic potency of AGEN1181ms in combination with immunogenic adjuvant chemotherapy evaluated in ICB resistant preclinical models of PDAC treated with conventional triple chemotherapy. C57BL/6 mice s.c. implanted with KPC (KrasG12D, P53-/- Pdx1-Cre) tumor chunks (100mm³) isolated from KPC-tumor bearing mice were treated with 6 biweekly i.p. injections of parental, Fc-enhanced AGEN1181ms, or isotype control antibodies either as monotherapy or combination with chemotherapy. Chemotherapy-treated mice received i.p. gemcitabine (70 mg/kg), i.p. cisplatin (4 mg/kg) and i.v. nab-paclitaxel (25 mg/kg) on days 1 and 4. AGEN1181ms synergized with chemotherapy, resulting in 100% primary tumor control, including a significant proportion of complete responders (50% CR).

AGEN1181+/- PD-1 enhances the activation of central and effector memory T cells in cynomolgus monkeys

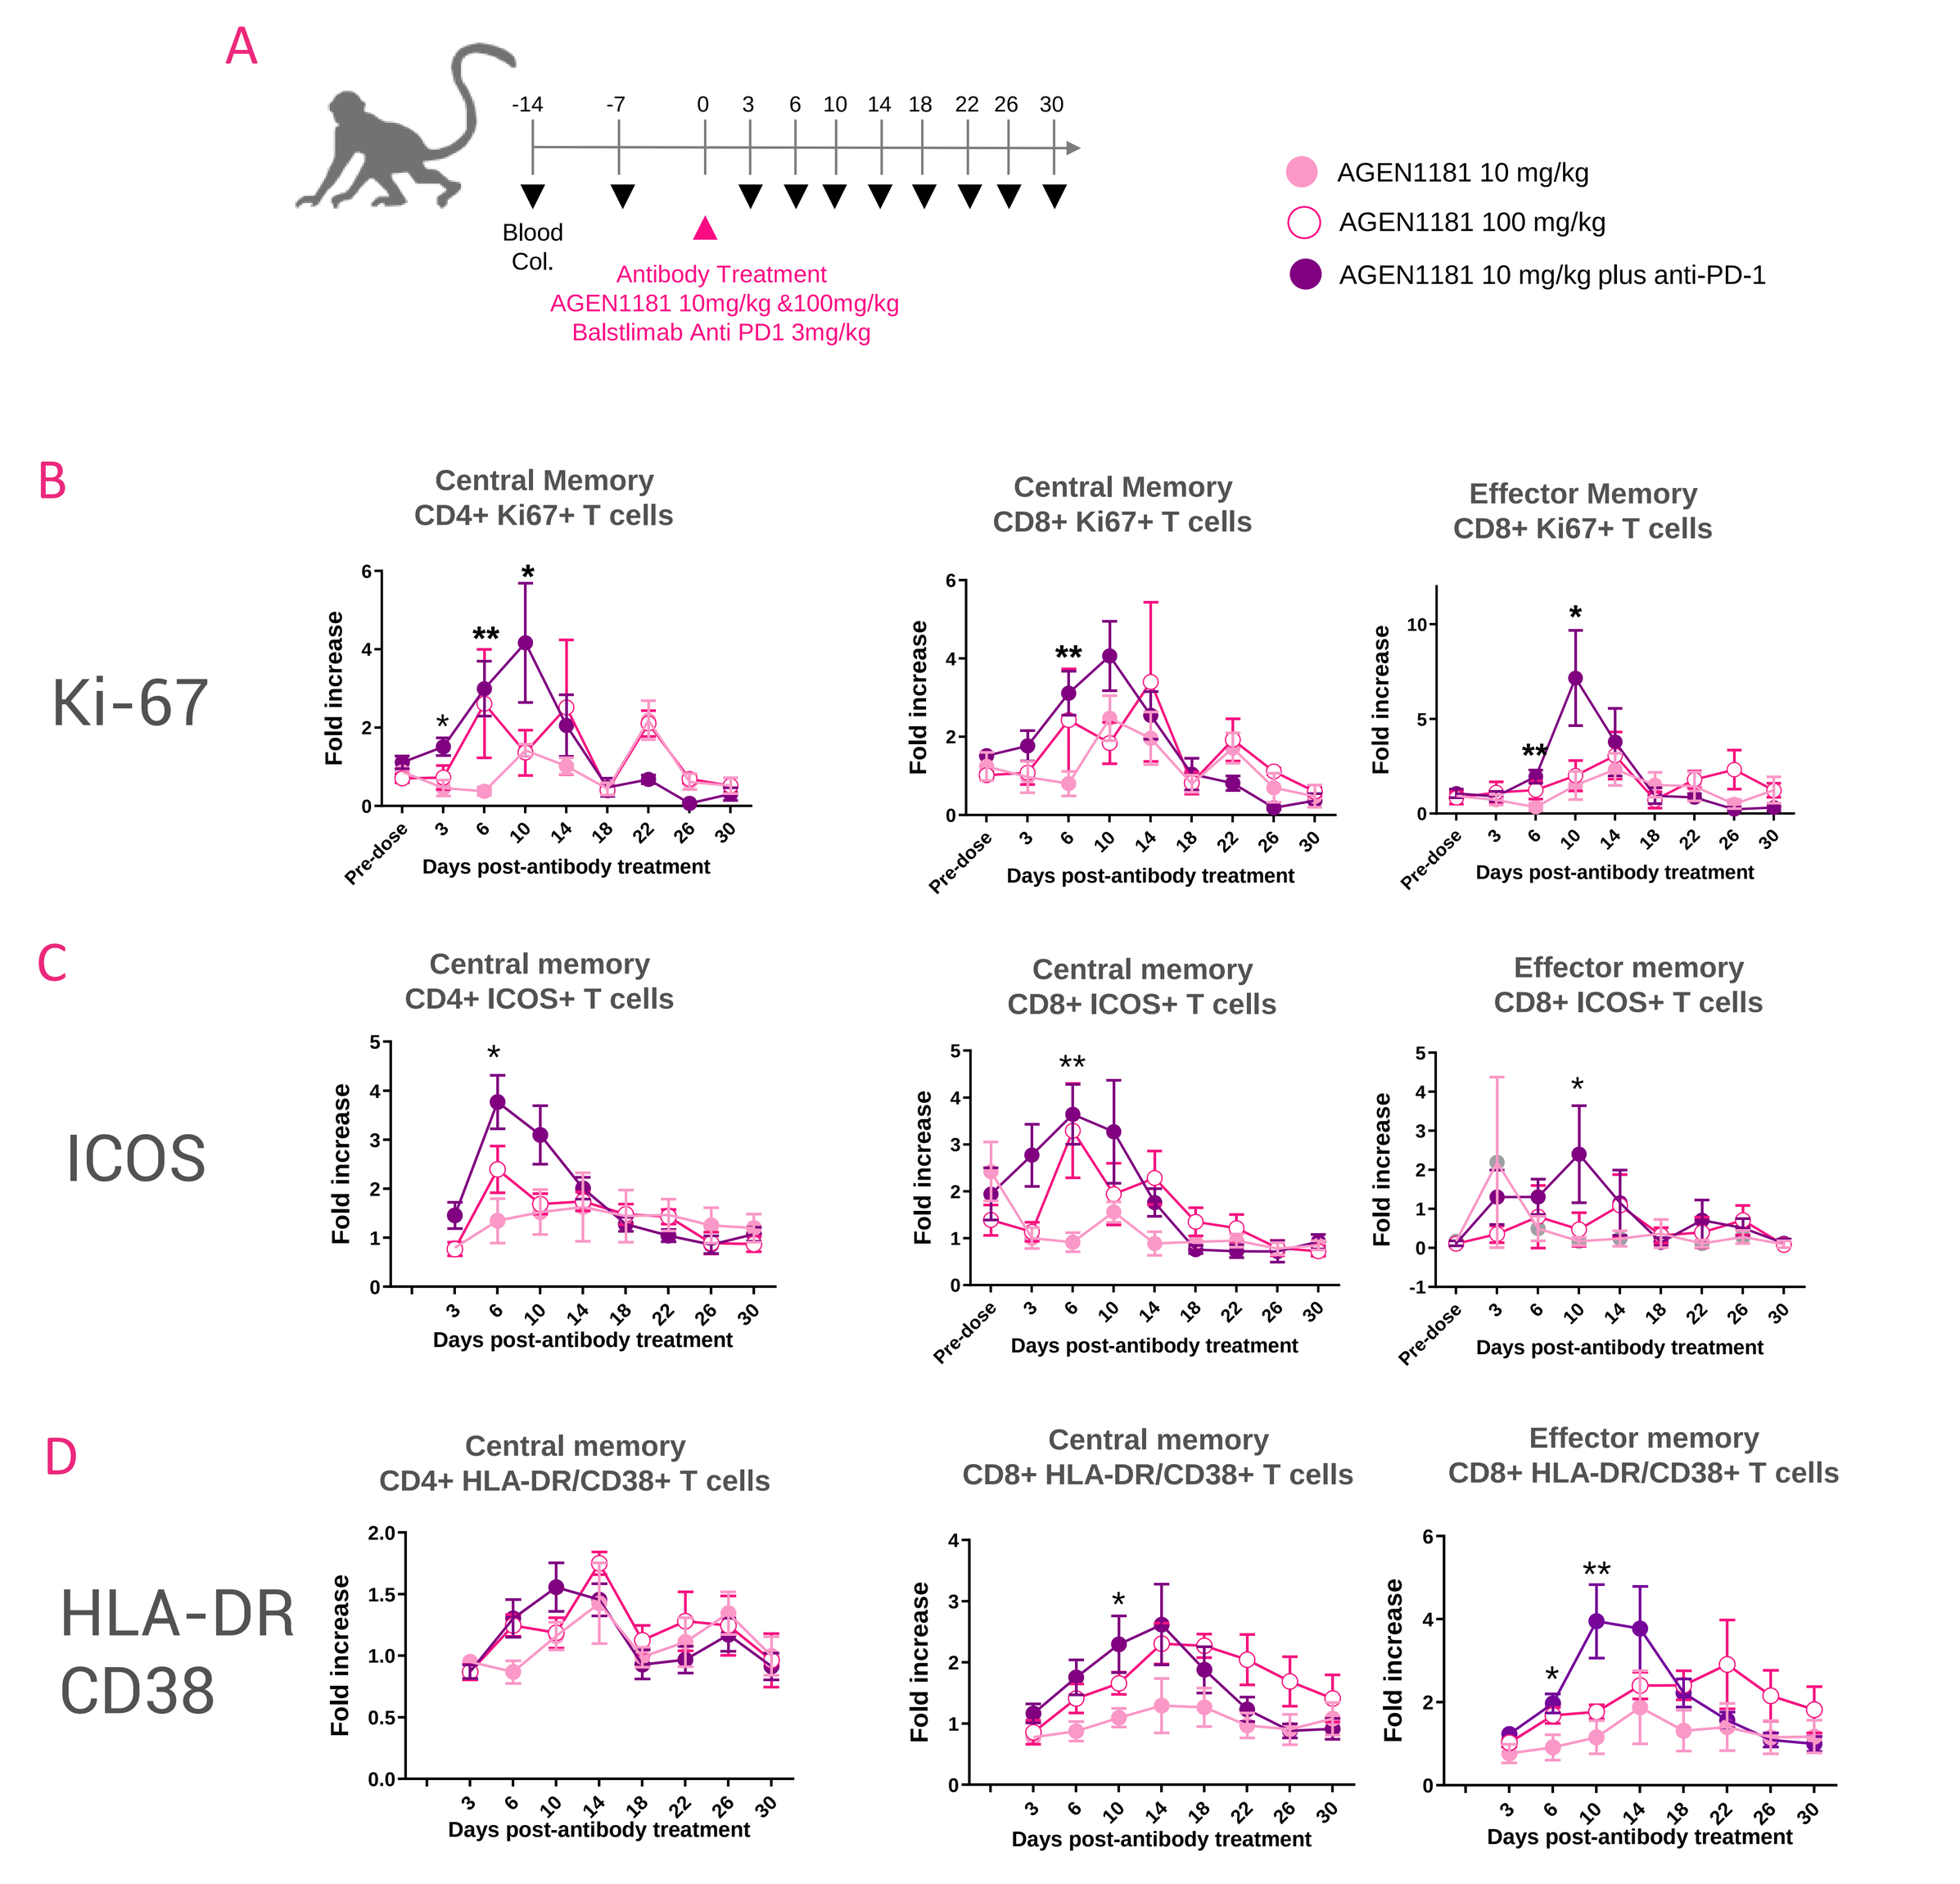
A
Blood Col.
Antibody Treatment
AGEN1181 10mg/kg &100mg/kg
Balstlimab Anti PD1 3mg/kg
AGEN1181 10 mg/kg
AGEN1181 100 mg/kg
AGEN1181 10 mg/kg plus anti-PD-1
B
Ki-67
Central Memory CD4+ Ki67+ T cells
Central Memory CD8+ Ki67+ T cells
Effector Memory CD8+ Ki67+ T cells
C
ICOS
Central memory CD4+ ICOS+ T cells
Central memory CD8+ ICOS+ T cells
Effector memory CD8+ ICOS+ T cells
D
HLA-DR CD38
Central memory CD4+ HLA-DR/CD38+ T cells
Central memory CD8+ HLA-DR/CD38+ T cells
Effector memory CD8+ HLA-DR/CD38+ T cells
Fold increase
Days post-antibody treatment

Figure 8: PD characterization of AGEN1181 on circulating T cells in the non-human primate.
A: Cynomologus monkeys were treated with single i.v. dose of AGEN1181 at 10 and 100 mg/kg as monotherapy and in combination (10mg/kg plus 3 mg/kg anti-PD-1). All treatments were safe and no acute toxicity was observed. Dose-dependent PD effects of monotherapy were enhanced by the combination with PD-1 based on B: increase in replicating central memory CD4 and CD8 T cells and effector memory CD8 T cells; C-D: acute activation markers of CD4 and CD8 T cells. Together, these data demonstrate a conserved broad effect of AGEN1181 on T cells as a priming agent and a functional enhancer which can be further enhanced by combination with anti-PD1.

Conclusions:

AGEN1181 is an Fc-enhanced anti-CTLA-4 mAb that demonstrates superior single agent and broader combination activity than conventional CTLA-4 mAbs

AGEN1181 engages multiple mechanisms of action to promote optimal anti-tumor immunity
- Promotes intratumoral Treg depletion and T cell infiltration
- Enhances T cell priming and the breadth and depth of tumor associated T cells
- Enhances T cell memory formation and activation to promote durable anti-tumor immunity

Promotes curative responses in checkpoint resistant preclinical cancer models such as PDAC-KPC as single therapy and in combination with immunogenic adjuvant chemotherapy.

Supplemental Information:

Expanding the Therapeutic Potential of anti-PD-1 and anti-CTLA-4 Therapy with Innovative Fc Engineering and Rationale Combinations for the Treatment of Solid Tumors.
AACR 2020 Poster # 922

On-going Trials:

Combination of AGEN1181 with Agenus's balstilimab (anti-PD-1) is advancing in the clinic (NCT03860272).
AACR 2021 Poster #1677

References:

[1]Waight et al. Cancer Cell 2018
[2]Danbee et al. PNAS 2018

Correspondence

Antoine Tanne: antoine.tanne@agenusbio.com
Dhan Chand: dhan.chand@agenusbio.com
Presented at The American Association for Cancer Research (AACR), April 9-14, 2021, Virtual Meeting